Pediatric Phase 2 Data Demonstrate That TransCon hGH Has An Anti-hGH Immunogenic Profile That is Comparable to Daily hGH

David Gilfoyle[1], PhD; Susanne Pihl[1]; Pierre Chatelain[2], MD; Michael Beckert[1], MD

[1]Ascendis Pharma A/S, Hellerup, Denmark; [2]Div. Endocrinologie Pediatrique – Hopital Mere-Enfant, University Claude Bernard Lyon 1, Lyon, France

This study was sponsored by Ascendis Pharma A/S

Background

TransCon Growth Hormone is a sustained-release prodrug of recombinant human Growth Hormone (hGH) that releases fully active, unmodified hGH into the blood compartment (Figure 1).

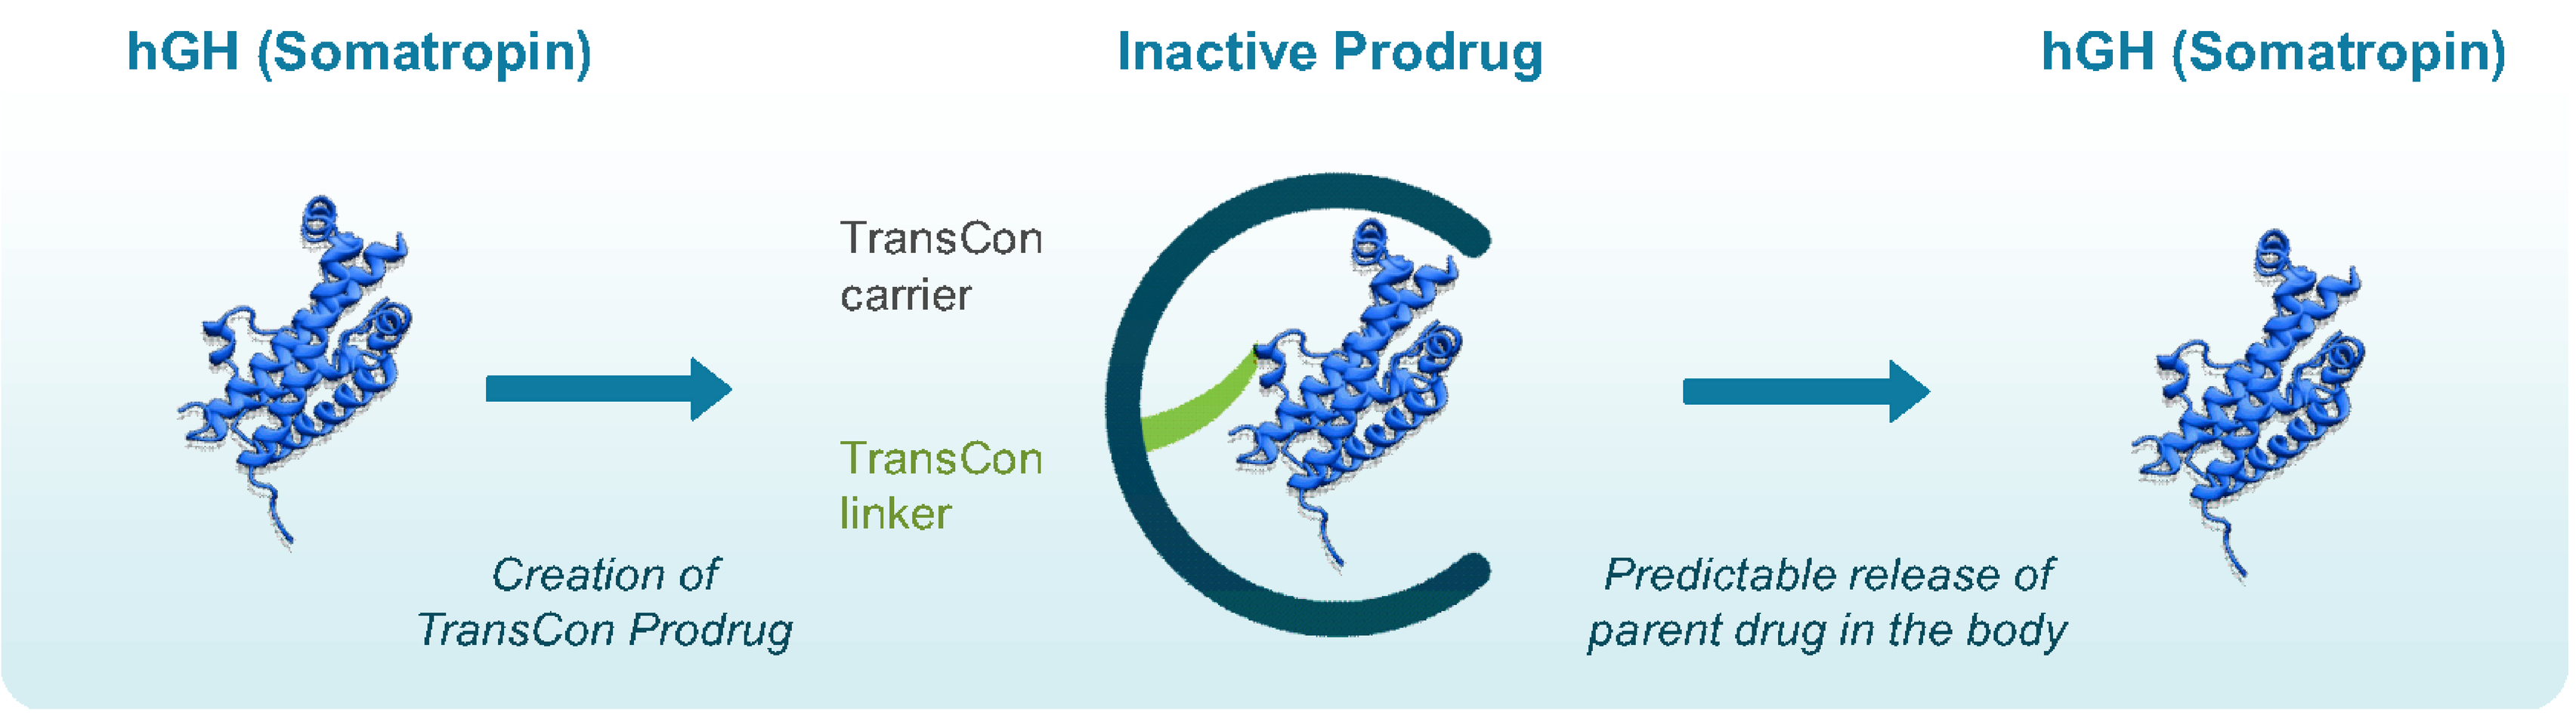
hGH (Somatropin)
Inactive Prodrug
hGH (Somatropin)
TransCon carrier
TransCon linker
Creation of TransCon Prodrug
Predictable release of parent drug in the body

