Baseline Demographics of the ACHieve Study: A Five-Year, Multi-National Observational Cohort Study of Children with Achondroplasia

Leanne M. Ward[1], Ravi Savarirayan[2], Philippe M. Campeau[3], Yuri Zarate[4], Ciara McDonnell[5], Janet Legare[6], Daniel Hoernschemeyer[7], Wolfgang Högler[8], Dirk Schnabel[9], Melita Irving[10], Yongguo Yu[11], Ying Zhang[12], Weijian Liu[12], Dorthe Viuff[13], Marie-Louise Hartoft-Nielsen[13]

[1]University of Ottowa, Ottowa, Toronto, [2]Royal Children's Hospital and University of Melbourne, Melbourne, Victoria AUS, [3]CHU Sainte-Justine Research Center, Montreal, QC, [4]University of Arkansas for Medical Sciences, Little Rock, AK USA, [5]Children's Health Ireland at Temple Street; Dublin, IRE, [6]University of Wisconsin School of Medicine and Publich Health, Madison, WI USA, [7]University of Wisconsin School of Medicine and Public Health; Madison, WI, [8]Department of Paediatrics and Adolescent Medicine, Johannes Kepler University Linz; Linz, Austria, [9]Charite Universitatsmedizin Berlin, [10]Guy's and St. Thomas' NHS Foundation Trust; London, UK, [11]Xinhua hospital Affiliate to Shanghai Jiaotong University School of Medicine; Shanghai, China, [12]Ascendis Pharma Inc., [13]Ascendis Pharma A/S

BACKGROUND

Achondroplasia (ACH)

- ACH is the most common short limbed skeletal dysplasia[1]
- Caused by a gain-of-function mutation in the fibroblast growth factor receptor 3 (FGFR3) gene and results in impairment of the endochondral ossification process[2]
- Global birth prevalence of ACH is approximately 4.6 per 100,000[3]

C-type natriuretic peptide (CNP)

- Promotes chondrocyte development through inhibition of the FGFR3 pathway, specifically through activation of NPR–B
- Potentially promising therapeutic target for treating growth failure and dwarfism, as it inhibits the overactive signalling through the mutated FGFR3 receptor causing ACH[4] (Figure 1)
- Due to its very short half-life (2–3 minutes), it can be speculated that a prolonged exposure of CNP is required for improved growth

