Design and Clinical Development of TransCon Growth Hormone for Growth Hormone Deficiency

Michael Beckert[1], MD; David Gilfoyle[1], PhD; Jan Møller Mikkelsen[2]; Grethe Rasmussen[1], PhD; Harald Rau[3], PhD; Kennett Sprogøe[1], PhD
[1]Ascendis Pharma A/S, Hellerup, Denmark; [2]Ascendis Pharma Inc., Palo Alto, USA; [3]Ascendis Pharma GmbH, Heidelberg, Germany

The studies were sponsored by Ascendis Pharma A/S.

Background

TransCon Growth Hormone is designed as a once-weekly sustained-release prodrug of recombinant human growth hormone (hGH, somatropin) (Figure 1). Based on the inert TransCon prodrug technology unmodified native hGH is released with a C_{max} and AUC comparable to daily therapy. TransCon Growth Hormone leverages the known pharmacology of daily hGH and is being developed for the treatment of Growth Hormone Deficiency (GHD) in children and adults.

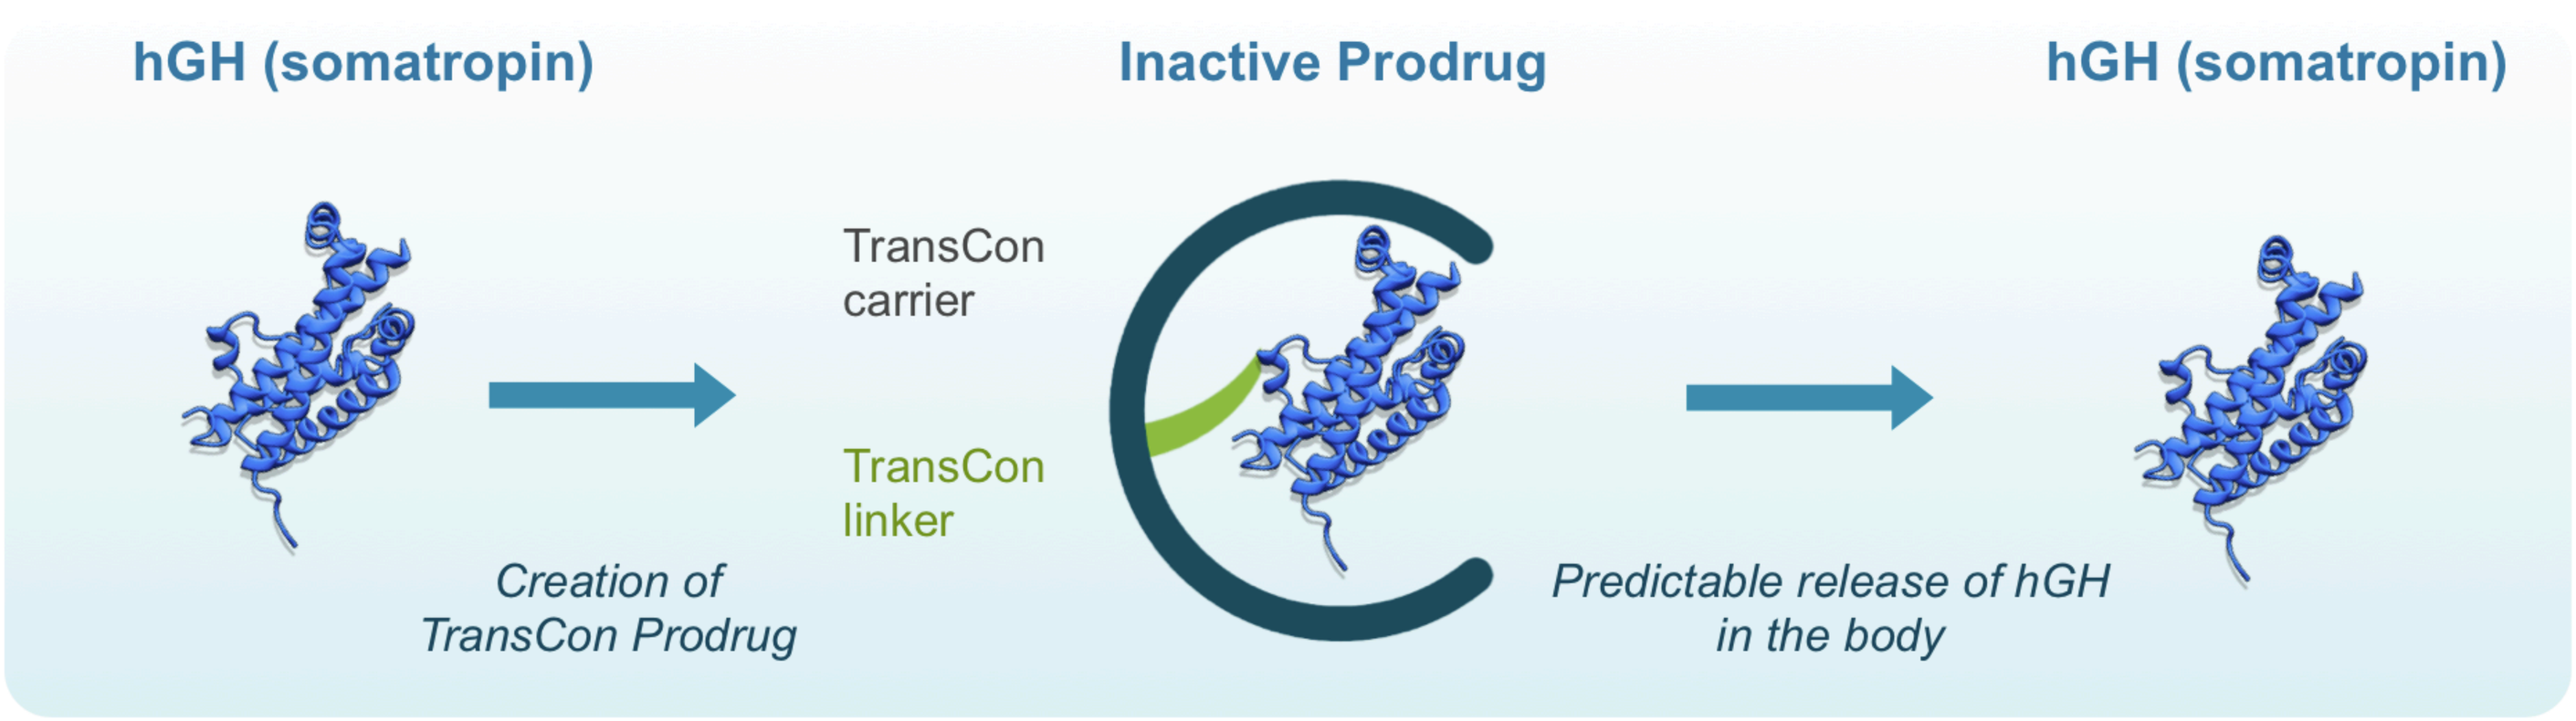
hGH (somatropin)
Inactive Prodrug
hGH (somatropin)
TransCon carrier
TransCon linker
Creation of TransCon Prodrug
Predictable release of hGH in the body

Figure 1: The TransCon hGH prodrug consists of hGH transiently bound to a polyethylene glycol carrier molecule *via* a TransCon linker. The released hGH is unmodified and designed to maintain the same mode of action and distribution in the body as endogenous hGH.

TransCon hGH can be administered in small volumes with a fine (30G or smaller) needle at comparable weekly dose-levels to daily hGH. The product can be stored at room temperature.

Objectives

The objective is to develop a safe and efficacious sustained-release hGH resulting in both hGH and IGF-1 serum concentrations within the therapeutic range, leveraging the safety, efficacy, tolerability and immunogenicity of daily hGH dosing, which has been established for use over decades.

