Efficacy and Safety of Up to 2 Years of Treatment with TransCon hGH (lonapegsomatropin) In Treatment-Naïve and Treatment-Experienced Children with Growth Hormone Deficiency

Aristides K. Maniatis[1], Samuel J. Casella[2], Ulhas M. Nadgir[3], Paul Hofman[4], Paul Saenger[5], Elena D. Chertok[6], Elena M. Aghajanova[7], Wenjie Song[8], Meng Mao[8], Steven Chessler[8], Allison Komirenko[8], Michael Beckert[9], Aimee D. Shu[8], Paul S. Thornton[10]

[1]Rocky Mountain Pediatric Endocrinology, Centennial, CO, USA; [2]Children's Hospital at Dartmouth-Hitchcock, Lebanon, NH; [3]Center of Excellence in Diabetes and Endocrinology, Sacramento, CA; [4]Liggins Institute, University of Auckland, Auckland, New Zealand; [5]NYU Langone Health, New York, NY; [6]Voronezh State Medical University, Voronezh, Russia; [7]Yerevan State Medical University, Yerevan, Armenia; [8]Ascendis Pharma, Inc, Palo Alto, CA; [9]Ascendis Pharma A/S, Hellerup, Denmark; [10]Cook Children's Health Care System, Ft. Worth, TX.

BACKGROUND

- Once-weekly lonapegsomatropin (TransCon hGH) is an investigational prodrug for growth hormone deficiency (GHD) that consists of 3 components: unmodified somatropin, an inert carrier that protects it, and a linker that temporarily joins the two[1,2] (Figure 1)

Figure 1. Lonapegsomatropin (TransCon hGH) Design

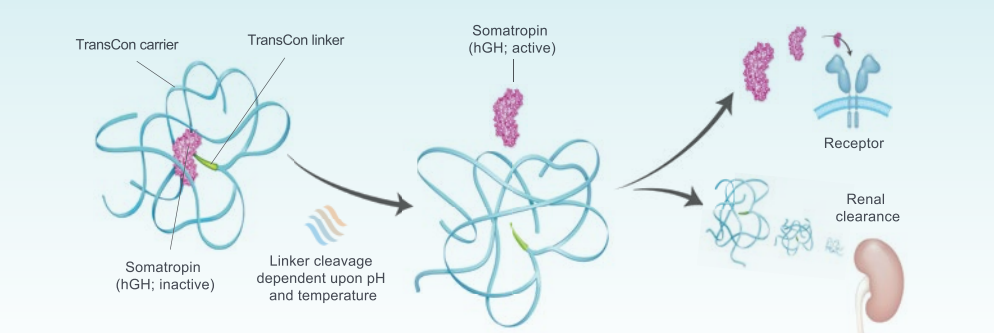
TransCon carrier
TransCon linker
Somatropin (hGH; active)
Receptor
Renal clearance
Somatropin (hGH; inactive)
Linker cleavage dependent upon pH and temperature

Once-weekly prodrug releases unmodified somatropin designed to mimic daily somatropin:

• Tissue Distribution • Physiological levels • Therapeutic effects: efficacy, safety and tolerability

- In the pivotal phase 3 heiGHt Trial evaluating treatment-naïve children with GHD, lonapegsomatropin demonstrated superior annualized height velocity (AHV) and statistically greater change from baseline in height standard deviation score (Δ height SDS) at 52 weeks compared to daily somatropin therapy (Genotropin) and had a similar safety and tolerability profile[3]
- In the phase 3 fliGHt Trial, children who switched from daily somatropin to lonapegsomatropin continued to grow well and maintained a good safety profile[4]
- Results are reported from heiGHt and fliGHt subjects who continued into the enliGHten open-label long-term extension (OLE) Trial for up to 52 weeks (data cut: June 1st 2020)

METHODS

TRIAL DESIGN

PHASE 3 HEIGHT TRIAL

- heiGHt was a 52-week, open-label, active-controlled, pivotal phase 3 trial in which treatment-naïve, prepubertal subjects (males 3-12; females 3-11 years old) with GHD were randomized 2:1 to receive once-weekly lonapegsomatropin 0.24 mg hGH/kg/week via vial/syringe or an equivalent weekly dose of daily Genotropin via pen device

