The Pivotal Phase 3 heiGHt Trial of Weekly TransCon™ hGH vs. Daily hGH in Treatment-Naïve Subjects with Pediatric Growth Hormone Deficiency

Aristides K. Maniatis[1], Paul Thornton[2], Paul Hofman[3], Elena Aghajanova[4], Elena Chertok[5], Maria Korpal-Szczyrska[6], Elene Giorgadze[7], Tatiana Kovalenko[8], Aimee D. Shu[9], David B. Karpf[9], Wenjie Song[9], Michael Beckert[9], Jonathan A. Leff[9], and the heiGHt Trial Investigators

[1]Rocky Mountain Pediatric Endocrinology, [2]Cook Children's, [3]University of Auckland, [4]Yerevan State Medical University, [5]Voronezh State Medical University, [6]Medical University of Gdansk, [7]LTD National Institute of Endocrinology, [8]Republican Children's Clinical Hospital of the Ministry of Health of the Udmurt Republic, [9]Ascendis Pharma, Inc.

BACKGROUND

TransCon™ hGH is a long-acting prodrug in development for patients with growth hormone deficiency (GHD). TransCon™ hGH consists of a parent drug, hGH, that is transiently bound to a carrier via a proprietary linker.[1] The carrier extends hGH circulation time in the body through a shielding effect that minimizes renal excretion and receptor binding (Figure 1). Over one week, TransCon™ hGH releases fully active, unmodified hGH via autohydrolosis of the TransCon™ linker at physiologic pH and temperature. TransCon™ hGH is designed to maintain the same mode of action and distribution as daily hGH, but with once-weekly dosing.[2]

Figure 1: TransCon™ Growth Hormone Releases Unmodified hGH

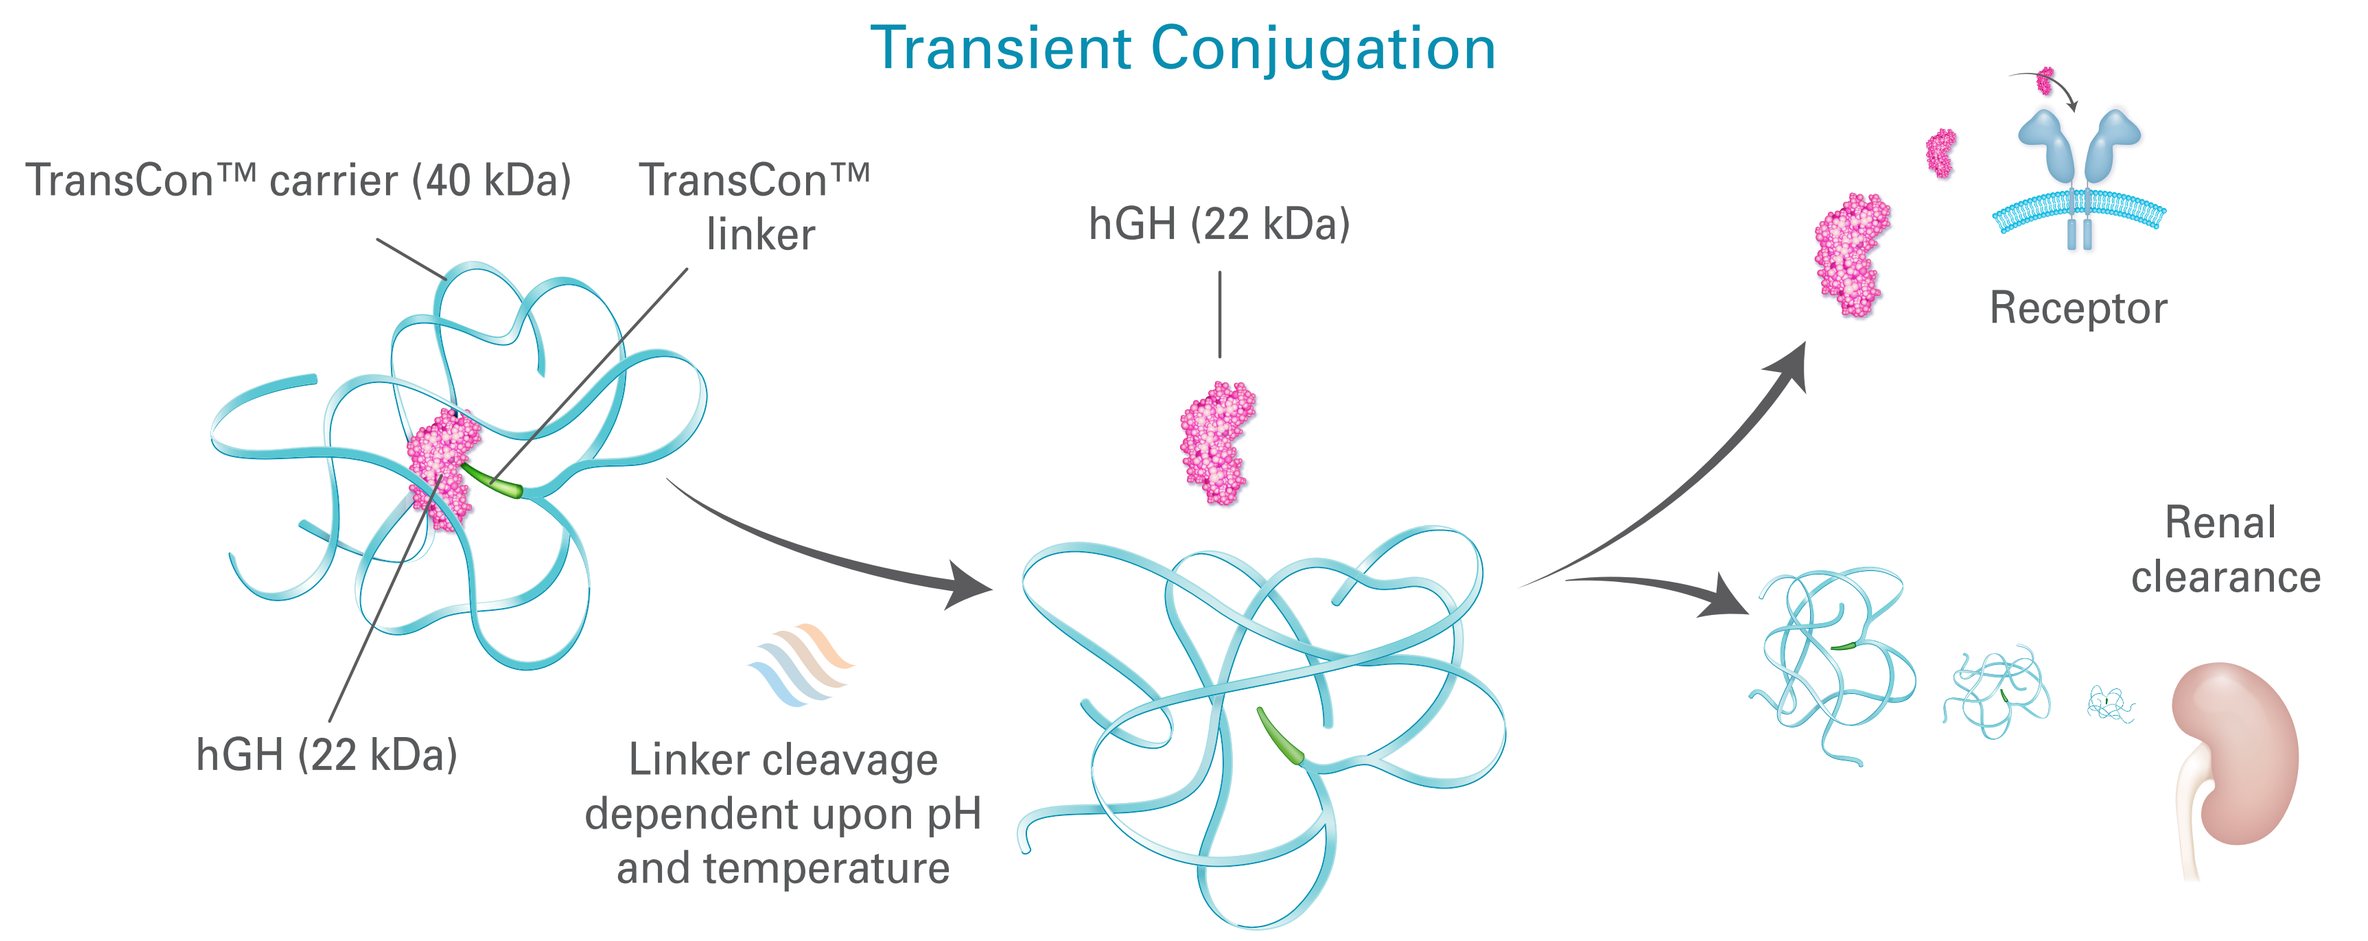
Transient Conjugation
TransCon™ carrier (40 kDa)
TransCon™ linker
hGH (22 kDa)
hGH (22 kDa)
Linker cleavage dependent upon pH and temperature
Receptor
Renal clearance