Figure 1: The TransCon hGH prodrug consists of hGH transiently bound to a polyethylene glycol carrier via a TransCon linker. The released hGH is unmodifed, and designed to maintain the same mode of action and distribution in the body as daily hGH.

Daily administered hGH replacement therapy:

- Well tolerated, without significant anti-hGH immunogenicity
- Low titer, non-neutralizing anti-hGH antibodies detected in 8% (7 of 87) of pediatric Growth Hormone Deficient (GHD) subjects treated with Genotropin [1]
- Patients with neutralizing anti-hGH antibodies, may not respond to hGH therapy, and may require long-term IGF-I therapy to facilitate growth
- New sustained-release therapies should maintain a comparable safety profile

TransCon hGH has been shown in a Phase 2 study in GHD children, to be safe and well tolerated, demonstrating an anti-hGH immunogenicity profile comparable to that reported for daily administered hGH.

Objectives

While permanent conjugation of carrier molecules to protein therapeutics has the potential to reduce immunogenicity through epitope shielding [2], the protein-carrier interface can also elicit unwanted immunogenicity [3].

TransCon hGH is designed to leverage the inherent low immunogenicity of recombinant hGH:

- In the prodrug form, the carrier shields both the protein and the protein-carrier interface
- Following release of unmodified hGH, the potentially immunogenic protein-carrier interface is removed

Sensitive anti-hGH binding and neutralizing antibody assays have been developed, validated and utilized to assess anti-hGH immunogenicity in a Phase 2 clinical study of GHD children and consequently support the TransCon hGH product concept.

Clinical Study Design

Pre-pubertal, treatment-naïve, children with GHD (53 treated patients) received:

- Weekly s.c. injections of TransCon hGH (0.14, 0.21 or 0.30 mg hGH/kg/week [n=12, 14 and 14, respectively]) or
- Daily s.c. injections of Genotropin® (0.21 mg hGH/kg/week [n=13])
- Six-month treatment period

Serum samples were collected and assessed for anti-hGH binding and, if appropriate, neutralizing antibodies at:

- Screening, pre-dose at Weeks 1, 5 and 13 and 1 week after the last dose (Week 27)

References:
[1] Assessment Report, Somatropin Biopartners, European Medicines Agency, EMA/CHMP/229458/2013, 30 May 2013
[2] FDA Guidance for Industry, Immunogenicity Assessment for Therapeutic Protein Products, August 2014
[3] Immunogenicity of Biopharmaceuticals, Weert, M. and Moller, E. H. (Eds.) ISBN: 978-0-387-75840-4 (2008)
[4] FDA Guidance for Industry, Assay Development for Immunogenicity Testing of Therapeutic Proteins, December 2009 (Draft Guidance)
[5] EMEA/CHMP Guideline on Immunogenicity Assessment of Biotechnology-Derived Therapeutic Proteins, April 2008
[6] Mire-Sluis, A. R. et al, J Immunol Methods, 289, 1 – 16 (2004)
[7] Shankar, G. et al, J. Pharm Biomed Anal, 48 (5), 1267-1281 (2008)
[8] Gupta, S. et al, J. Pharm Biomed Anal, 55 (5), 878-888 (2011)

Immunogenicity Assessment

The potential presence of anti-hGH antibodies were assessed in serum using assays developed and validated in accordance with appropriate regulatory and industry guidelines [4 – 8, inclusive]:

Assay Parameter	Binding Assay	Neutralizing Assay
Assay Format	Bridging ELISA	Cell-based (Nb2-11) proliferation assay
Antibody Evaluation Stages	Screening, confirmation and titration (as appropriate)	Screening and confirmation (as appropriate)
Confirmation Principle	Immunodepletion with hGH	Alternative stimulus with prolactin
Isotype Detection	Any hGH-specific Ig isotype based on bridging assay format	IgG, IgM and IgA
Assay Cut-point Determination	Plate specific cut-point derived from normalization factors which assumed a 5% false positive rate (screening assay) based on assessment of 50 male and female children with GHD	
Sensitivity	< 500 ng/mL (based on a control antibody)	
Interference	Hemolysed and lipemic serum assessed	
Drug Tolerance	Assessed for TransCon hGH and hGH - confirmed acceptable at typical C_{trough} concentrations in the presence of currently observed treatment-emergent responses	