PHASE 3 FLIGHT TRIAL

- fliGHt was a 26-week, open-label phase 3 trial in which treatment-experienced subjects (6 months to 17 years old; subjects < 3 years old could be treatment-naïve) with GHD switched from their previous daily somatropin to lonapegsomatropin 0.24 mg hGH/kg/week via vial/syringe

ENLIGHTEN OLE TRIAL

- All subjects who enrolled in the long-term extension trial received lonapegsomatropin at their previous dose via vial/syringe (daily somatropin subjects from heiGHt started lonapegsomatropin 0.24 mg hGH/kg/week) (Figure 2)
- Subjects in the US switched to the TransCon hGH Auto-Injector when available

Figure 2. Lonapegsomatropin Phase 3 Clinical Program

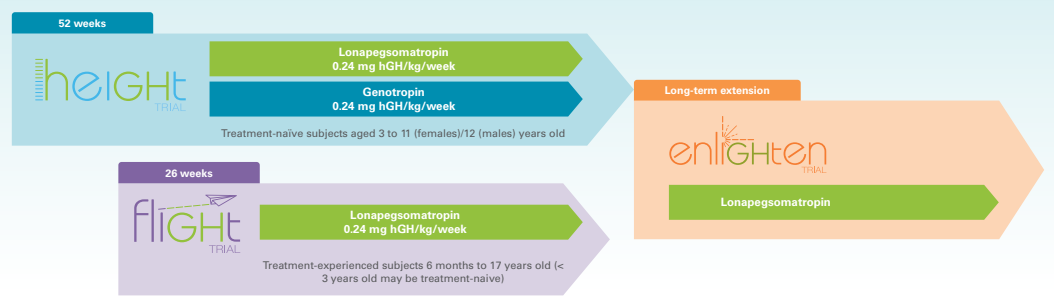
52 weeks
heiGHt TRIAL
Lonapegsomatropin 0.24 mg hGH/kg/week
Genotropin 0.24 mg hGH/kg/week
Treatment-naïve subjects aged 3 to 11 (females)/12 (males) years old
26 weeks
fliGHt TRIAL
Lonapegsomatropin 0.24 mg hGH/kg/week
Treatment-experienced subjects 6 months to 17 years old (< 3 years old may be treatment-naive)
Long-term extension
enliGHten TRIAL
Lonapegsomatropin

OUTCOMES

EFFICACY

- Growth outcomes were evaluated approximately every 13 weeks
- Three groups were analyzed:
 - Subjects treated with lonapegsomatropin in heiGHt, followed by continuation of lonapegsomatropin in enliGHten
 - Subjects treated with daily somatropin in heiGHt, followed by lonapegsomatropin in enliGHten
 - Subjects treated with lonapegsomatropin in fliGHt, followed by continuation of lonapegsomatropin in enliGHten
- Comparisons between the two heiGHt treatment groups allowed for the evaluation of safety and efficacy outcomes as they had similar baseline demographics and comparable treatment histories
- IGF-1 was obtained on post-dose Day 5 (±1) in fliGHt and enliGHten; in heiGHt, average IGF-1 for lonapegsomatropin was calculated based on a population pharmacodynamic model

SAFETY

- Safety was evaluated throughout the trial periods and is summarized by trial

STATISTICAL ANALYSIS

- A by-visit ANCOVA model was used to analyze numeric endpoints

RESULTS

DISPOSITION, DEMOGRAPHICS, AND BASELINE CHARACTERISTICS

- Nearly all subjects who completed heiGHt (158/159) and fliGHt (140/144) continued into enliGHten (Table 1)
 - Eight (3.4%) subjects have prematurely withdrawn from the trial for the following reasons: 4 subjects (1.3%) due to withdrawn consent, 2 (0.7%) for protocol violation, and 2 for "other" reasons
 - As of the data cut, 2 subjects have achieved near adult height (AHV < 2 cm/year over the last 9 months or bone age > 14 years [females] or >16 years [males]) and thus have completed the trial
- As previously described, baseline demographics were balanced between groups in heiGHt (Table 2). Subjects enrolled in fliGHt were primarily treatment-experienced (98%) and ranged from 1.2 to 17.4 years old at the start of the fliGHt Trial
- Upon entry into enliGHten, subjects from fliGHt were generally older and more advanced in Tanner Stage compared to those entering from heiGHt (Table 3)