Parent drug is transiently bound to a TransCon™ linker-soluble carrier moiety, which inactivates and shields parent drug from clearance

Following injection, the linker is designed to autohydrolyze at specific rates to predictably release unmodified parent drug

Designed to distribute parent drug like the endogenous compound; linker-carrier is cleared renally

TransCon™ hGH is a long-acting prodrug consisting of parent drug, unmodified hGH, transiently bound to a carrier, mPEG (40 kDa), via a proprietary TransCon™ linker. Based on physiologic pH and temperature, the TransCon™ linker autohydrolyzes following first-order kinetics, releasing fully active, unmodified hGH over a one-week period designed to allow the same tissue distribution and receptor activation as endogenous GH. The release of hGH liberates the inert carrier, allowing elimination from the body. The carrier and linker are cleared primarily by renal filtration and to a minor extent by hepatobiliary excretion.

Once-weekly prodrug designed to release unmodified hGH and mimic daily hGH:

- Tissue distribution
- Physiological levels
- Therapeutic effects: safety, efficacy, and tolerability

OBJECTIVES

The primary objective of the heiGHt Trial was to demonstrate non-inferiority in annualized height velocity (AHV) of prepubertal children with GHD after 52 weeks of treatment with once-weekly TransCon™ hGH versus daily Genotropin® (Pfizer, NY).

Key secondary objectives included:

- Evaluation of the safety of weekly TransCon™ hGH administered over 52 weeks compared to daily Genotropin®
- Evaluation and comparison of the change in height Standard Deviation Score (SDS) over 52 weeks of weekly TransCon™ hGH compared to daily Genotropin®
- Evaluation of serum IGF-1 over 52 weeks in weekly TransCon™ hGH or daily Genotropin®
- Description of the pharmacodynamic profile of IGF-1 over 168 hours (7 days) following a weekly administration

METHODS

- heiGHt was a global, pivotal, phase 3 randomized, open-label, active-controlled trial comparing once-weekly TransCon™ hGH to daily Genotropin® at equivalent doses (Figure 2)
- Males and females (aged 3-12 or 3-11 years, respectively) in Tanner stage 1 diagnosed with isolated GHD (or as part of multiple pituitary deficiency on stable replacement therapy) via 2 different GH stimulation tests (peak GH ≤10 ng/mL) were eligible
- Subjects deemed eligible for the trial were randomly assigned to one of two treatment groups, either TransCon™ hGH 0.24 mg/kg/wk administered subcutaneously (SC) once weekly or daily Genotropin® administered SC once per day in a dose equivalent to 0.24 mg/kg/wk (ie, 0.034 mg/kg/d) for 52 weeks
- Randomization (2:1, TransCon™ hGH to Genotropin®) was stratified by age (≤ 6 years vs > 6 years), gender, and peak stimulated GH level (≤ 5 ng/mL vs > 5 ng/mL)
- TransCon™ hGH and Genotropin® were administered to the thigh, buttocks or abdomen in a rotating fashion
- Subjects were treated and followed for 52 weeks
- Topline results are reported