Figure 1. CNP Can Counteract the Pathology of ACH

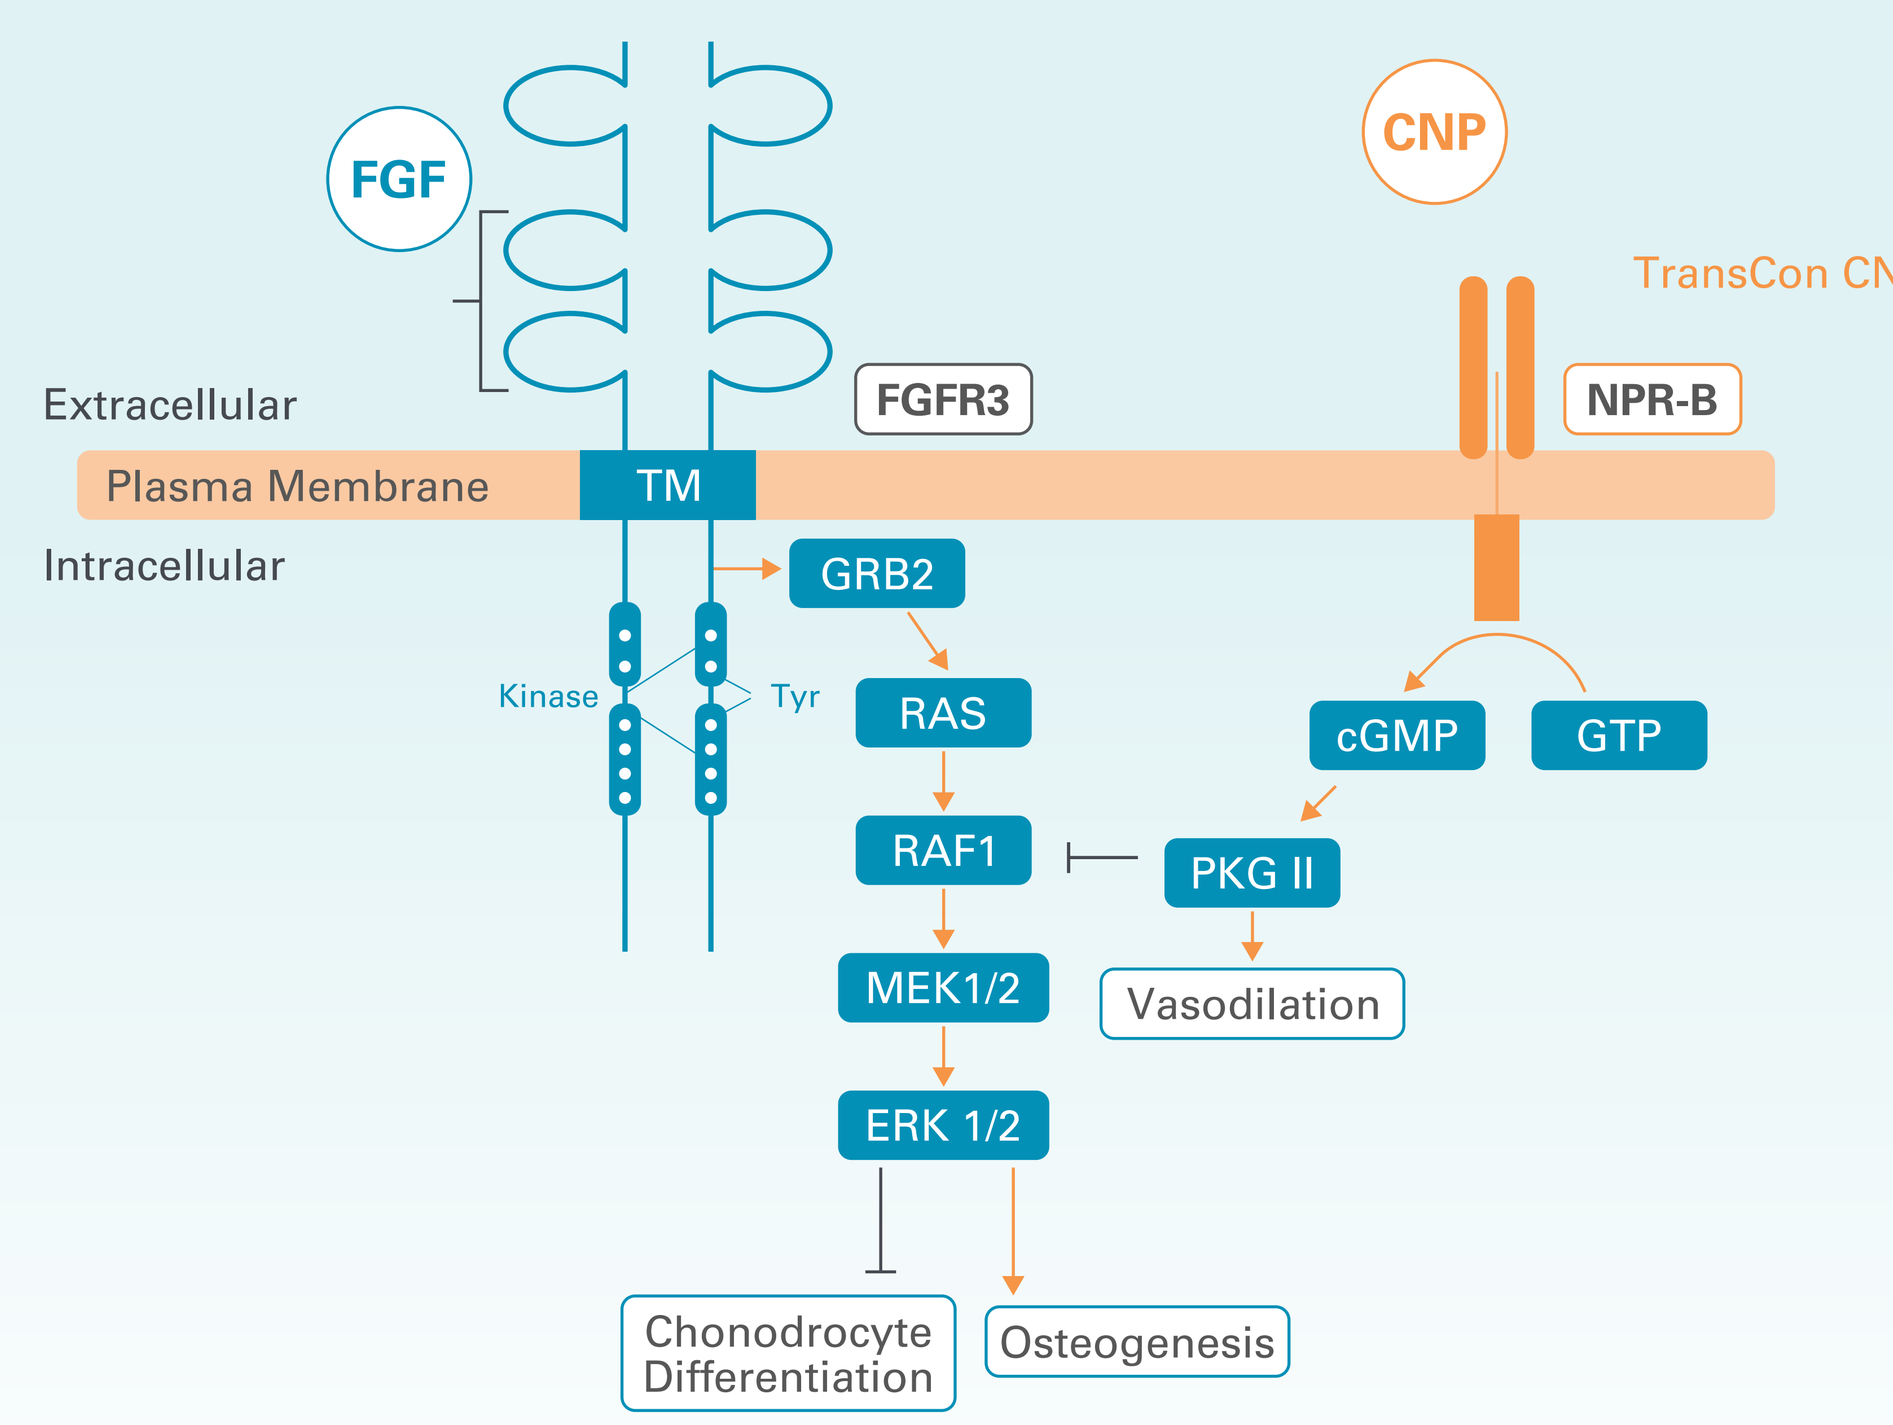
FGF
CNP
TransCon CNP
Extracellular
FGFR3
NPR-B
Plasma Membrane
TM
Intracellular
GRB2
Kinase
Tyr
RAS
cGMP
GTP
RAF1
PKG II
MEK1/2
Vasodilation
ERK 1/2
Chonodrocyte Differentiation
Osteogenesis

CNP does not alter the function of FGF receptors or change endogenous levels of FGF ligands, hereby reducing the risk of interfering with normal FGF biology

FGF, fibroblast growth factor; FGFR3, fibroblast growth factor receptor 3; CNP, C-type natriuretic peptide; NPR-B, natriuretic peptide receptor B; GRB2, Growth factor receptor-bound protein 2; cGMP, cyclic guanosine monophosphate; GTP, guanosine 5'-triphosphate; PKG II, protein kinase G II; RAF, rapidly accelerated fibrosarcoma kinases; MEK, Mitogen-activated protein kinase; ERK, extracellular signal-regulated kinase

Current clinical treatment approach

- Does not address underlying etiology, therefore includes symptomatic management, such as surgery and requires lifelong follow-up care[5]
- Surgical lengthening of lower limbs requires long-term hospitalization and risks serious complications such as infections, fractures, or deviation from the bone axis[6]

Morbidity

- Although changes in clinical management have improved survival, morbidity remains high and poses many challenges for individuals with ACH (Figure 2)

Figure 2. Achondroplasia Morbidity[7]