Table 1. Subject Disposition

	heiGHt		fliGHt
	Lonapegsomatropin n (%)	Daily somatropin n (%)	Lonapegsomatropin n (%)
Enrolled and dosed in parent trial	105	56	146
Completed parent trial	104 (99.0)	55 (98.2)	144 (98.6)
Enrolled and dosed in enliGHten	103 (98.1)	55 (98.2)	140 (95.9)
Withdrew from enliGHten[a]	3 (2.9)	1 (1.8)	4 (2.9)
Completed enliGHten[b]	0	0	7 (5.0)

[a]Denominator based on subjects enrolled and dosed in enliGHten.
[b]Each subject's participation is expected to last up until: the product is approved and commercially available, alternative arrangements have been made for continued subject access to hGH treatment, or treatment for pediatric growth hormone deficiency is no longer considered appropriate (including – but not limited to – when there is evidence of closed epiphyses [bone age >14.0 years for females; >16.0 years for males]). When any of the 3 criteria is met, the subject will be considered to have completed the extension trial. The total duration of participation for each subject in the trial will therefore depend on the conditions listed above and may be different in different countries and regions.

- The mean dose of lonapegsomatropin remained approximately 0.24 mg hGH/kg/wk for subjects from heiGHt at Week 104 and was 0.20 mg hGH/kg/wk for subjects from fliGHt at Week 78

Table 2. Demographics and Baseline Characteristics at Start of heiGHt and fliGHt

	heiGHt		fliGHt
	Lonapegsomatropin n (%)	Daily somatropin n (%)	Lonapegsomatropin n (%)
Age, n	105	56	146
Mean (SD)	8.5 (2.7)	8.5 (2.8)	10.6 (3.9)
Min, max	3.3, 13.1	3.2, 12.9	1.2, 17.4
Gender, n	105	56	146
Males, n (%)	86 (81.9)	46 (82.1)	110 (75.3)
Height SDS, n	105	56	146
Mean (SD)	-2.9 (0.8)	-3.0 (0.9)	-1.4 (0.8)
Δ Average Parental Height, n	103	56	141
Mean (SD)	-2.3 (1.1)	-2.6 (1.3)	-1.1 (1.0)
IGF-1 SDS, n	105	56	1461
Mean (SD)	-2.1 (0.9)	-2.0 (1.0)	0.9 (1.3)
Peak stimulated GH level prior to initiating GH therapy, n	105	56	143
Mean (SD)	5.9 (2.8)	5.5 (3.0)	5.9 (2.6)
≤5 ng/mL, n (%)	37 (35.2)	21 (37.5)	52 (35.6)
>5 ng/mL, n (%)	68 (64.8)	35 (62.5)	91 (62.3)

Demographics and baseline characteristics are presented for the time of enrollment into fliGHt; subjects had been previously treated for an average of 1.14 years of daily somatropin therapy.

Table 3. Demographics and Disease Characteristics at Start of enliGHten

	heiGHt		fliGHt
	Lonapegsomatropin n (%)	Daily somatropin n (%)	Lonapegsomatropin n (%)
Age, n	103	55	140
Mean (age)	9.5 (2.7)	9.5 (2.8)	11.1 (3.9)
Min, max	4.4, 14.1	4.2, 13.9	1.7, 17.8
Height SDS, n	103	55	140
Mean (SD)	-1.9 (0.7)	-2.1 (0.8)	-1.1 (0.8)
IGF-1 SDS, n	102	54	138
Mean (SD)	-0.7 (1.0)[a]	-0.1 (1.2)	1.6 (1.3)[b]
Tanner Stage, n	103	55	140
Stage I, n (%)	92 (89.3)	45 (81.8)	77 (55.0)
Stage II, n (%)	11 (10.7)	8 (14.5)	21 (15.0)
Stage III, n (%)	0	2 (3.6)	22 (15.7)
Stage IV, n (%)	0	0	17 (12.1)
Stage V, n (%)	0	0	3 (2.1)

[a]In heiGHt, IGF-1 SDS was drawn before dosing with lonapegsomatropin and represents a trough value.
[b]In fliGHt, IGF-1 SDS was drawn at Day 5 (±1) after dosing with lonapegsomatropin and represents average IGF-1 SDS.