Figure 2: Phase 3 heiGHt Trial Design

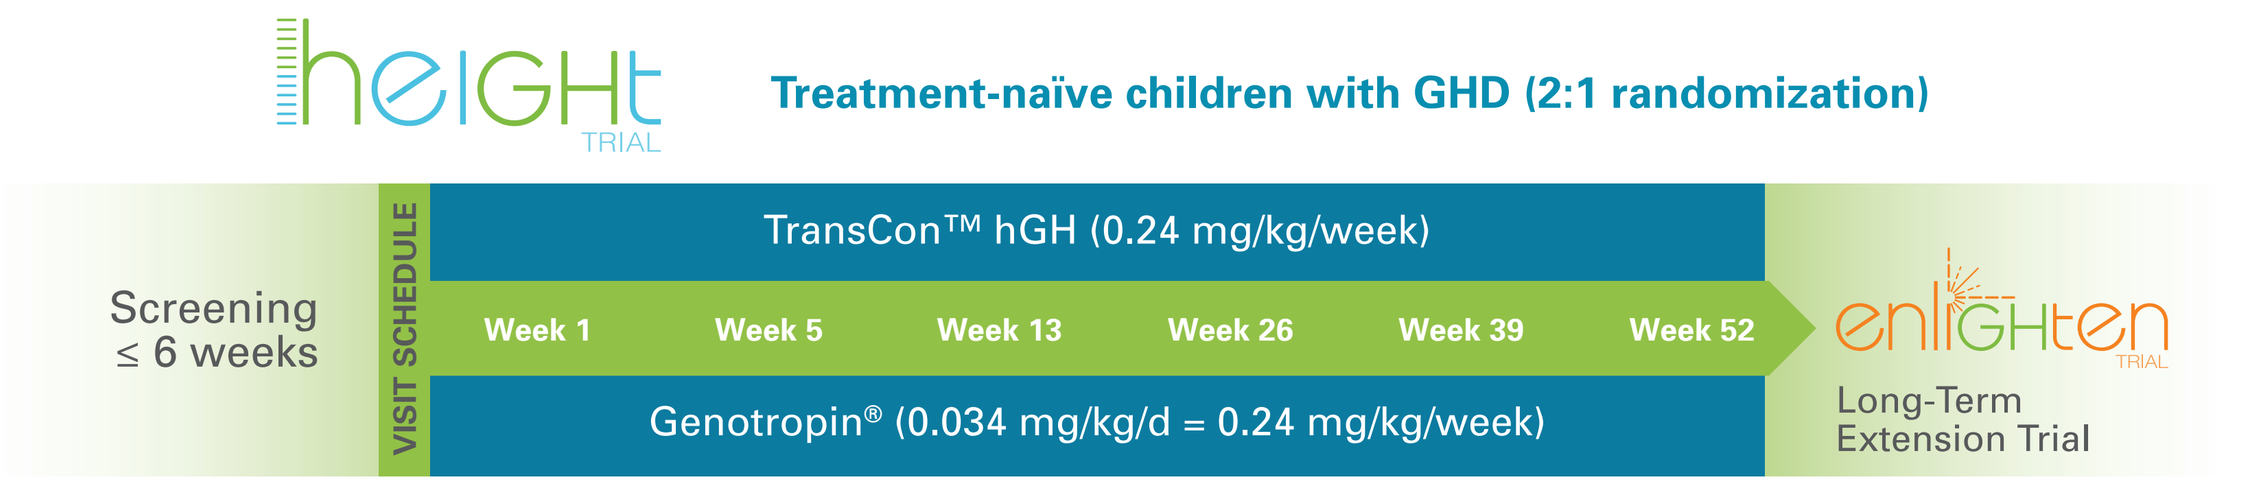
heiGHt TRIAL
Treatment-naïve children with GHD (2:1 randomization)
Screening ≤ 6 weeks
VISIT SCHEDULE
TransCon™ hGH (0.24 mg/kg/week)
Week 1
Week 5
Week 13
Week 26
Week 39
Week 52
Genotropin® (0.034 mg/kg/d = 0.24 mg/kg/week)
enliGHten TRIAL
Long-Term Extension Trial

Key Inclusion Criteria

- Prepubertal children with GHD
- Height SDS ≤ -2.0
- IGF-1 SDS ≤ -1.0
- 2 GH stimulation tests (GH ≤ 10 ng/mL)
- Bone age ≥ 6 months behind chronological

Objective:

- Demonstrate non-inferiority

Key Endpoints

- Annualized Height Velocity (AHV) at 52 weeks (primary endpoint)
- AHV at earlier time points
- Change in HT SDS over 52 weeks
- Change in serum IGF-1 and IGFBP-3 levels
- Change in IGF-1 SDS and IGFBP-3 SDS
- Normalization of IGF-1 SDS
- hGH and IGF-1 levels over 168 hours at week 13 (PK/PD subset)

RESULTS

Table 1: Demographics and Baseline Characteristics

	TransCon™ hGH (n=105) Mean	Genotropin® (n=56) Mean	Total (N=161) Mean
Chronological Age (CA)(years)	8.51	8.48	8.50
Bone Age (BA)(years)	5.84	5.98	5.88
BA/CA	0.69	0.70	0.69
Height SDS	-2.89	-3.00	-2.93
BMI	16.06	16.46	16.20
BMI SDS	-0.32	-0.14	-0.25
Delta Mid-Parental Height SDS	-2.32	-2.55	-2.40
IGF-1 SDS	-2.08	-1.96	-2.04
Peak GH (ng/mL)	5.89	5.48	5.75
Caucasian (%)	95.2	92.9	94.4
Male (%)	81.9	82.1	82.0

A total of 161 subjects from 54 sites were randomized and dosed. In the intention-to-treat population, 159/161 (98.8%) completed the trial, while 2 subjects withdrew. One of 105 (1.0%) in the TransCon™ hGH cohort grew fatigued with the medical system following unrelated acute appendicitis and an appendectomy, while 1/56 (1.8%) in the daily Genotropin® cohort was lost to follow-up prior to the final visit. The cohorts were well balanced with respect to demographics and baseline characteristics across all parameters.

RESULTS

Table 2: TransCon™ hGH Met its Primary Endpoint of Non-inferiority and Was Superior to Genotropin® in AHV at Week 52

	TransCon™ hGH (n=105)	Genotropin® (n=56)	Estimate of Treatment Difference	*P*-value
LS Mean AHV at Week 52 (cm/year)	11.2	10.3	0.86	0.0088
Standard Error	0.23	0.30	0.33	
95% Confidence Interval (cm/year)	10.71 – 11.62	9.73 – 10.89	0.22 – 1.50	

LS Mean=Least Squares Mean; ANCOVA model was applied after missing data were imputed by multiple imputation method.

At Week 52, the AHV for TransCon™ hGH was 11.2 cm while that for Genotropin® was 10.3 cm. The treatment difference was 0.86 cm in favor of TransCon™ hGH with a 95% confidence interval of 0.22 to 1.50, which demonstrates not only non-inferiority but also superiority (*p*=0.0088) over Genotropin®. Further analysis of the AHV revealed that the treatment difference reached statistical significance at Weeks 26 (p=0.0017) (not shown), 39 (p=0.0061) (not shown) and 52 (p=0.0088).