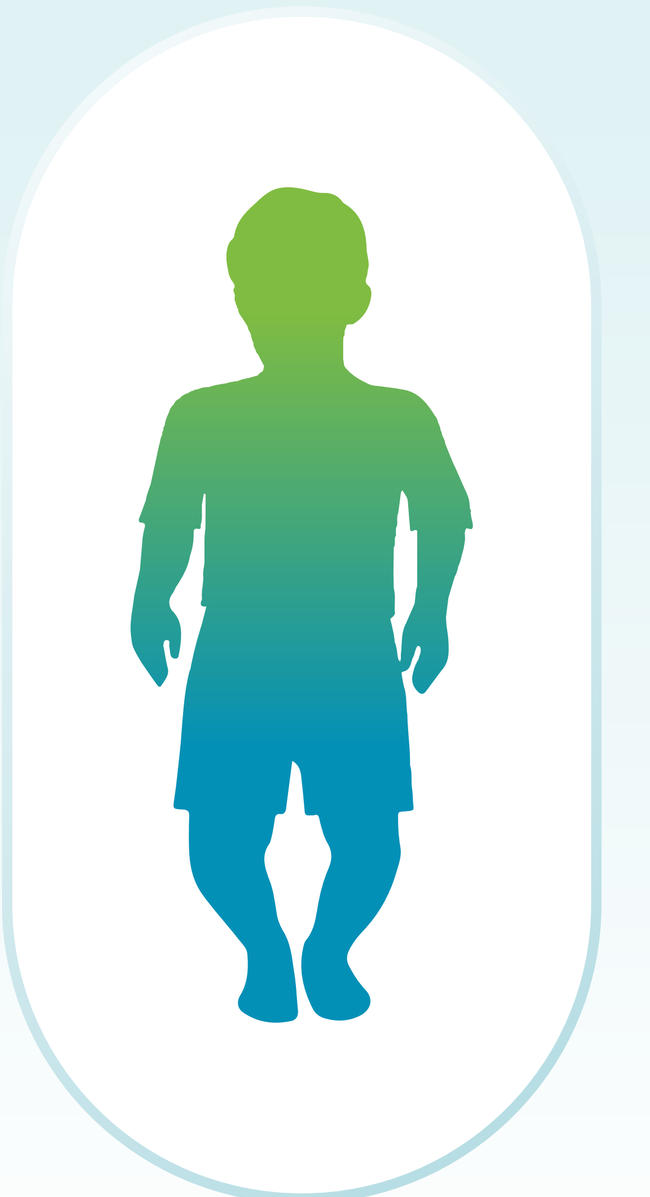

Up to **85%** of patients require intervention for obstructive sleep apnea and respiratory insufficiency

25% of children have hearing loss increasing to >50% in adulthood

22% have osteotomy

15-30% have fixed kyphotic deformity

Up to **28%** require cervicomedullary decompression by age 4

10% of children have neurological signs of spinal stenosis

80% of adults have clinical signs and symptoms related to spinal stenosis

AIMS AND OBJECTIVES

- The precise timing of comorbidity onset and the natural history of many of ACH features are not fully understood
- Prospective natural history studies are needed to provide important observational insights into the experience of children living with ACH and to inform the design and conduct of pathology-targeted intervention studies
- The goal of this analysis is to describe the design of a natural history study, ACHieve, that targets skeletal morbidity and provide interim longitudinal data in children with ACH
 - — The results of which will serve as an indirect reference for comparing clinical outcomes in children with ACH receiving an investigational drug, TransCon CNP

METHODS

STUDY DESIGN

- The ACHieve study is a multi-center, longitudinal, observational cohort study in children with ACH from birth up to 8 years at enrollment
- Children are evaluated every six months for up to 5 years in 25 centers from North America, Europe, China, and Oceania
- At each assessment, children undergo comprehensive anthropometric studies (including body proportionality measurements and recumbent or standing height) and information on the timing and nature of ACH-related comorbidities and their treatments are collected
- The first patient was enrolled in 2019 and a total of 149 children are included in this analysis

ACHieve STUDY

Primary Outcome Measures

- Annualized height velocity (centimeters/year) in children with ACH, for up to 5 years

Secondary Outcome Measures

- Collection of natural history of achondroplasia manifestations and symptoms in children with ACH, for up to 5 years

KEY INCLUSION CRITERIA

- Clinical diagnosis of ACH
- Age 0 – 8 years old at enrollment
- Able to stand without assistance if the child is 24 months or older

KEY EXCLUSION CRITERIA

Prior treatment with:

- — Human growth hormone (hGH) or other medicinal products intended to affect stature or body proportionality
- — Medicinal products intended to affect stature or body proportionality within the previous 6 months of screening
- — Any investigational medicinal product or device intended to affect stature or body proportionality at any time

History or presence of:

- — Injury or disease of the growth plate(s), other than ACH, that affects growth potential of long bones
- — Bone-related surgery that affects growth potential of long bones. Limb-lengthening with full recovery is allowed with a minimum of 12 months of bone healing
- — Forms of skeletal dysplasias other than ACH or medical conditions that result in short stature or abnormal bone growth
- — Malignant disease, other than basal cell epithelioma/carcinoma or completely resected squamous skin cancer with no recurrence for 12 months per medical records

BASELINE DEMOGRAPHICS AND INTERIM LONGITUDINAL DATA

CURRENT STATUS

- 5-year timeframe to characterize the natural history of ACH. A total of 200 children are targeted for enrollment, follow-up is anticipated to last until 2024
 - — As of 11 February 2021, 149 enrolled across 12 countries (Figure 3)