Design and Methods

TransCon Growth Hormone was investigated in a total of two Healthy Volunteer (HV) Phase 1 and two Phase 2 studies in adults and children with GHD. Daily hGH with an equivalent dose-level was included in most trials to enable comparison of hGH-levels, IGF-1 levels; and efficacy by height velocity in GHD.

Phase 1 HV and Phase 2 AGHD Studies

A total of 68 healthy adult subjects were included in two Phase 1 single-dose pharmacokinetic and pharmacodynamic studies, investigating TransCon hGH dose-levels from 0.04 mg hGH/kg/week up to 0.36 mg hGH/kg/week. In these Phase 1 studies, TransCon hGH was shown to

1) Be safe and well tolerated,
2) Be suitable for a once-weekly dosing regimen,
3) Provide a pharmacokinetic hGH and pharmacodynamic IGF-1 response comparable to daily hGH treatment throughout the dosing period (Figure 2 and 3).

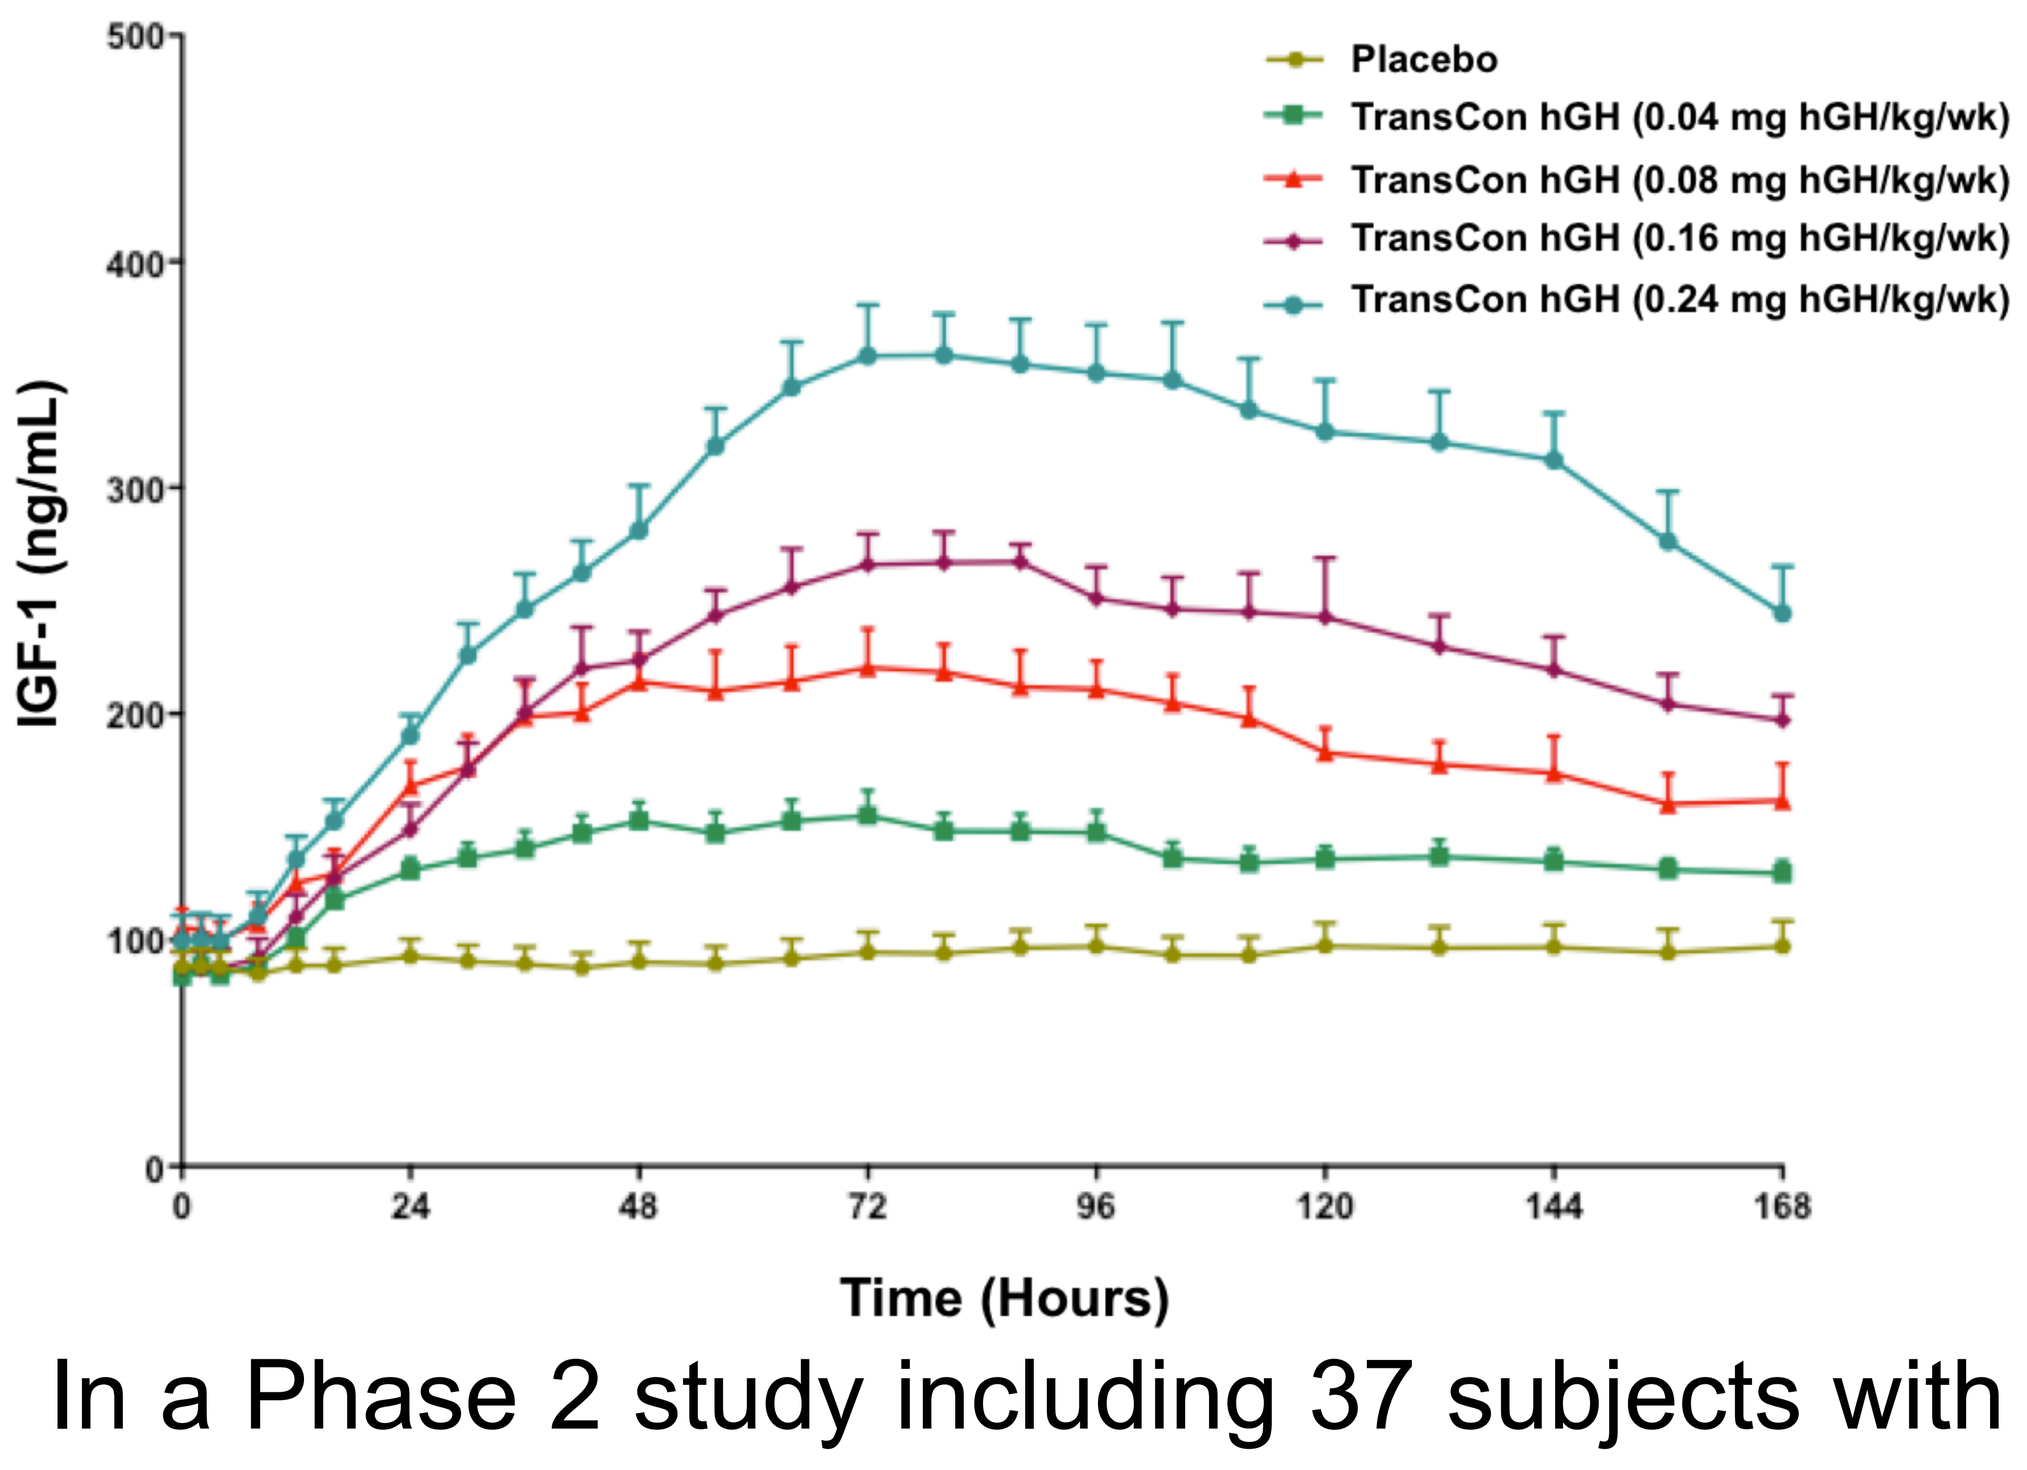
Placebo
TransCon hGH (0.04 mg hGH/kg/wk)
TransCon hGH (0.08 mg hGH/kg/wk)
TransCon hGH (0.16 mg hGH/kg/wk)
TransCon hGH (0.24 mg hGH/kg/wk)
IGF-1 (ng/mL)
0
100
200
300
400
500
0
24
48
72
96
120
144
168
Time (Hours)