Figure 3: Schematic of Possible Outcomes of the heiGHt Trial

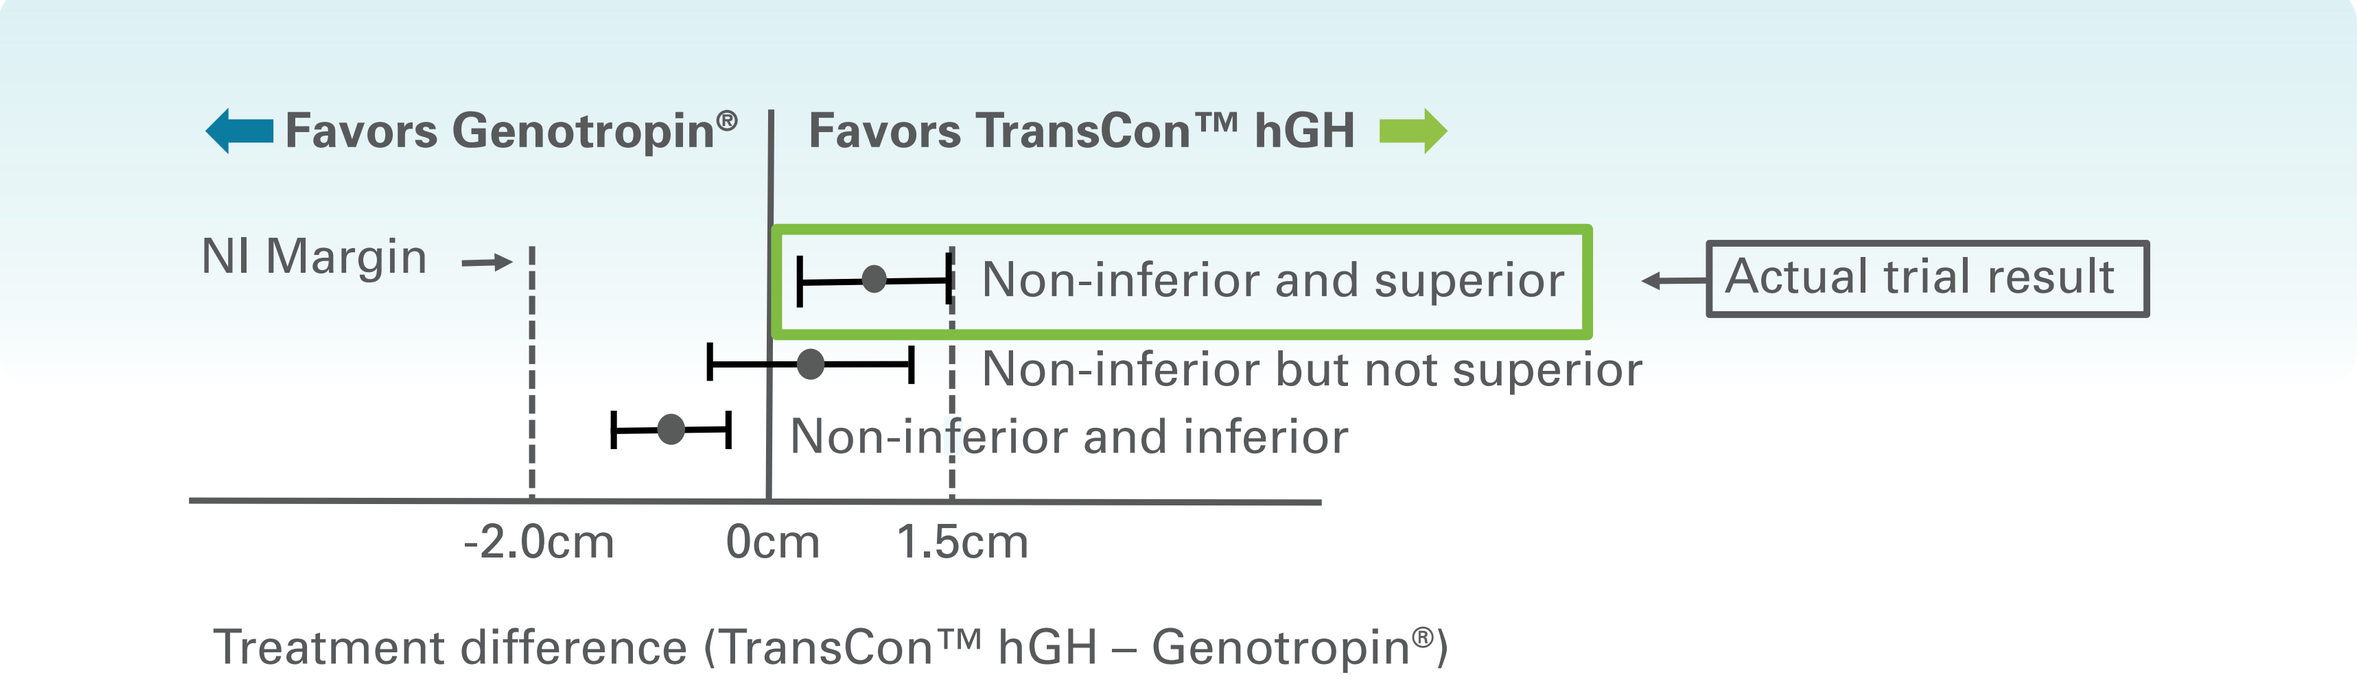
Favors Genotropin®
Favors TransCon™ hGH
NI Margin
Non-inferior and superior
Actual trial result
Non-inferior but not superior
Non-inferior and inferior
-2.0cm
0cm
1.5cm
Treatment difference (TransCon™ hGH – Genotropin®)

Table 3: Numerically Lower Incidence of Poor Responders with TransCon™ hGH

Poor responders defined as AHV < 8.0 cm/year[3]

At Week 52	TransCon™ hGH (n=104)* n (%)	Genotropin® (n=55)* n (%)
Responder	100 (96.2)	49 (89.1)
Poor Responder	4 (3.8)	6 (10.9)

*Excludes one subject/group with missing Week 52 data (98.8% subjects completed study).

In a post-hoc analysis, the proportion of responders in each group was evaluated, defined as achieving at least 8.0 cm/year in AHV. Of those subjects administered TransCon™ hGH, 4/104 (3.8%) were poor responders as compared to 6/55 (10.9%) administered daily Genotropin®.

Figure 4: Change in Height SDS Over 52 Weeks for Equivalent Doses of TransCon™ hGH and Daily Genotropin®

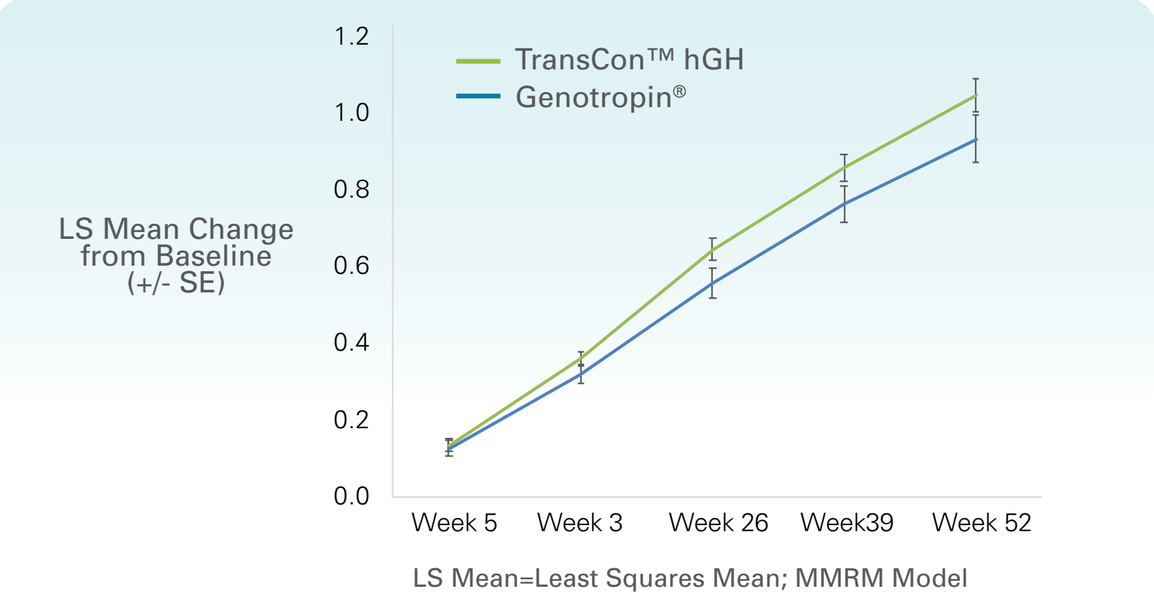
TransCon™ hGH
Genotropin®
LS Mean Change from Baseline (+/- SE)
1.2
1.0
0.8
0.6
0.4
0.2
0.0
Week 5
Week 3
Week 26
Week39
Week 52
LS Mean=Least Squares Mean; MMRM Model

A comparison of TransCon™ hGH vs. daily Genotropin® height SDS at Weeks 1, 13, 26, 39, and 52 showed a similar trend as AHV, with the difference in mean height SDS difference increasing throughout the trial. MMRM=Mixed Model Repeated Measures.