HEIGHT AND PHARMACODYNAMIC OUTCOMES

- The treatment difference in least-squares (LS) mean Δ height SDS (lonapegsomatropin vs daily somatropin) at the end of heiGHt (Week 52) was 1.10 vs 0.96 ($P = 0.015$), and was numerically, but not significantly, greater at Week 104 (1.61 vs 1.49, $P = 0.158$ in favor of lonapegsomatropin (Figure 3)

Figure 3. Δ Height SDS Over 104 Weeks for heiGHt Subjects

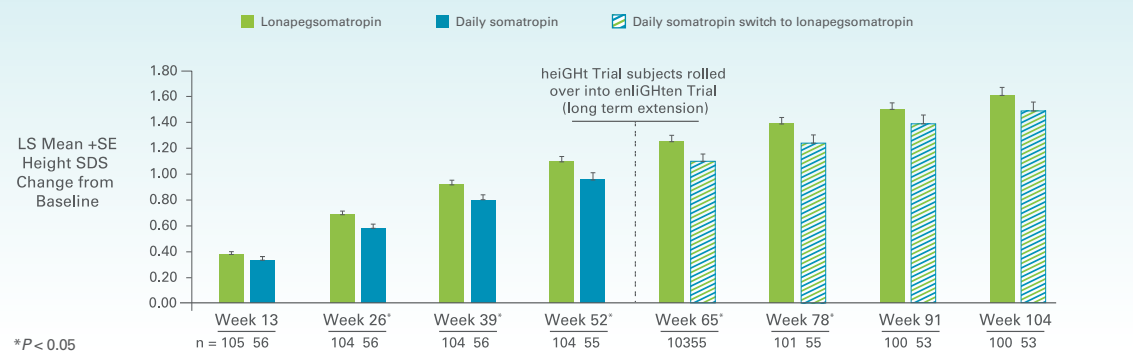
Lonapegsomatropin
Daily somatropin
Daily somatropin switch to lonapegsomatropin
heiGHt Trial subjects rolled over into enliGHten Trial (long term extension)
LS Mean +SE Height SDS Change from Baseline
Week 13, Week 26*, Week 39*, Week 52*, Week 65*, Week 78*, Week 91, Week 104
n = 105 56, 104 56, 104 56, 104 55, 10355, 101 55, 100 53, 100 53
*P < 0.05

- For heiGHt subjects, AHV increased in both groups to within the expected range for 2nd year therapy (Figure 4)
 - Among heiGHt subjects who switched from daily somatropin to lonapegsomatropin, a lower-than-expected attenuation in 2nd year AHV (as seen in the crossover of lines and decrease in slope in Figure 4) suggested an improved treatment effect of lonapegsomatropin relative to the previous daily somatropin

Figure 4. AHV over 104 Weeks for heiGHt subjects

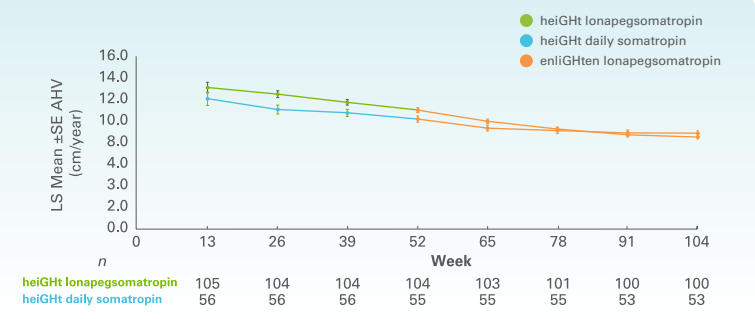
heiGHt lonapegsomatropin
heiGHt daily somatropin
enliGHten lonapegsomatropin
LS Mean ±SE AHV (cm/year)
Week
n
heiGHt lonapegsomatropin 105 104 104 104 103 101 100 100
heiGHt daily somatropin 56 56 56 55 55 55 53 53

- In heiGHt, average IGF-1 SDS values were higher for lonapegsomatropin-treated subjects compared with daily somatropin-treated subjects, paralleling the observed improved growth outcomes (Figure 5)
- Beyond 52 weeks, average IGF-1 SDS for heiGHt subjects who started on lonapegsomatropin generally remained stable without further increase; for heiGHt subjects who switched from daily somatropin to lonapegsomatropin, an initial increase in average IGF-1 SDS with subsequent stabilization was observed (Figure 5)