Figure 3. ACHieve Study Enrollment by Country

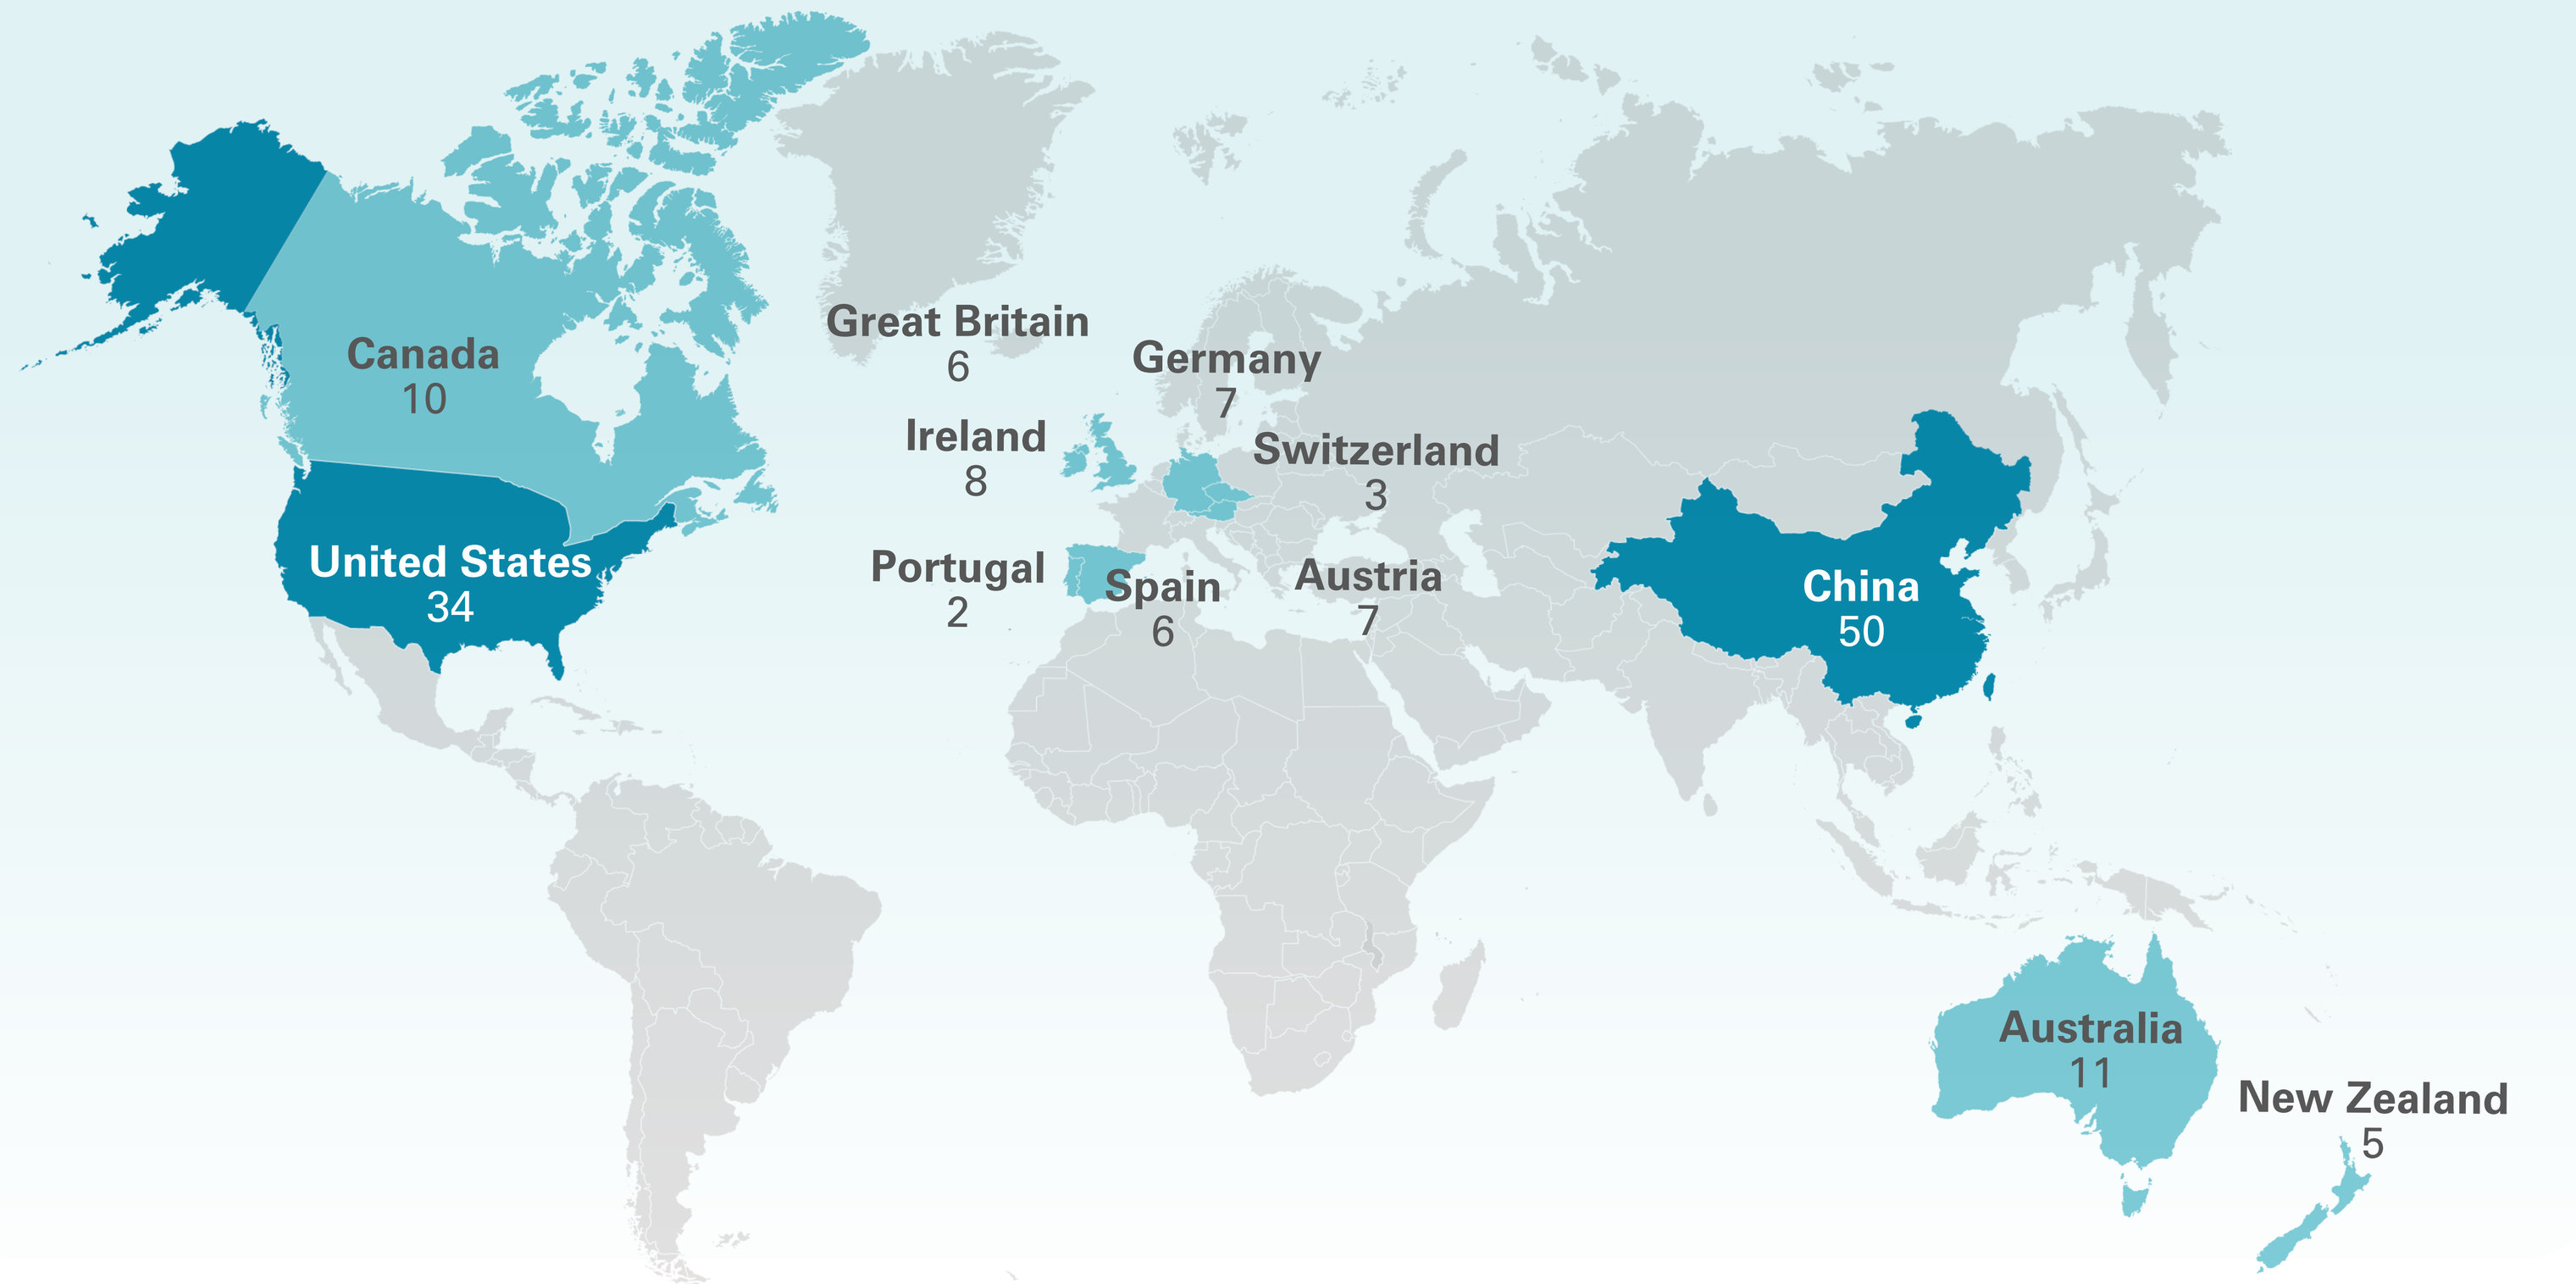
Canada 10
United States 34
Great Britain 6
Ireland 8
Portugal 2
Spain 6
Germany 7
Switzerland 3
Austria 7
China 50
Australia 11
New Zealand 5