Figure 5: IGF-1 Profile Over 1 Week of Testing at Week 13 (n=11)

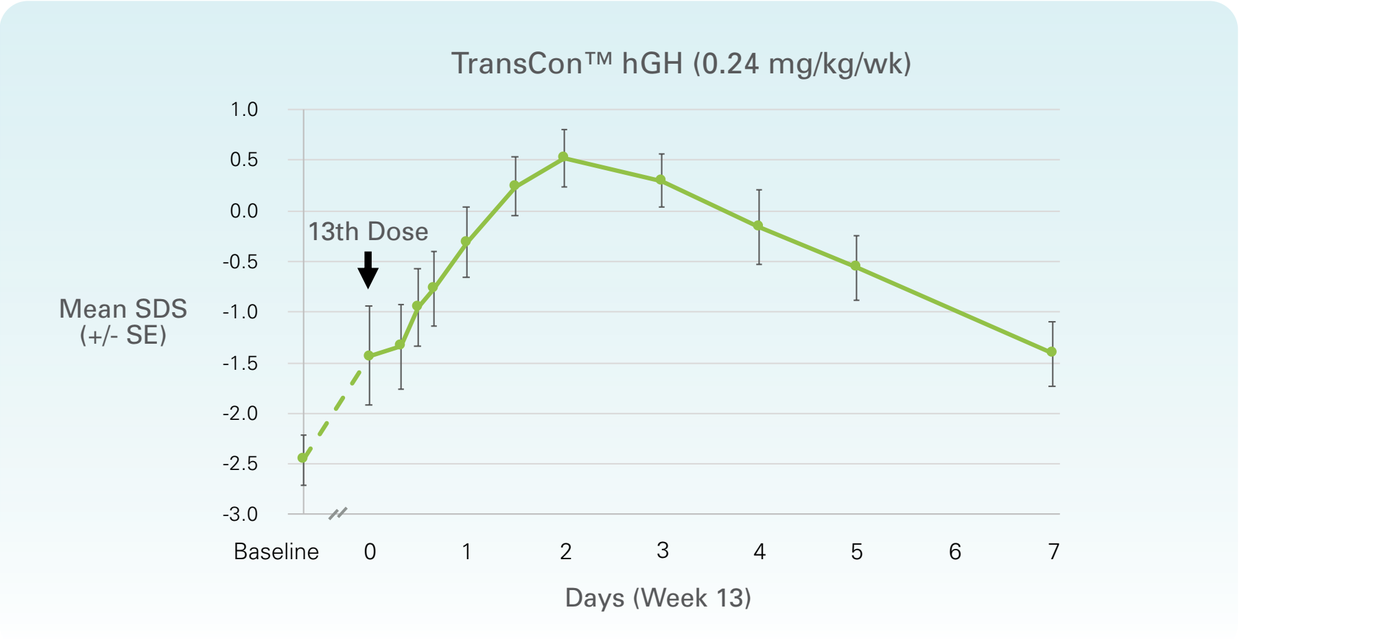
TransCon™ hGH (0.24 mg/kg/wk)
Mean SDS (+/- SE)
13th Dose
1.0
0.5
0.0
-0.5
-1.0
-1.5
-2.0
-2.5
-3.0
Baseline
0
1
2
3
4
5
6
7
Days (Week 13)

In the subset of subjects (n=11) with intensive PK and PD monitoring during Week 13, results were similar to those reported in the TransCon™ hGH pediatric phase 2 trial (Chatelain, 2017). Mean peak IGF-1 SDS was achieved on Day 2 post-dose. At the end of the week, IGF-1 SDS returned to steady-state baseline (Hours recorded = baseline, 0, 8, 12, 16, 24, 36, 48, 72, 96, 120, 168).

Figure 6: IGF-1 SDS Over 52 Weeks for Equivalent Doses of TransCon™ hGH and Daily Genotropin® (N=161)

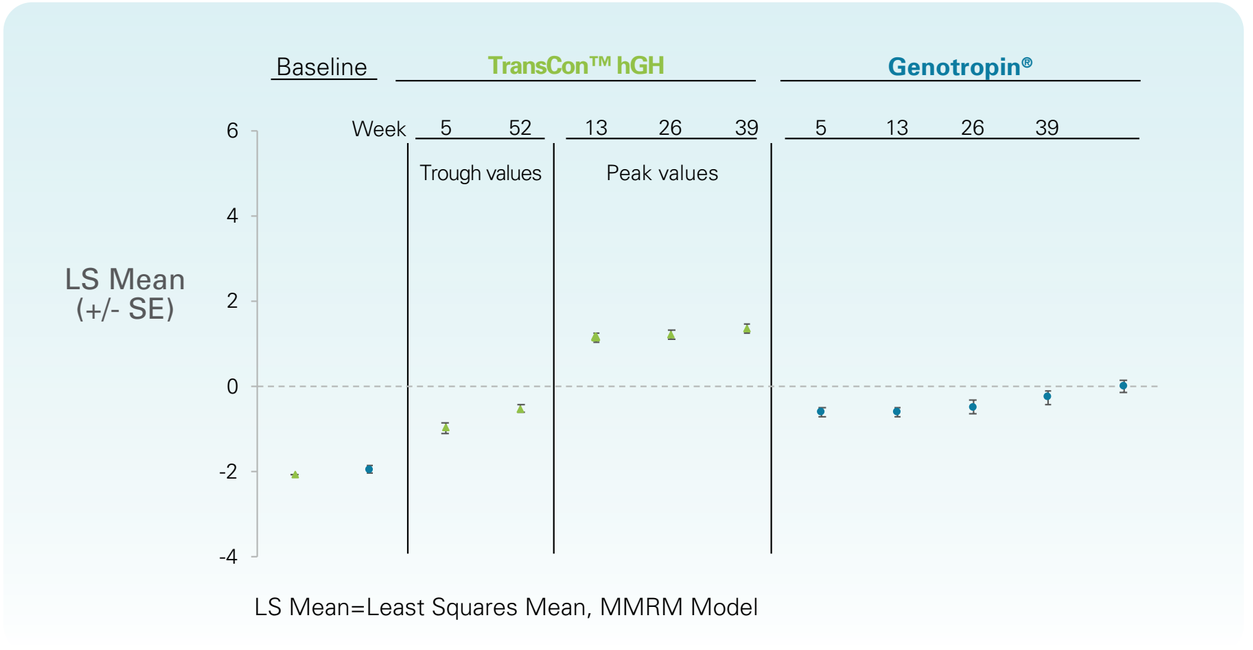
Baseline
TransCon™ hGH
Genotropin®
Week
5
52
13
26
39
5
13
26
39
Trough values
Peak values
LS Mean (+/- SE)
6
4
2
0
-2
-4
LS Mean=Least Squares Mean, MMRM Model