Figure 5. Average IGF-1 SDS over 104 Weeks for heiGHt subjects

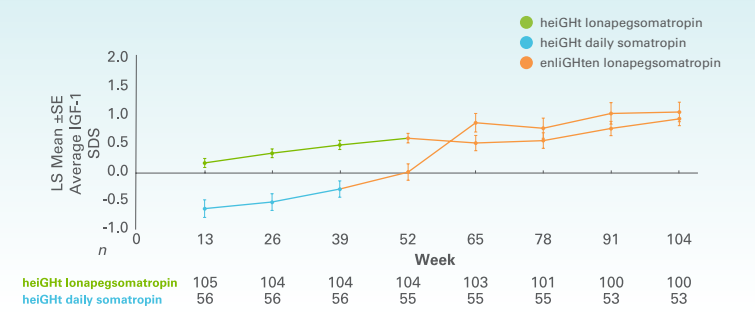
heiGHt lonapegsomatropin
heiGHt daily somatropin
enliGHten lonapegsomatropin
LS Mean ±SE Average IGF-1 SDS
Week
n
heiGHt lonapegsomatropin 105 104 104 104 103 101 100 100
heiGHt daily somatropin 56 56 56 55 55 55 53 53

Dose adjustments per investigator judgement were allowed in enliGHten.

- fliGHt subjects continued to approach their average parental height, with height SDS improving from –1.42 at fliGHt baseline to –0.69 at Week 78 (Figure 6); LS mean (SE) AHV at Week 78 was 8.3 cm/year and was consistent with clinical expectations given the characteristics of the enrolled subjects[5]

Figure 6. Sustained Improvement in Height SDS

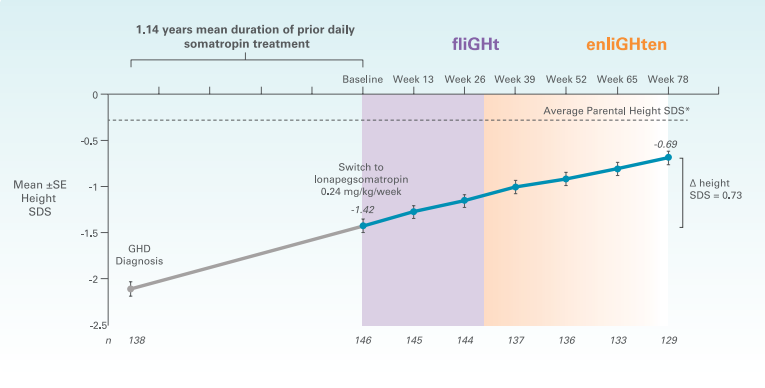
1.14 years mean duration of prior daily somatropin treatment
fliGHt
enliGHten
Baseline Week 13 Week 26 Week 39 Week 52 Week 65 Week 78
Average Parental Height SDS*
Mean ±SE Height SDS
Switch to lonapegsomatropin 0.24 mg/kg/week
-1.42
-0.69
Δ height SDS = 0.73
GHD Diagnosis
n 138 146 145 144 137 136 133 129

*Based on N=146 at fliGHt baseline.

- For fliGHt subjects, mean (SD) average IGF-1 SDS increased from 0.85 (1.3) at fliGHt baseline to 1.63 (1.3) at Week 26 and 1.81 (1.1) at Week 78

SAFETY OUTCOMES

- The adverse event (AE) profile of lonapegsomatropin was consistent across the phase 3 studies; treatment emergent AEs (TEAEs) were generally mild and no serious TEAEs were considered to be related to study drug (Table 4)
 - In enliGHten, the most common TEAEs were upper respiratory tract infection (21.1%), nasopharyngitis (11.1%), cough (8.7%), and pyrexia (8.4%); these are consistent with other clinical trials evaluating daily somatropin in children with GHD[6,7]
- A low titer non-neutralizing anti-drug binding antibodies was detected in < 10% of lonapegsomatropin-treated subjects

Table 4. Summary of Adverse Events Across All Trials

	heiGHt Trial (52 weeks)		fliGHt Trial (26 weeks)	enliGHten Trial (52 weeks)
Category, n (%)	Lonapegsomatropin (n = 105)	Daily somatropin (n = 56)	Lonapegsomatropin (n = 146)	Lonapegsomatropin (N = 298)
Treatment-emergent Adverse Events (TEAEs)	81 (77)	39 (70)	83 (57)	195 (65.4)
TEAEs Related to Study Drug	12 (11)	10 (18)	6 (4.1)	13 (4.4)
Serious Adverse Events (SAEs)	1 (1.0)	1 (1.8)	1 (0.7)	10 (3.4)
SAEs Related to Study Drug	0	0	0	0
TEAEs Leading to Any Action on Study Drug	2 (1.9)	1 (1.8)	2 (1.4)	5 (1.7)
TEAEs Leading to Discontinuation of Study Drug	0	0	0	0