Table 1. ACHieve Preliminary Demographics and Baseline Characteristics

Demographic (N = 149)	
Age (years)	
Means (SD)	3.7 (2.3)
Median (Min, Max)	3.1 (0.3, 9.0*)
Age Group (years) – n (%)	
< 2	40 (26.8)
2 – < 5	69 (46.3)
≥ 5	40 (26.8)
Sex – n (%)	
Female:Male	78 (52.3):71 (47.7)
BMI (kg/m²) Mean (SE) by Age Group (years)	
< 2	19.1 (0.4)
2 – < 5	20.9 (0.2)
≥ 5	21.1 (0.6)
Height SDS Mean (SE) by Age Group (years)**	
< 2	–3.6 (0.2)
2 – < 5	–4.6 (0.1)
≥ 5	–5.5 (0.2)
AHV* (cm/yr) Mean (SE) by Age Group (years)**	
< 2	15.2 (1.1)
2 – < 5	5.6 (0.8)
≥ 5	3.6 (0.2)
Race – n (%)	
White	84 (56.4)
Asian	56 (37.6)
American Indian or Alaska Native	1 (0.7)
Black or African American	1 (0.7)
Other	7 (4.7)
Height (cm) Mean (SD) by Age Group (years)	
< 2	65.0 (4.8)
2 – < 5	79.0 (5.0)
≥ 5	93.3 (5.2)
Age at ACH Diagnosis – n (%)≠	
Pre-Birth	19 (12.8)
At Birth	17 (11.4)
0 – 6 Months	70 (47.0)
> 6 Months	42 (28.2)
Type of Mutations – n (%)≠	
1138G > A or 1138G > C	134 (89.9)
Other	7 (4.7)
Unknown	8 (5.4)

*Age is rounded to the one decimal, **height expressed in Standard Deviation Score (SDS) of normal height children is derived using CDC 2000 (United States of America)/Kuczmarski method, ***Baseline AHV based on heights antecedent to study baseline (range approximately 12 months), ≠1 subject with missing data