Mean IGF-1 SDS levels for TransCon™ hGH were lower at trough and higher at peak than IGF-1 levels for daily Genotropin®; all 3 parameters trended upward with time and generally remained within normal range (ie, under 2.0 SDS). The mean observed TransCon™ hGH IGF-1 SDS at baseline was -2.08, increasing to -0.7 at Week 52 compared to 0 for daily Genotropin®.

Table 4: Change in Bone Age and BA/CA Over 52 Weeks

Bone Age, years	TransCon™ hGH n=105 Mean (SD)	Genotropin® n=56 Mean (SD)
Baseline	5.84 (2.60)	5.98 (2.68)
Week 52	7.16 (2.72)	7.35 (2.94)
Change from Baseline	**1.36 (0.87)**	**1.35 (0.72)**

BA/CA Ratio	TransCon™ hGH n=105 Mean (SD)	Genotropin® n=56 Mean (SD)
Baseline	0.69 (0.16)	0.70 (0.14)
Week 52	0.75 (0.15)	0.76 (0.14)
Change from Baseline	**0.06 (0.10)**	**0.05 (0.08)**

Over 52 weeks, the mean bone age advanced by 1.36 and 1.35 years in the TransCon™ hGH and daily Genotropin® cohorts. The BA/CA ratio increased to 0.75 and 0.76, for TransCon™ hGH and Genotropin®, respectively, for a BA/CA ratio increase of 0.06 over the trial.

Table 5: Summary of Adverse Events: Safety Population

	TransCon™ hGH n=105 n (%)	Genotropin® n=56 n (%)	Total N=161 n (%)
Treatment-emergent Adverse Events (TEAE)	81 (77.1%)	39 (69.6%)	120 (74.5%)
TEAEs Related to Study Drug	12 (11.4%)	10 (17.9%)	22 (13.7%)
Serious Adverse Events (AEs)	1 (1.0%)	1 (1.8%)	2 (1.2%)
Serious AEs Related to Study Drug	0	0	0
TEAEs Leading to Any Action on Study Drug	2 (1.9%)	1 (1.8%)	3 (1.9%)
TEAEs Leading to Discontinuation of Study Drug	0	0	0

Treatment-emergent AEs were common in both groups (77.1% on TransCon™ hGH and 69.6% on Genotropin®). For those related to study drug, values were 11.4% and 17.9% for TransCon™ hGH and Genotropin®, respectively. Two SAEs were reported during the trial, one on TransCon™ hGH (an appendectomy rated as unrelated to study drug), and one on Genotropin® (a concussion rated as unrelated to study drug). Two AEs in the TransCon™ hGH group resulted in a reduction of dose, due to elevations in IGF-1, while one subject on Genotropin® had a dose reduction due to facial swelling, with a normal IGF-1 value.

Figure 7: Safety Analyses

- Low titer anti-hGH binding antibodies were detected with a similar incidence in both TransCon™ hGH and Genotropin® groups in less than 10% of subjects
 - All were non-neutralizing and antibodies all resolved over time
- Mean fasting glucose (Figure 7a) and hemoglobin A1c (Figure 7b) values were stable and within the normal range for both arms:

Figure 7a

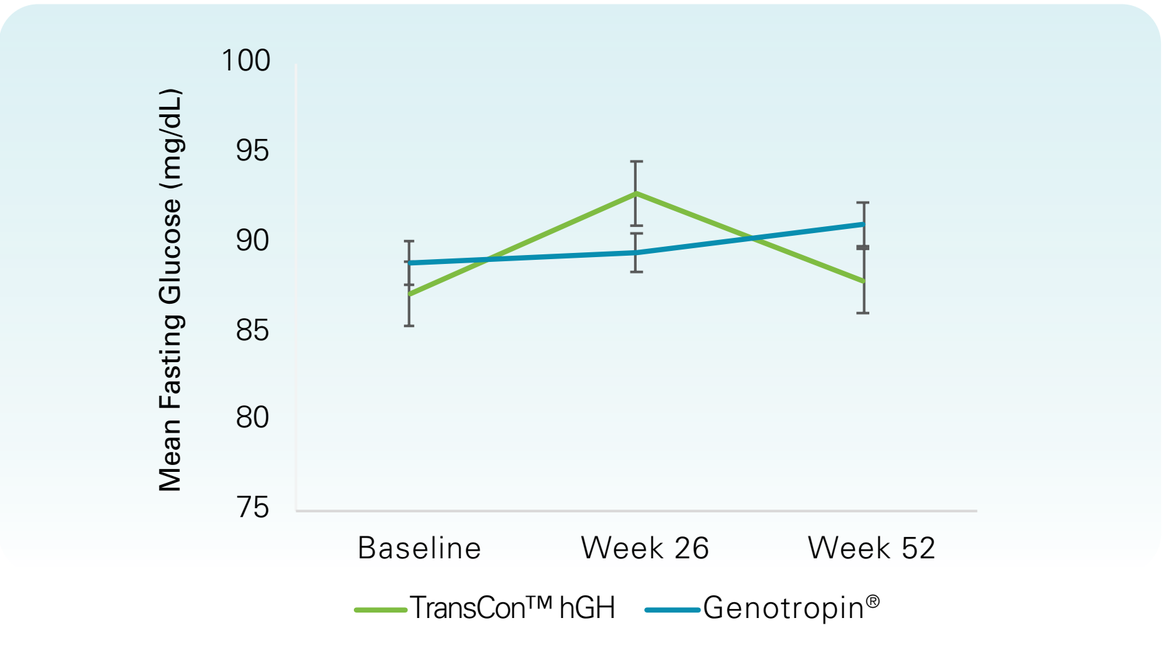
Mean Fasting Glucose (mg/dL)
100
95
90
85
80
75
Baseline
Week 26
Week 52
TransCon™ hGH
Genotropin®