- Hemoglobin A1c, cortisol, and free thyroxine were stable and generally remained within the normal range throughout the trials (Figure 7)

Figure 7. Laboratory Parameters Across All Trials

Figure 7A

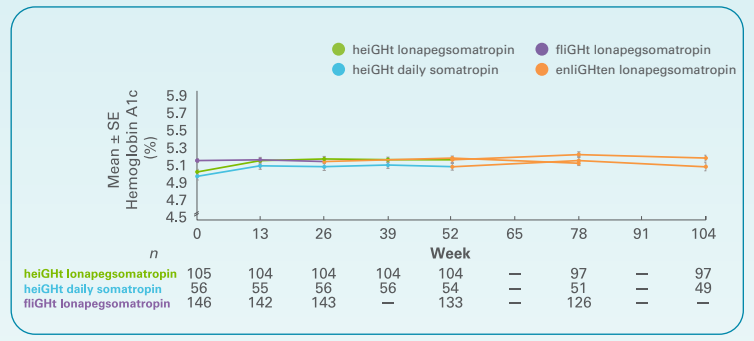
heiGHt lonapegsomatropin
heiGHt daily somatropin
fliGHt lonapegsomatropin
enliGHten lonapegsomatropin
Mean ± SE Hemoglobin A1c (%)
Week
n
heiGHt lonapegsomatropin 105 104 104 104 104 — 97 — 97
heiGHt daily somatropin 56 55 56 56 54 — 51 — 49
fliGHt lonapegsomatropin 146 142 143 — 133 — 126 — —

Figure 7B

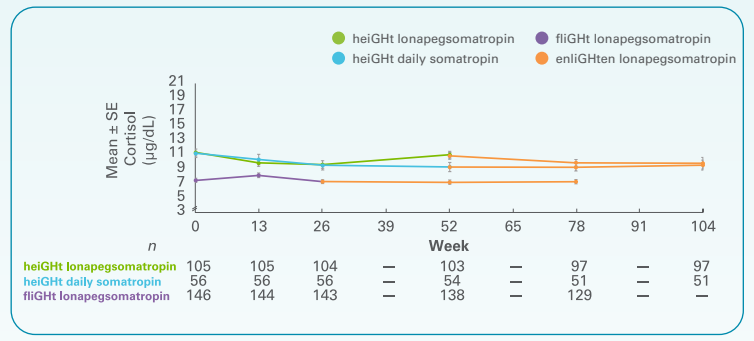
heiGHt lonapegsomatropin
heiGHt daily somatropin
fliGHt lonapegsomatropin
enliGHten lonapegsomatropin
Mean ± SE Cortisol (μg/dL)
Week
n
heiGHt lonapegsomatropin 105 105 104 — 103 — 97 — 97
heiGHt daily somatropin 56 56 56 — 54 — 51 — 51
fliGHt lonapegsomatropin 146 144 143 — 138 — 129 — —

Figure 7C

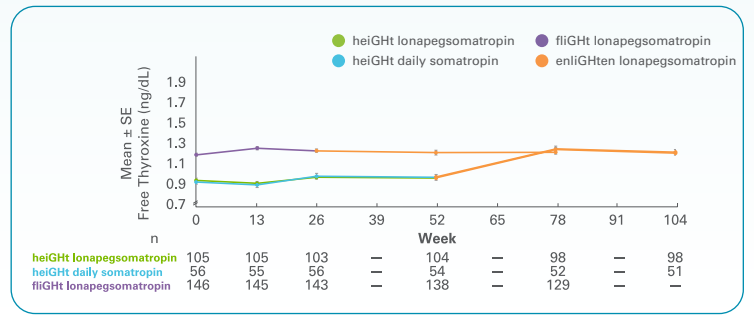
heiGHt lonapegsomatropin
heiGHt daily somatropin
fliGHt lonapegsomatropin
enliGHten lonapegsomatropin
Mean ± SE Free Thyroxine (ng/dL)
Week
n
heiGHt lonapegsomatropin 105 105 103 — 104 — 98 — 98
heiGHt daily somatropin 56 55 56 — 54 — 52 — 51
fliGHt lonapegsomatropin 146 145 143 — 138 — 129 — —

Central laboratory and assays used for laboratory parameters differed between trials.