Figure 4. Anthropometric Parameters by Visit

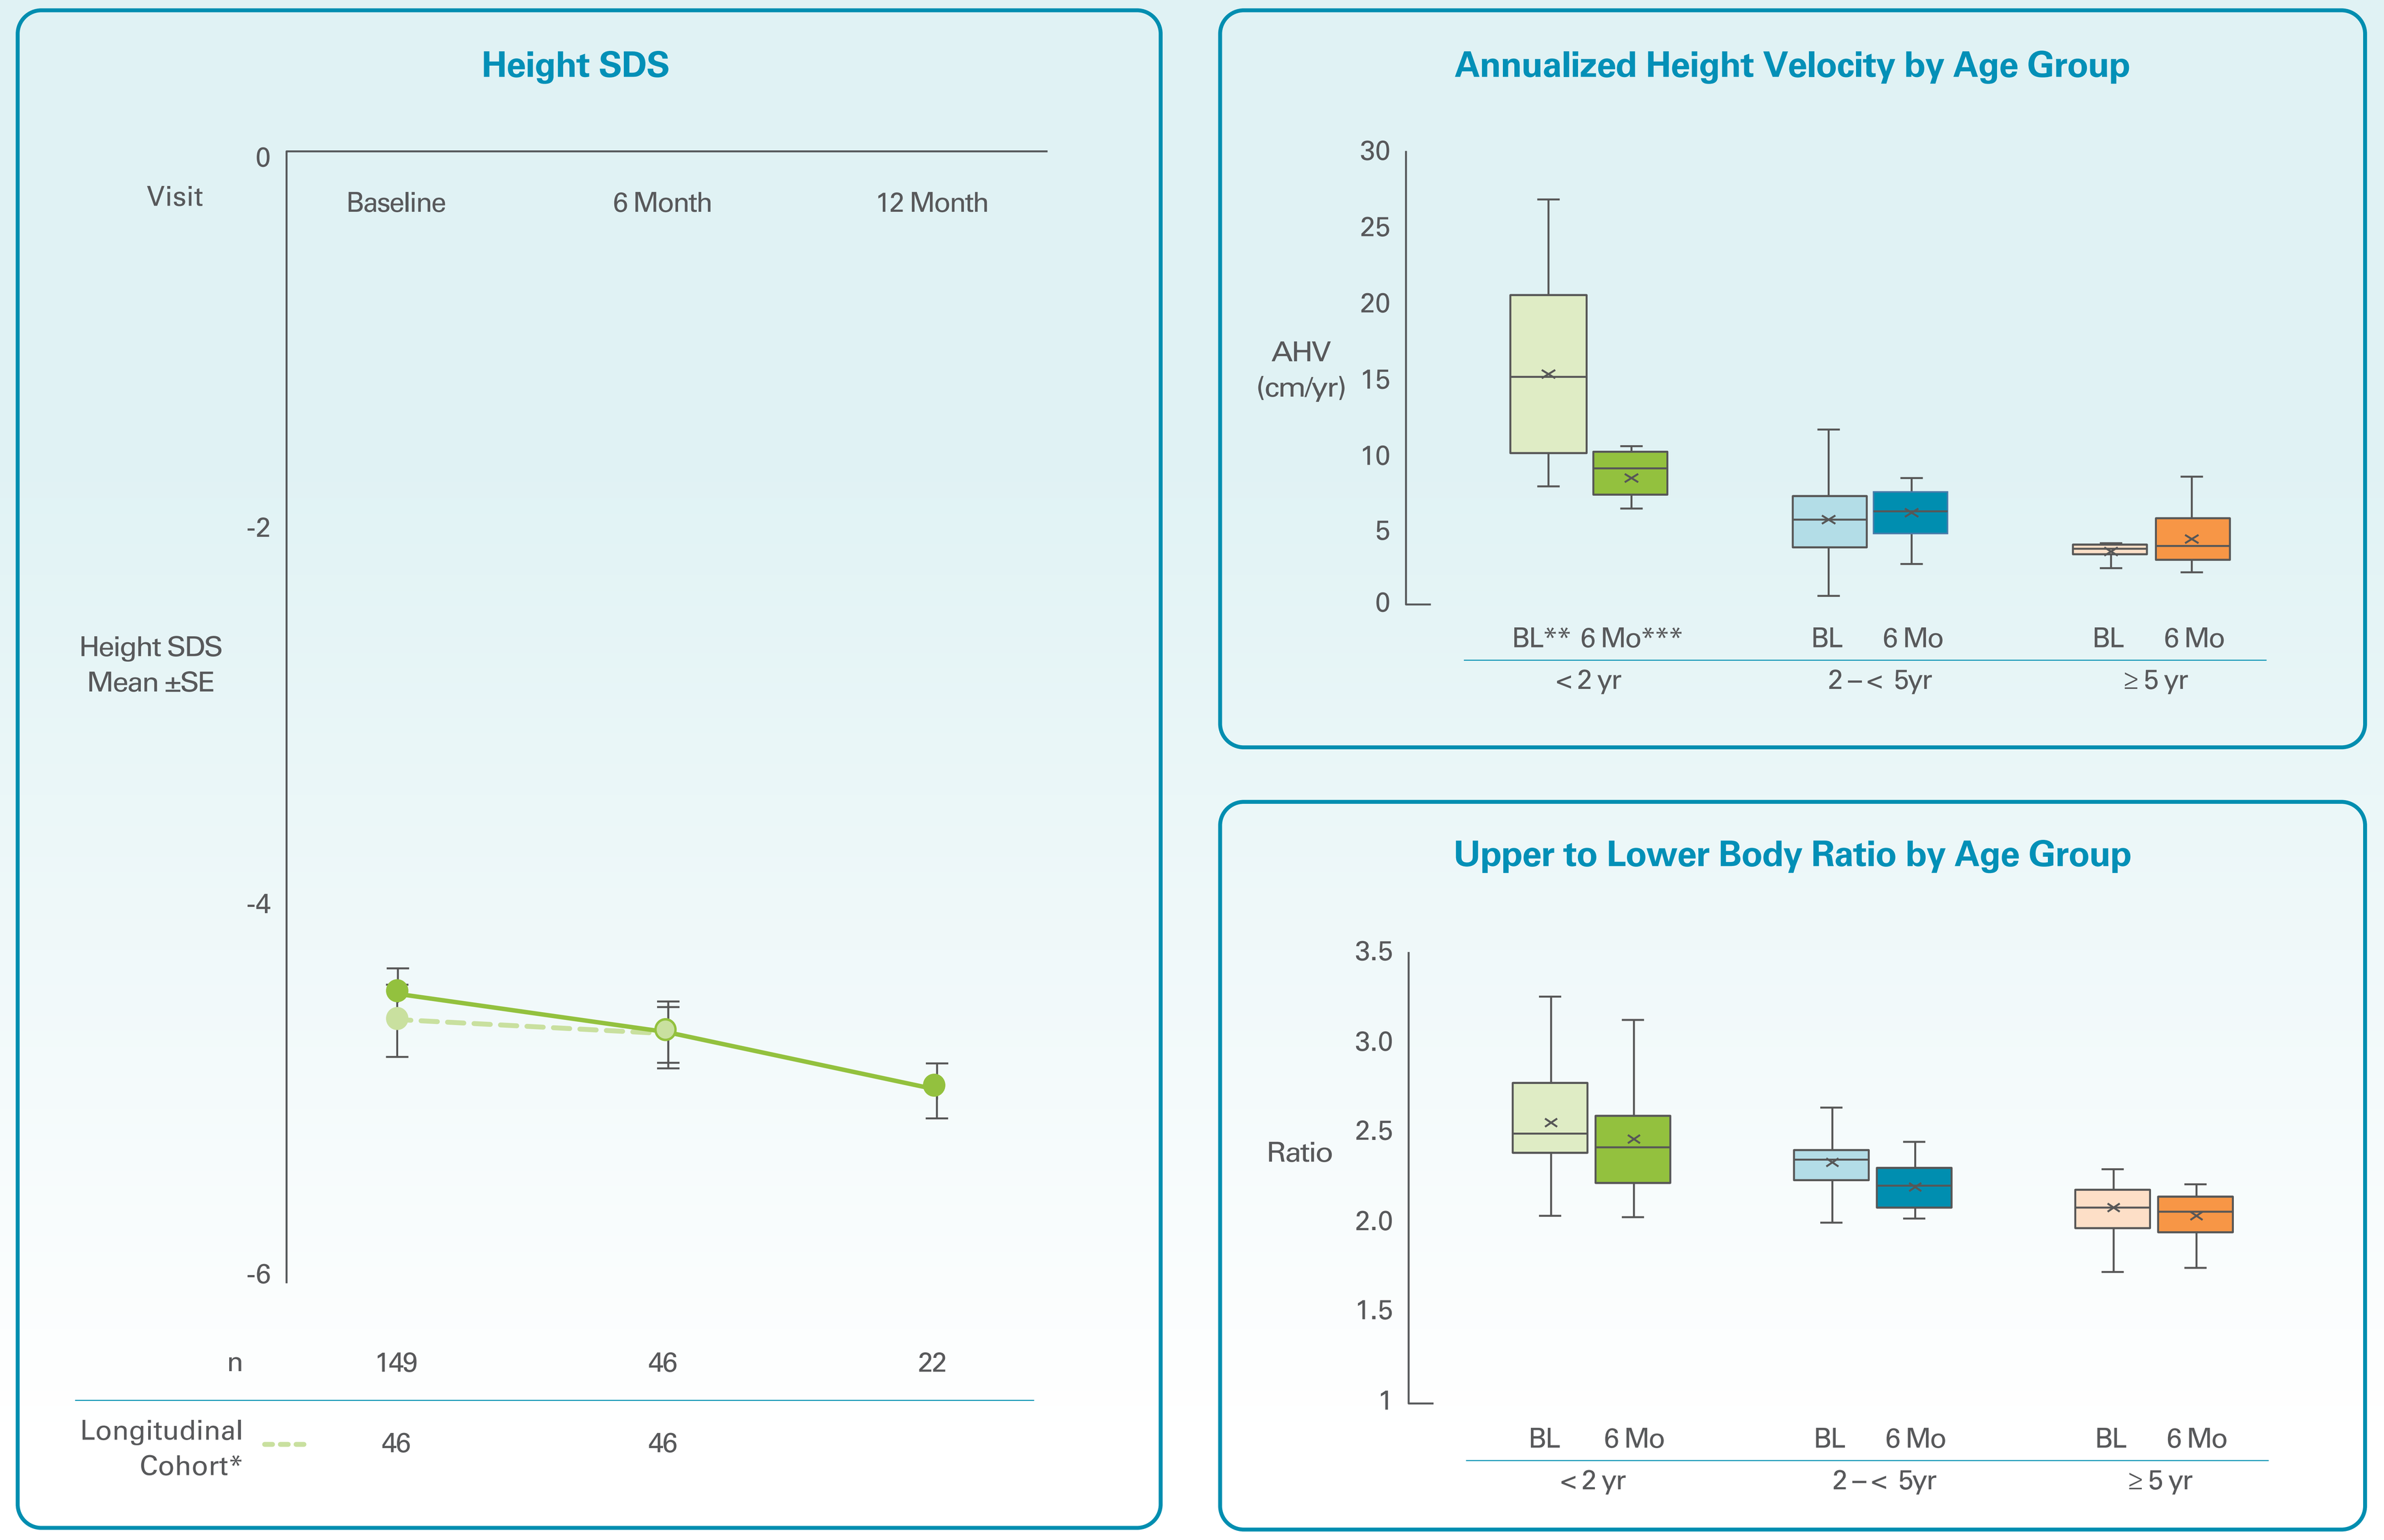
Height SDS
Visit
Baseline
6 Month
12 Month
0
-2
-4
-6
Height SDS Mean ±SE
n
149
46
22
Longitudinal Cohort*
46
46
Annualized Height Velocity by Age Group
AHV (cm/yr)
30
25
20
15
10
5
0
BL** 6 Mo***
BL 6 Mo
BL 6 Mo
< 2 yr
2 –< 5yr
≥ 5 yr
Upper to Lower Body Ratio by Age Group
Ratio
3.5
3.0
2.5
2.0
1.5
1
BL 6 Mo
BL 6 Mo
BL 6 Mo
< 2 yr
2 –< 5yr
≥ 5 yr