Figure 7b

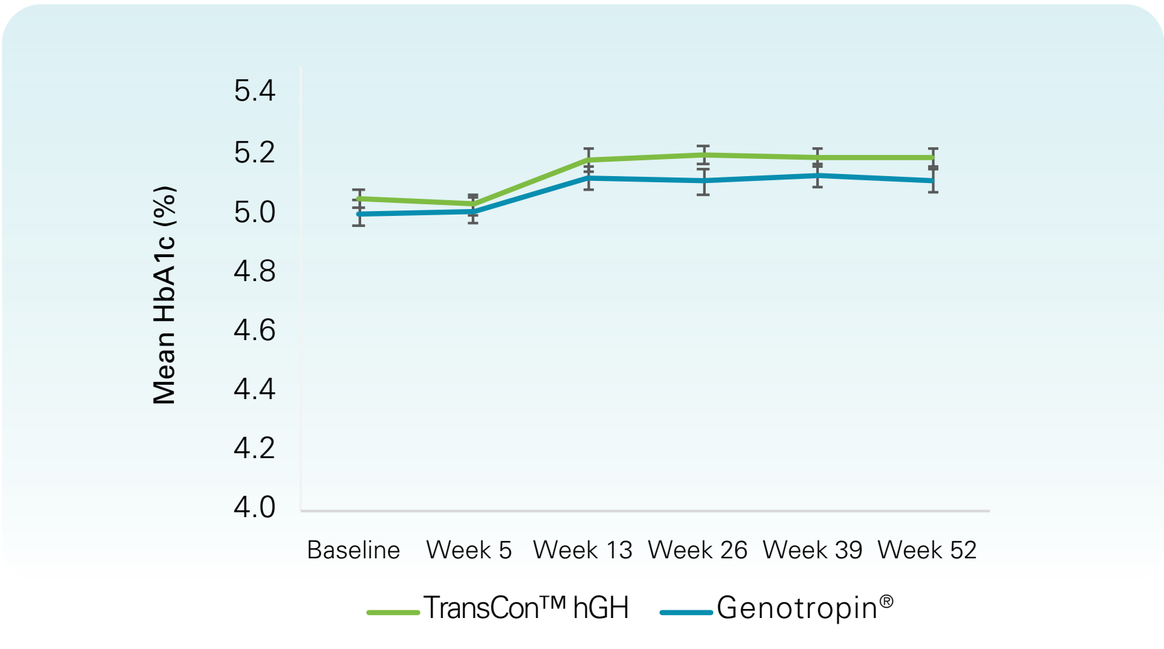
Mean HbA1c (%)
5.4
5.2
5.0
4.8
4.6
4.4
4.2
4.0
Baseline
Week 5
Week 13
Week 26
Week 39
Week 52
TransCon™ hGH
Genotropin®