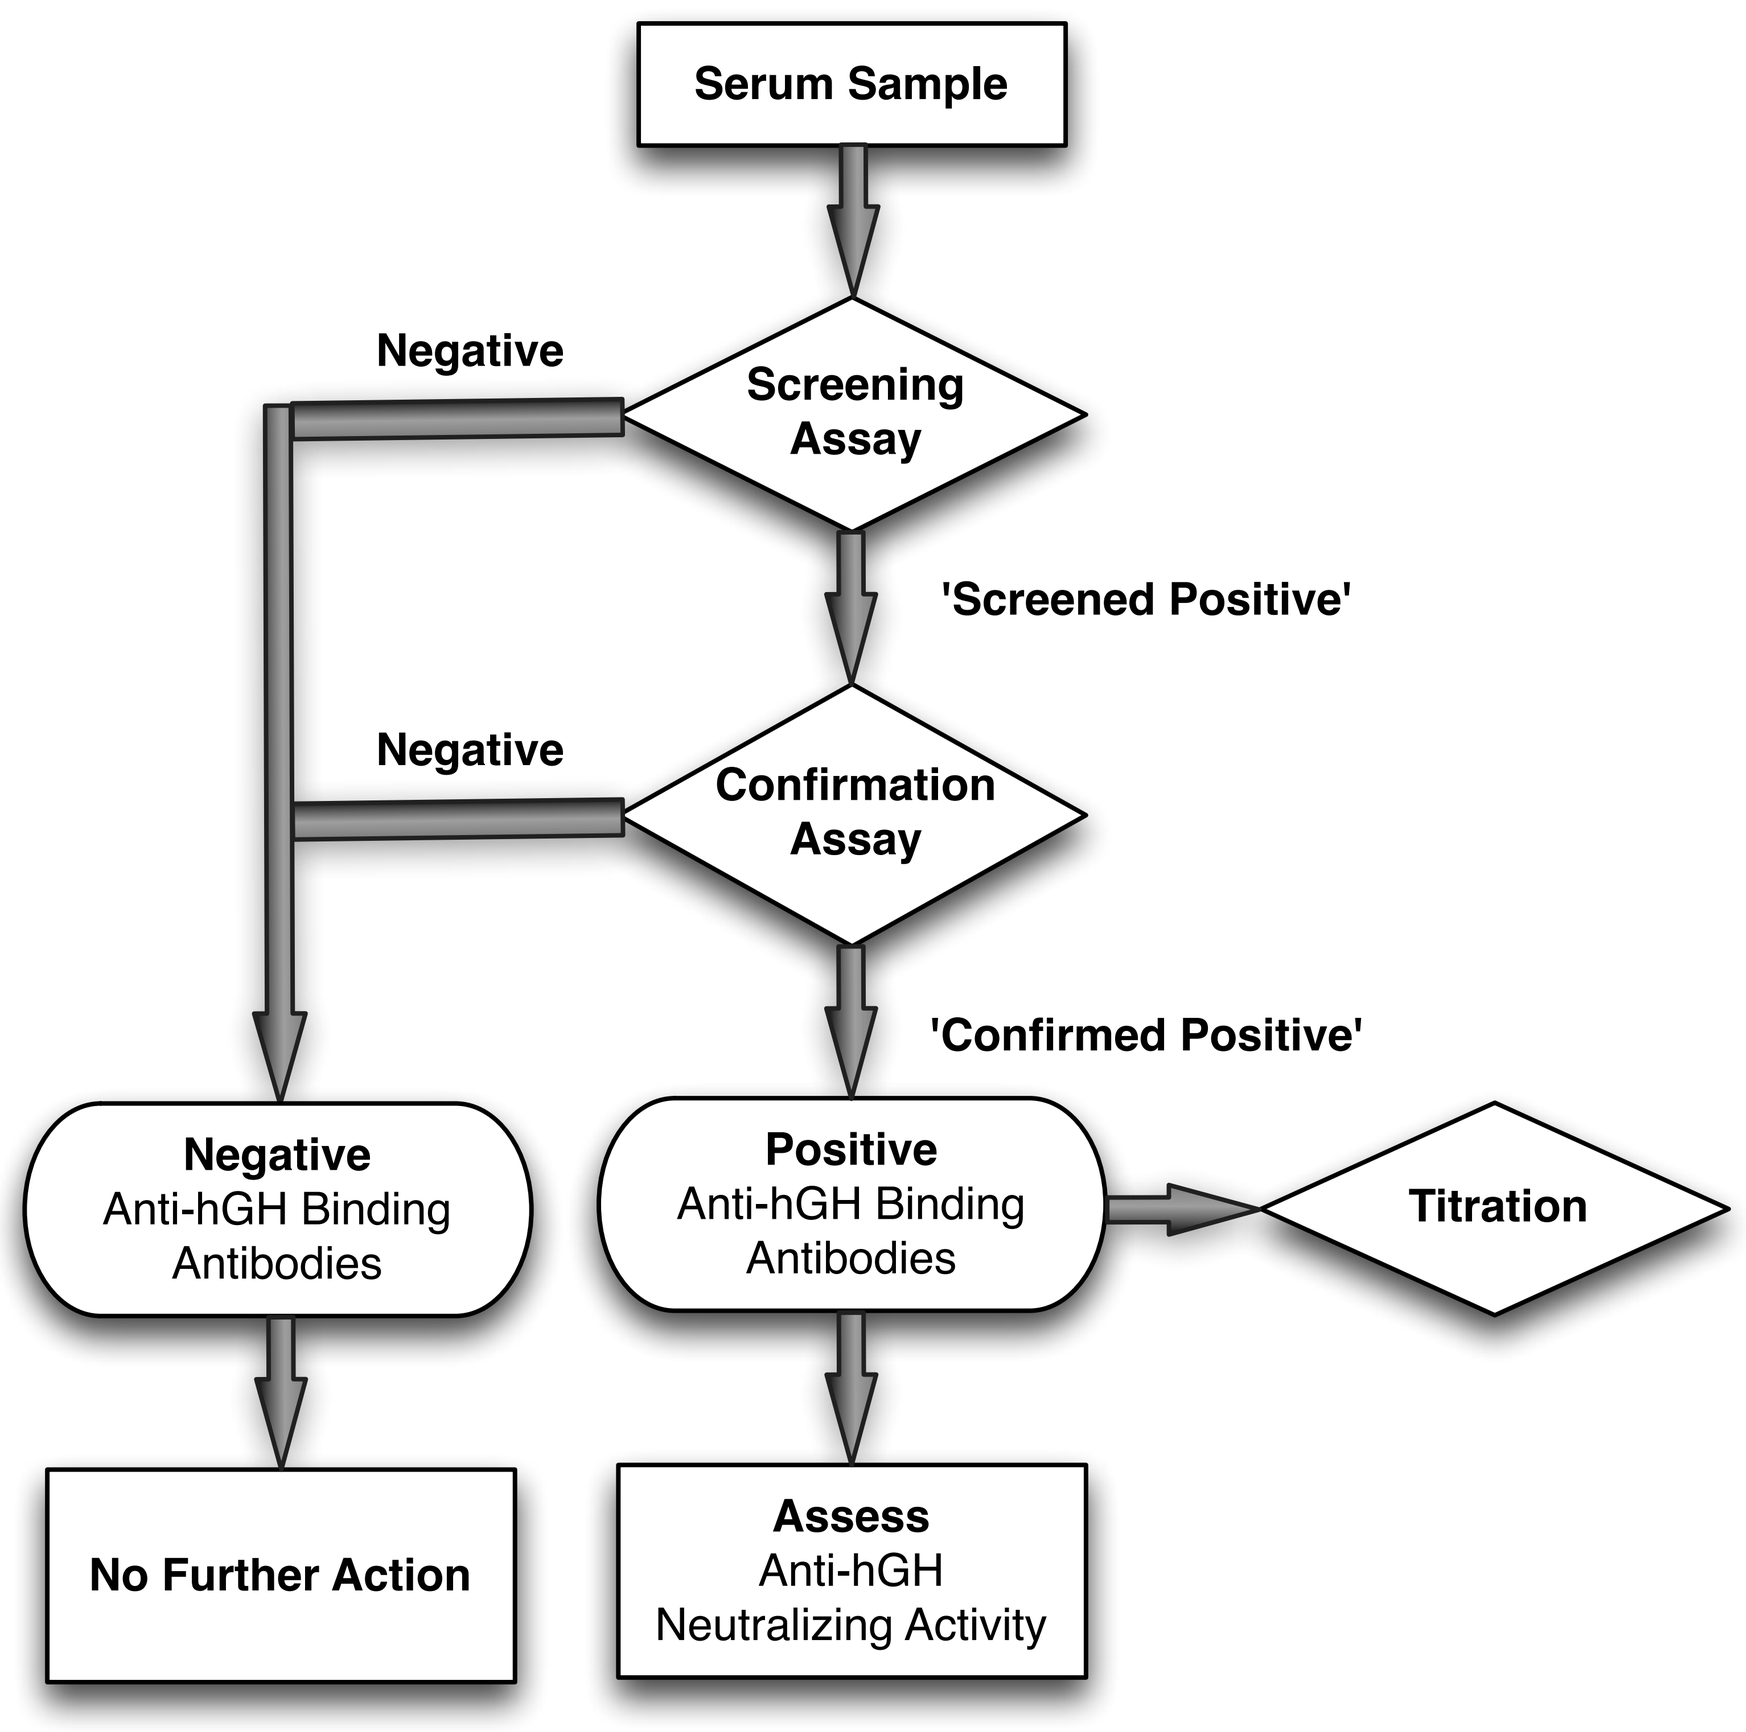
Serum Sample
Screening Assay
Negative
'Screened Positive'
Confirmation Assay
Negative
'Confirmed Positive'
Negative Anti-hGH Binding Antibodies
Positive Anti-hGH Binding Antibodies
Titration
No Further Action
Assess Anti-hGH Neutralizing Activity