*subjects at baseline who have completed Month 6, **Baseline AHV based on heights antecedent to study baseline (range-approximately 12 months), ***after 6 months
BL = baseline, Mo = month

SUMMARY AND FUTURE DIRECTIONS

Summary

- The ACHieve natural history study of children with ACH has been launched in 12 countries with close to 150 participants
- Most children were diagnosed in the first six months of life, and carry one of two mutations in the FGFR3 gene
- Initial 6-month data suggest that the annualized growth velocity and the upper to lower body ratio has age group-dependent patterns

Future Directions

- The ACHieve study will continue to collect data on the timing, frequency, and characteristics of growth patterns, body proportionality, and comorbidities in children with ACH
- These observations may serve as an indirect reference for future intervention trials targeting the pathology of the underlying skeletal dysplasia with an investigational drug, TransCon CNP (Figure 5)

Figure 5. TransCon CNP Design

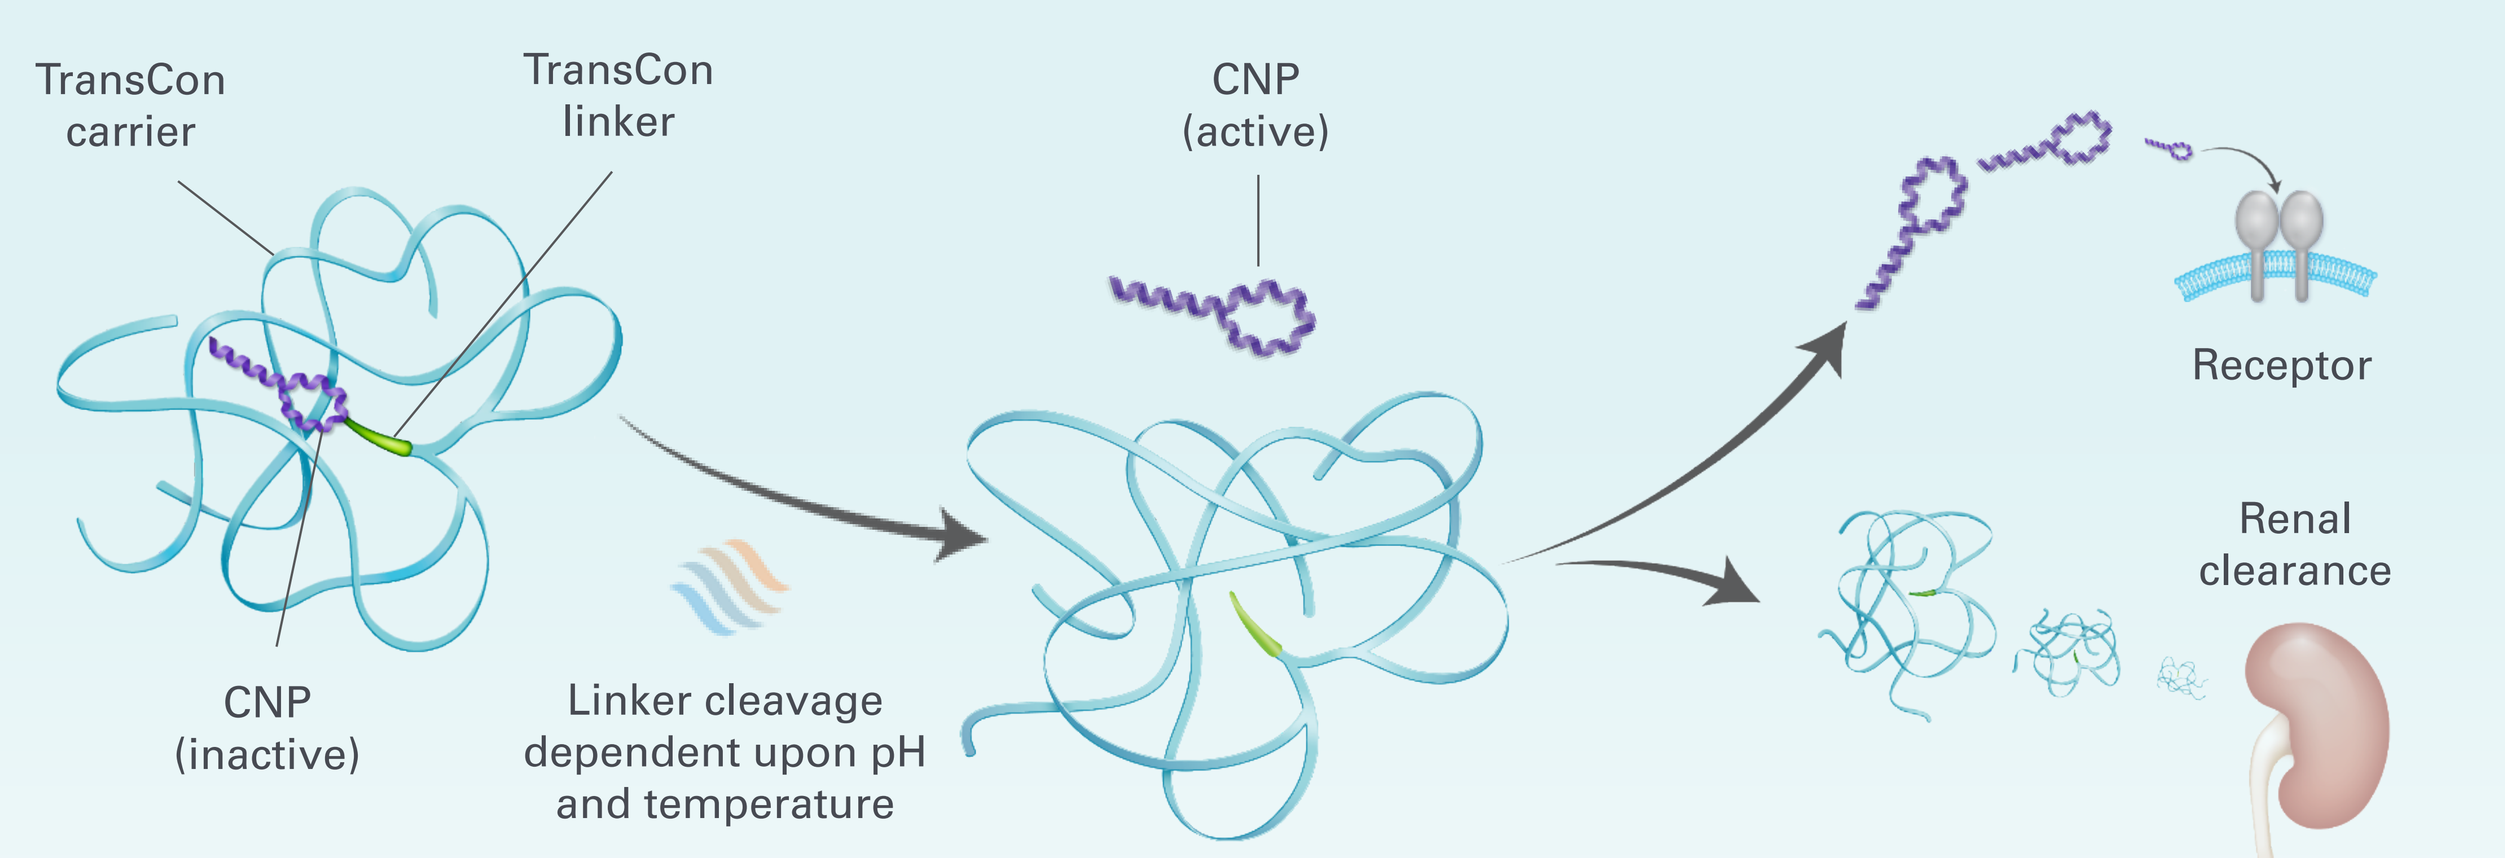
TransCon carrier
TransCon linker
CNP (active)
Receptor
Renal clearance
CNP (inactive)
Linker cleavage dependent upon pH and temperature

- TransCon technology is designed to provide effective shielding of CNP[8]:
 - — From neutral endopeptidase degradation in subcutaneous tissue and blood compartment
 - — Minimize binding of TransCon CNP to the NPR-C receptor
 - — Reduce binding of TransCon CNP to the NPR-B receptor in vasculature to avoid hypotension
- CNP liberated from TransCon CNP maintains small enough size to allow penetration into growth plates

- TransCon CNP is an investigational, sustained-release prodrug of CNP. CNP is transiently bound via the TransCon Linker to a chemically inert carrier that prolongs the peptide's overall circulatory half-life. This is achieved by minimizing renal clearance of the TransCon CNP prodrug and shielding of the CNP molecule from proteolytic degradation and from binding to its primary activating and clearance receptors, NPR-B and NPR-C
- Following cleavage of the TransCon Linker under physiological pH and temperature, active CNP peptide is slowly and continuously released
- The Phase 1 clinical trial with TransCon CNP demonstrated that single doses up to 150 μg CNP/kg were well-tolerated in healthy adult male volunteers, with no clinically significant trends observed in clinical laboratory assessments, vital sign measurements, ECG parameters, or physical examination findings[9,10]

REFERENCES

1. Horton WA, *et al. Lancet.* 2007;370:162-72. **2.** Rousseau F, *et al. Nature.*1994;371(6494):252-4. **3.** Foreman PK, et al. *Am J Med Genet.* 2020;182A:2297-2316. **4.** Lorget F, et al. *Am J Hum Genet.* 2012;91(6):1108-14. **5.** Pauli RM. *Orphanet J Rare Dis.* 2019;14(1):1-49. **6.** Miccoli M, et al. *Horm Res Paediatr.* 2016;86(1):27-34. **7.** Ireland PJ, et al. *Appl Clin Genet.* 2014;7:117-125. **8.** Breinholt VM, et al. *J Pharmacol Exp Ther.* 2019;370(3):459-471. **9.** Miccoli M, et al. *Horm Res Paediatr.* 2016;86(1):27-34. **10.** Ota S, et al. Presented at: 14th Annual Meeting of the International Skeletal Dysplasia Society; Sept 11-14, 2019; Oslo, Norway.