- Over time, treatment with lonapegsomatropin was associated with mean BMI SDS that stabilized towards 0 (Figure 8)

Figure 8. BMI SDS Across All Trials

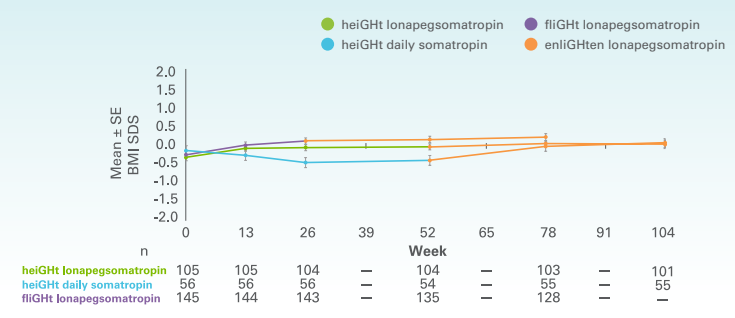
heiGHt lonapegsomatropin
heiGHt daily somatropin
fliGHt lonapegsomatropin
enliGHten lonapegsomatropin
Mean ± SE BMI SDS
Week
n
heiGHt lonapegsomatropin 105 105 104 — 104 — 103 — 101
heiGHt daily somatropin 56 56 56 — 54 — 55 — 55
fliGHt lonapegsomatropin 145 144 143 — 135 — 128 — —

SWITCHING TO THE TRANSCON HGH AUTO-INJECTOR

- Once available, subjects in enliGHten at select sites in the US were switched from vial and syringe to the TransCon hGH Auto-Injector; currently, 160 subjects are using the GH Auto-Injector in enliGHten
- As indicated by the Device Usability Questionnaire, the TransCon hGH Auto-Injector was generally found to be comfortable, easy-to-use, and safe (Figure 8)

Figure 8. Device Usability Questionnaire (DUQ) Ratings at Week 6 After TransCon hGH Auto-Injector Initiation (n = 139)

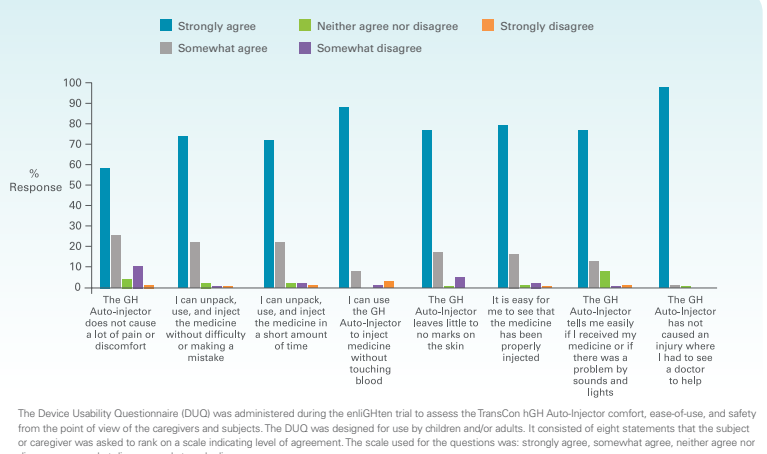
Strongly agree
Somewhat agree
Neither agree nor disagree
Somewhat disagree
Strongly disagree
% Response
The GH Auto-injector does not cause a lot of pain or discomfort
I can unpack, use, and inject the medicine without difficulty or making a mistake
I can unpack, use, and inject the medicine in a short amount of time
I can use the GH Auto-Injector to inject medicine without touching blood
The GH Auto-Injector leaves little to no marks on the skin
It is easy for me to see that the medicine has been properly injected
The GH Auto-Injector tells me easily if I received my medicine or if there was a problem by sounds and lights
The GH Auto-Injector has not caused an injury where I had to see a doctor to help

The Device Usability Questionnaire (DUQ) was administered during the enliGHten trial to assess the TransCon hGH Auto-Injector comfort, ease-of-use, and safety from the point of view of the caregivers and subjects. The DUQ was designed for use by children and/or adults. It consisted of eight statements that the subject or caregiver was asked to rank on a scale indicating level of agreement. The scale used for the questions was: strongly agree, somewhat agree, neither agree nor disagree, somewhat disagree and strongly disagree.