Serum samples were assessed in a multiple stage process initially to detect anti-hGH binding antibodies (refer to left figure). Samples confirmed positive for anti-hGH binding antibodies, and with an appropriate assay response were titered by dilution with negative control serum until the assay response fell below the screening assay cut-point.

All samples confirmed positive for anti-hGH binding antibodies were assessed for anti-hGH neutralizing antibody activity.

Results – Immunogenicity

One subject (0.14 mg hGH/kg/week; 2.5% of subjects administered TransCon hGH [n=40]) developed a treatment-emergent anti-hGH immune response:

- Detected initially at Week 13
- Titration at Week 27 indicated the presence of very low levels of anti-hGH binding antibodies
- Antibodies confirmed to be non-neutralizing
- Drug levels at Week 27 were confirmed to be below levels considered to significantly interfere with the antibody assays

The presence of anti-hGH antibodies did not appear to impact:

- Pharmacokinetic (TransCon hGH and hGH) or pharamcodynamic (IGF-I) profiles compared to antibody negative subjects
- Annualized height velocity (19.0 cm, in the upper half of the treatment cohort)

Conclusion

TransCon hGH has demonstrated an immunogenicity profile in a pedatric population comparable to that observed with daily administered hGH:

- Confirms immunogenicity data for TransCon hGH in two Phase 1 clinical studies in healthy volunteers and a Phase 2 clinical study in adults with GHD (AGHD)
- Detection of anti-hGH binding antibodies at a frequency comparable to that observed for Genotropin® indicates that analytical methodology are fit for purpose to support future clinical development of TransCon hGH

Based on the promising clinical results, the global Phase 3 heiGHt Trial in GHD children has been initiated in mid-2016.

Disclosure Statement (Conflicts of Interest): Authors marked 1 above are employees of Ascendis Pharma A/S. The author marked 2 above was the Coordinating Investigator for Ascendis Clinical Study ACP-001_CT-004.