Safety Analyses
Figure 8: Similar Tolerability: Pain and Itching

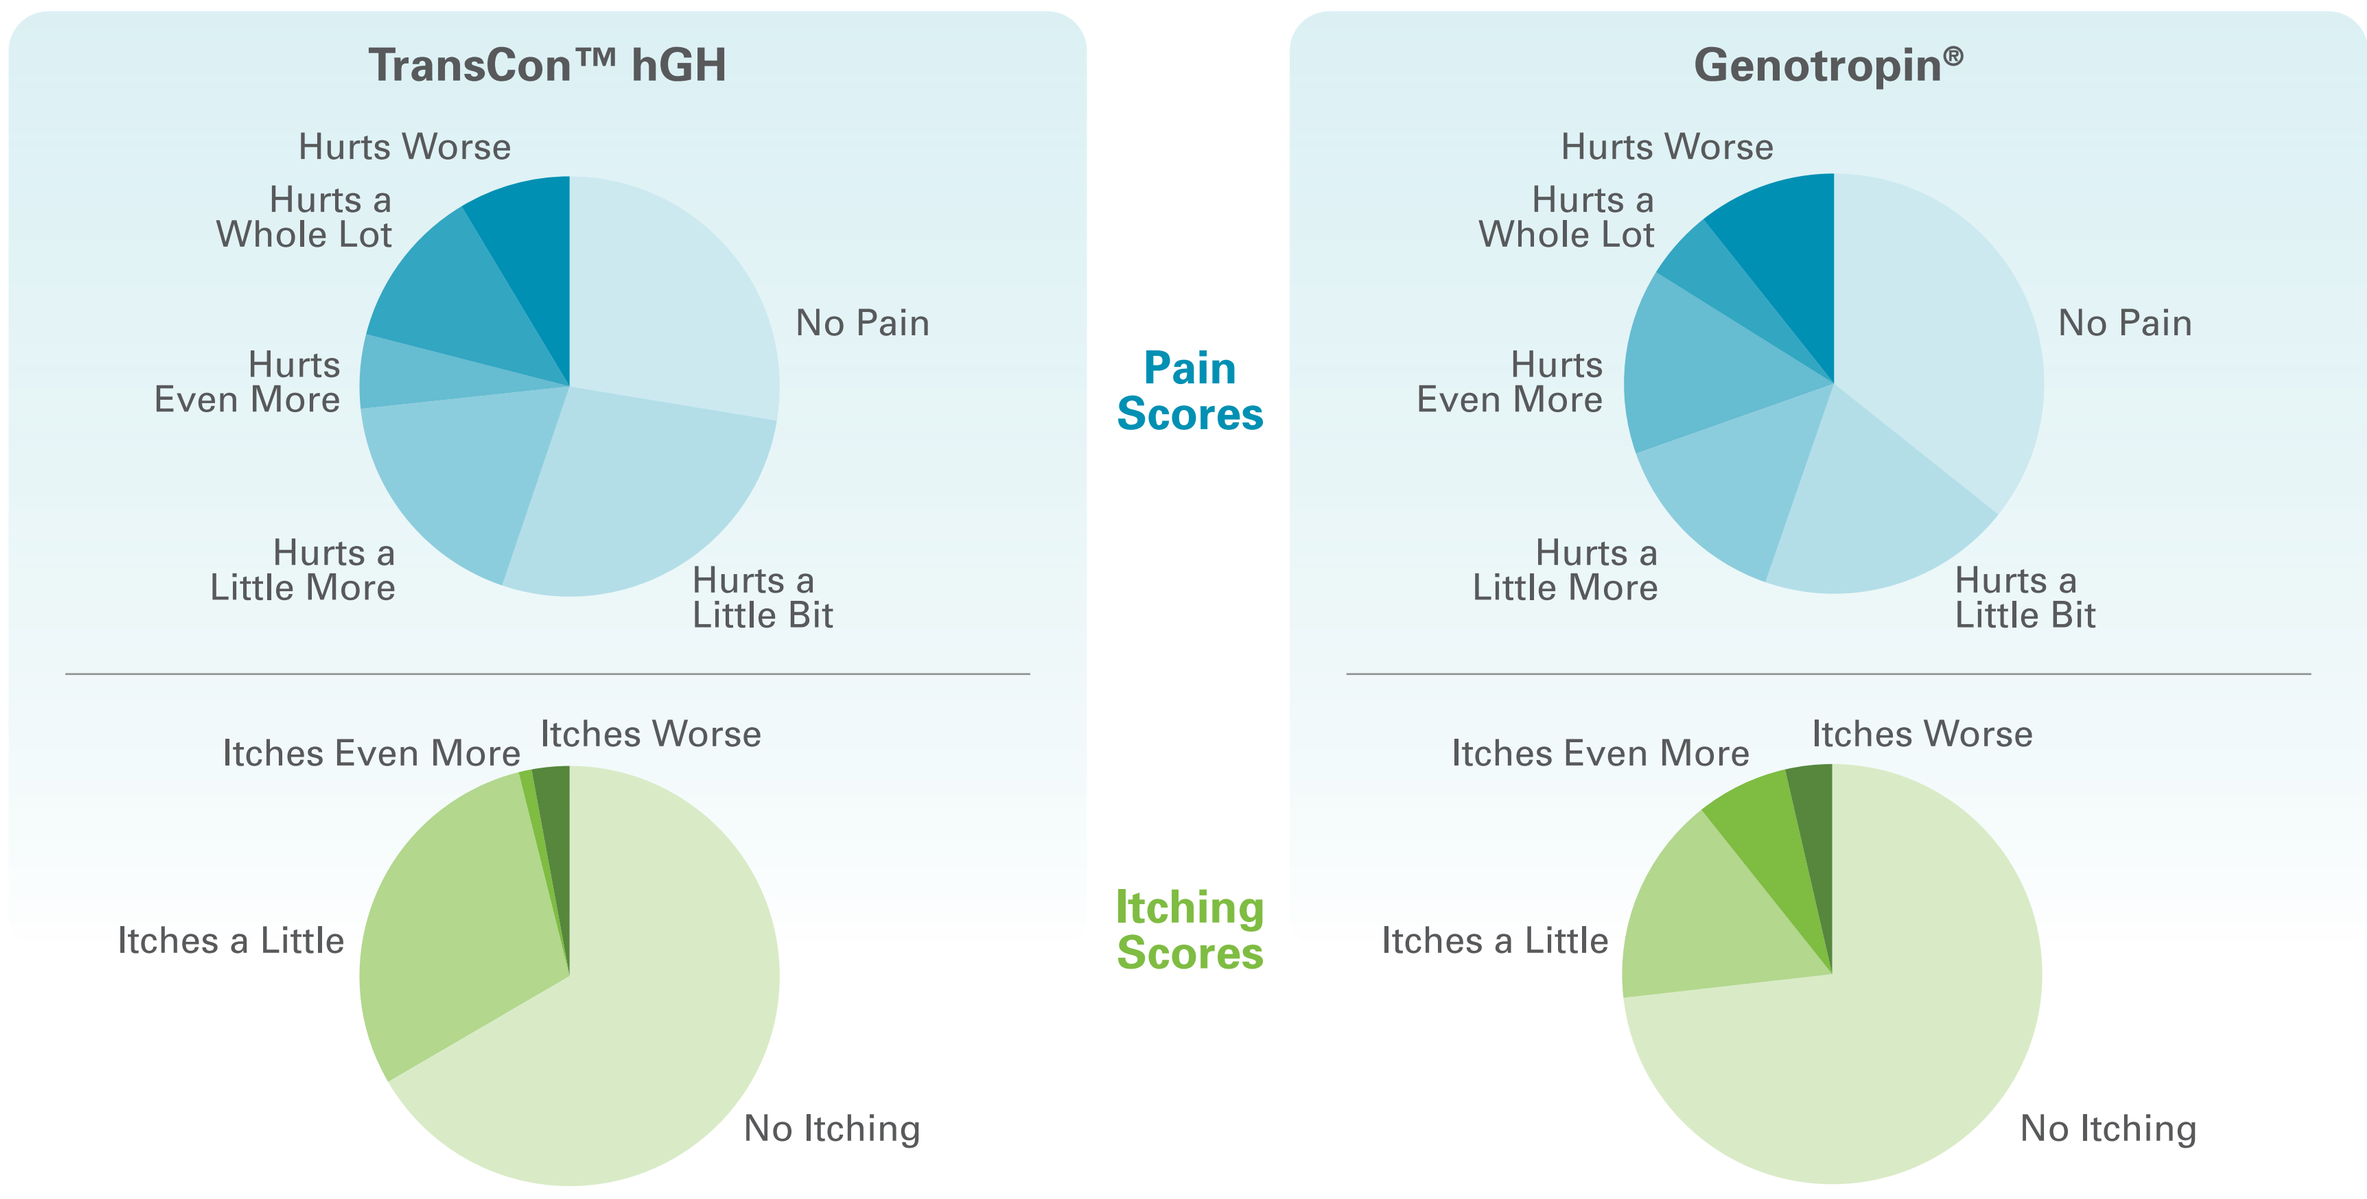
TransCon™ hGH
Genotropin®
Pain Scores
Hurts Worse
Hurts a Whole Lot
Hurts Even More
Hurts a Little More
Hurts a Little Bit
No Pain
Itching Scores
Itches Even More
Itches Worse
Itches a Little
No Itching

Pain and itching were assessed using a Subject Diary, where each dose was logged and rated for injection-site reactions by the subject or caregiver (for Pain assessment,using the Wong-Baker FACES Pain Rating Scale). Proportion of responses are represented for the categories of Pain and Itching, for those receiving TransCon™ hGH and those receiving Genotropin®. The pie charts reflect each subject's worst rating for either Pain or Itching over 52 weeks of receiving TransCon™ hGH or Genotropin®. Responses were similar between treatment groups for each score. Two subjects in each treatment arm experienced mild injection-site reactions that were considered adverse events.

CONCLUSIONS

- Treatment with TransCon™ hGH achieved the primary objective of non-inferiority in AHV at 52 weeks, and further showed superiority over Genotropin® (*p*=0.0088)
 - TransCon™ hGH: 11.2 cm/year
 - Genotropin®: 10.3 cm/year
- IGF-1 SDS scores were generally within the normal range following treatment for both groups
- Safety and tolerability of TransCon™ hGH were consistent with daily Genotropin®

IMPLICATIONS

- Availability of a safe and well-tolerated long-acting hGH therapy has been long awaited and elusive
- The standard of care for pediatric GHD for over the past 30 years has been recombinant hGH administered daily. While daily hGH has both an excellent safety profile and satisfactory efficacy, the frequency of its administration causes a significant burden on daily life for both children with GHD and their caregivers
- The heiGHt Trial results represent another step closer to development of a safe, well-tolerated, and effective long-acting treatment for pediatric GHD

REFERENCES:
[1]Sprogøe K et al. The rationale and design of TransCon Growth Hormone for the treatment of growth hormone deficiency. *Endocr Connect.* 2017;6(8):R171-R181.
[2]Chatelain P et al. A randomized phase 2 study of long-active TransCon GH vs daily GH in childhood GH deficiency. *J Clin Endocrinol Metab.* 2017;102(5):1673-1682.
[3]Bakker B, et al. Height velocity targets from the National Cooperative Growth Study for first-year growth hormone responses in short children. *J Clin Endocrinol Metab.* 2008;93(2):352-357.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group
© April 2019 Ascendis Pharma A/S