- Overall, fewer local tolerability reactions were reported with the TransCon hGH Auto-Injector compared with syringe/needle (Figure 9)

Figure 9. Local Tolerability from Subject Diary

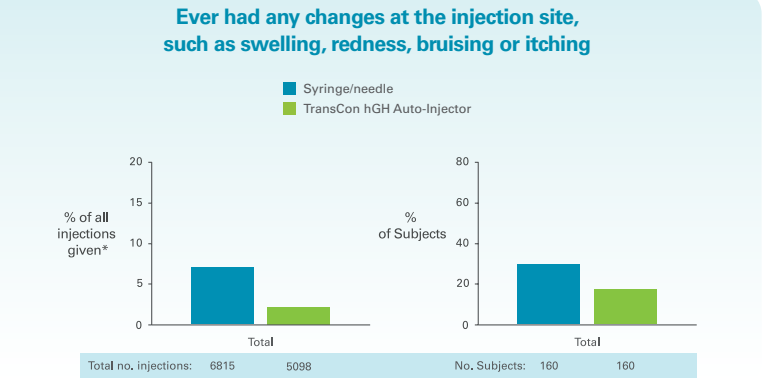
Ever had any changes at the injection site, such as swelling, redness, bruising or itching
Syringe/needle
TransCon hGH Auto-Injector
% of all injections given*
% of Subjects
Total
Total no. injections: 6815 5098
No. Subjects: 160 160

Local tolerability was defined as an injection site reaction deemed abnormal from those ordinarily observed in SC injections (including pain, intensity, or duration). Between visits, local tolerability will be evaluated and documented by the subject/parent/legal guardian/caregiver in the subject diary. At clinic visits, assessment of local tolerability was performed by injection site examination by trial staff (documented as part of the physical exam), in conjunction with subject diary review.

Left graph: Of all subjects who switched to the TransCon hGH Auto-Injector in enliGHten, the % of total injections given which were recorded in the patient diary as "ever had any changes at the injection site, such as swelling, redness, bruising or itching" is depicted. Overall, fewer injections given by the TransCon hGH Auto-Injector were rated as such.

Right graph: Of all subjects who switched from vial/syringe to the TransCon hGH Auto-Injector in enliGHten (n=160), the % of subjects who ever reported abnormal injection site reactions (i.e., "ever had any changes" etc) in the patient diary is depicted. Overall, fewer subjects responded that injections given by the TransCon hGH Auto-Injector caused local tolerability reactions.

CONCLUSIONS

- Across the broad population of the phase 3 program, subjects treated with lonapegsomatropin for up to 2 years continued to grow well, with a safety profile comparable to daily somatropin, including a similar AE profile, stable BMI, stable laboratory parameters, and low immunogenicity
- Among subjects who switched from daily somatropin to lonapegsomatropin, a lower-than-expected attenuation in 2nd year AHV suggested an improved treatment effect of lonapegsomatropin relative to the previous daily somatropin
- High retention rates were observed across the phase 3 program, with > 98% of subjects continuing from heiGHt and fliGHt into enliGHten
- In enliGHten, where doses could be adjusted based on investigator judgement, mean doses were at or just below 0.24 hGH mg/kg/week, indicating no evidence of higher doses required over time
- The TransCon hGH Auto-Injector was found to be well-tolerated and easy-to-use

REFERENCES:

1. Sprogøe K et al. *Endocr Connect,* 2017;6(8):R171-R181. 2. Chatelain P et al. *J Clin Endocrinol Metab,* 2017;102(5):1673-1682. 3. Thornton P et al. *Transcon Growth Hormone In The Treatment Of Pediatric Growth Hormone Deficiency: Results Of The Phase 3 Height Trial.* Presented at ENDO March 2019; New Orleans, LA. 4. Maniatis A et al. *Phase 3 fliGHt Trial: Experience of Switching from Daily Growth Hormone Therapy to Once-Weekly TransCon hGH in Children with Growth Hormone Deficiency.* Presented at ENDO June 2020. 5. Bakker B et al. *J Clin Endocrinol Metab.* 2008;93(2):352-357. 6. Cohen GM et al. *J Clin Endocrinol Metab.* 2002;87(1):90-98. 7. Peterkova V et al. *Horm Res.* 2007;68(6): 288-293.

This study was sponsored by Ascendis Pharma Endocrinology Division A/S.

All product candidates are investigational. Not for use in promotion or product commercialization.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group. © March 2021 Ascendis Pharma A/S