Figure 2: Pharmacodynamic profile of mean plasma IGF-1 levels during the initial 7 days following a single s.c. administration of TransCon hGH in a Phase 1 study with 44 Healthy Volunteers (M. Beckert et al., ENDO 2010, San Diego; NCT01010425).

In a Phase 2 study including 37 subjects with Adult Growth Hormone Deficiency (AGHD), with four repeated-dosings, TransCon hGH demonstrated dose-linearity over a dose-range of 0.02 mg hGH/kg/week up to 0.08 mg hGH/kg/week and comparable efficacy (IGF-1) to the daily hGH control group on exactly the same weekly dose-level (0.04 mg hGH/kg/week).

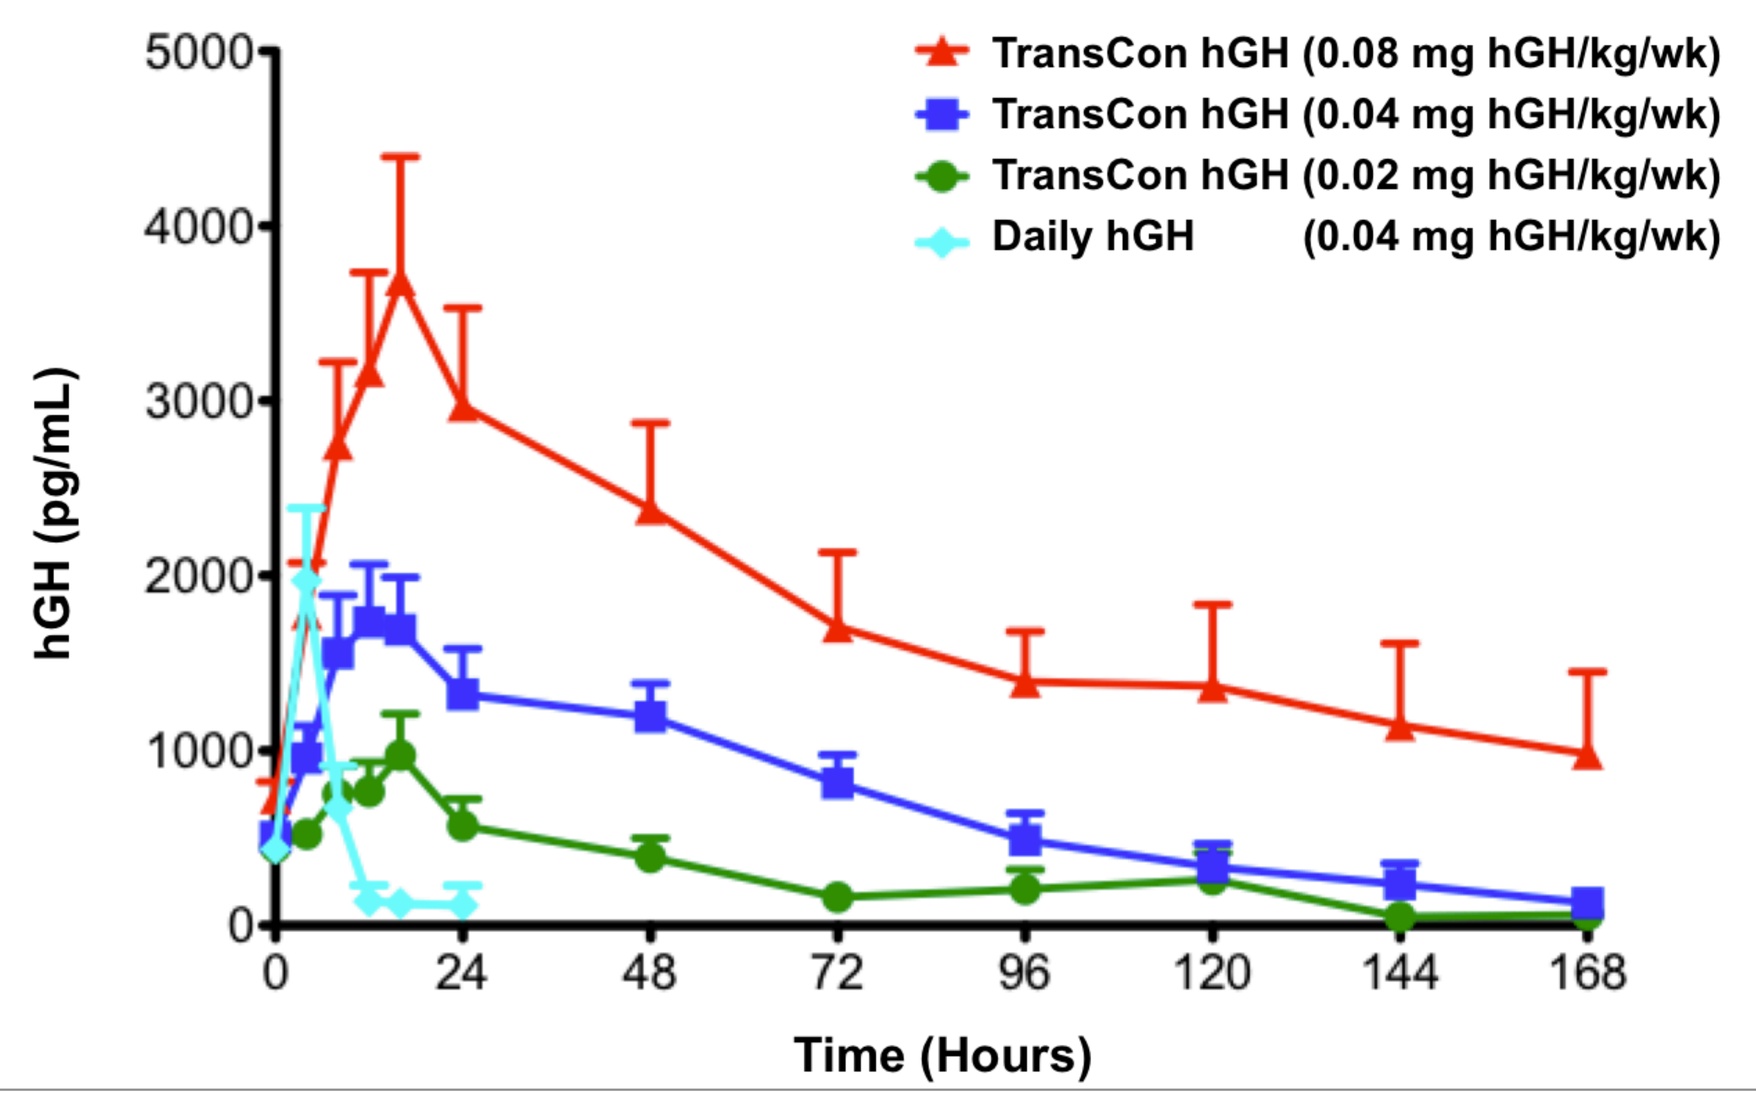
TransCon hGH (0.08 mg hGH/kg/wk)
TransCon hGH (0.04 mg hGH/kg/wk)
TransCon hGH (0.02 mg hGH/kg/wk)
Daily hGH (0.04 mg hGH/kg/wk)
hGH (pg/mL)
0
1000
2000
3000
4000
5000
0
24
48
72
96
120
144
168
Time (Hours)

Figure 3: Pharmacokinetic profile of hGH levels for TransCon hGH and daily hGH at Week 4 in Phase 2 AGHD study with 37 subjects (C. Hoybye et al., ENDO 2012, Houston; NCT01247675).

Results – Phase 2 Pediatric GHD Study

This pediatric Phase 2 clinical study was designed to investigate the safety, efficacy, pharmacokinetics and pharmacodynamics of TransCon hGH compared to daily hGH over a treatment period of six months.

The results of this Phase 2 study in 53 pediatric patients with GHD confirmed the safety, tolerability and the suitability of TransCon hGH for once-weekly dosing. An equivalent dose-level to daily hGH demonstrated slightly numerically higher growth rates compared to daily hGH treatment (Figure 4). Maximum hGH blood concentration was also comparable between equivalent weekly doses of TransCon hGH and daily hGH (Figure 5). No drug-related SAEs occurred, no lipoatrophy, nodule formation or anti-hGH neutralizing antibodies were seen. IGF-1 changes suggest a dose response and levels were in the expected range (P. Chatelain et al., ENDO 2016, Boston; NCT01947907).

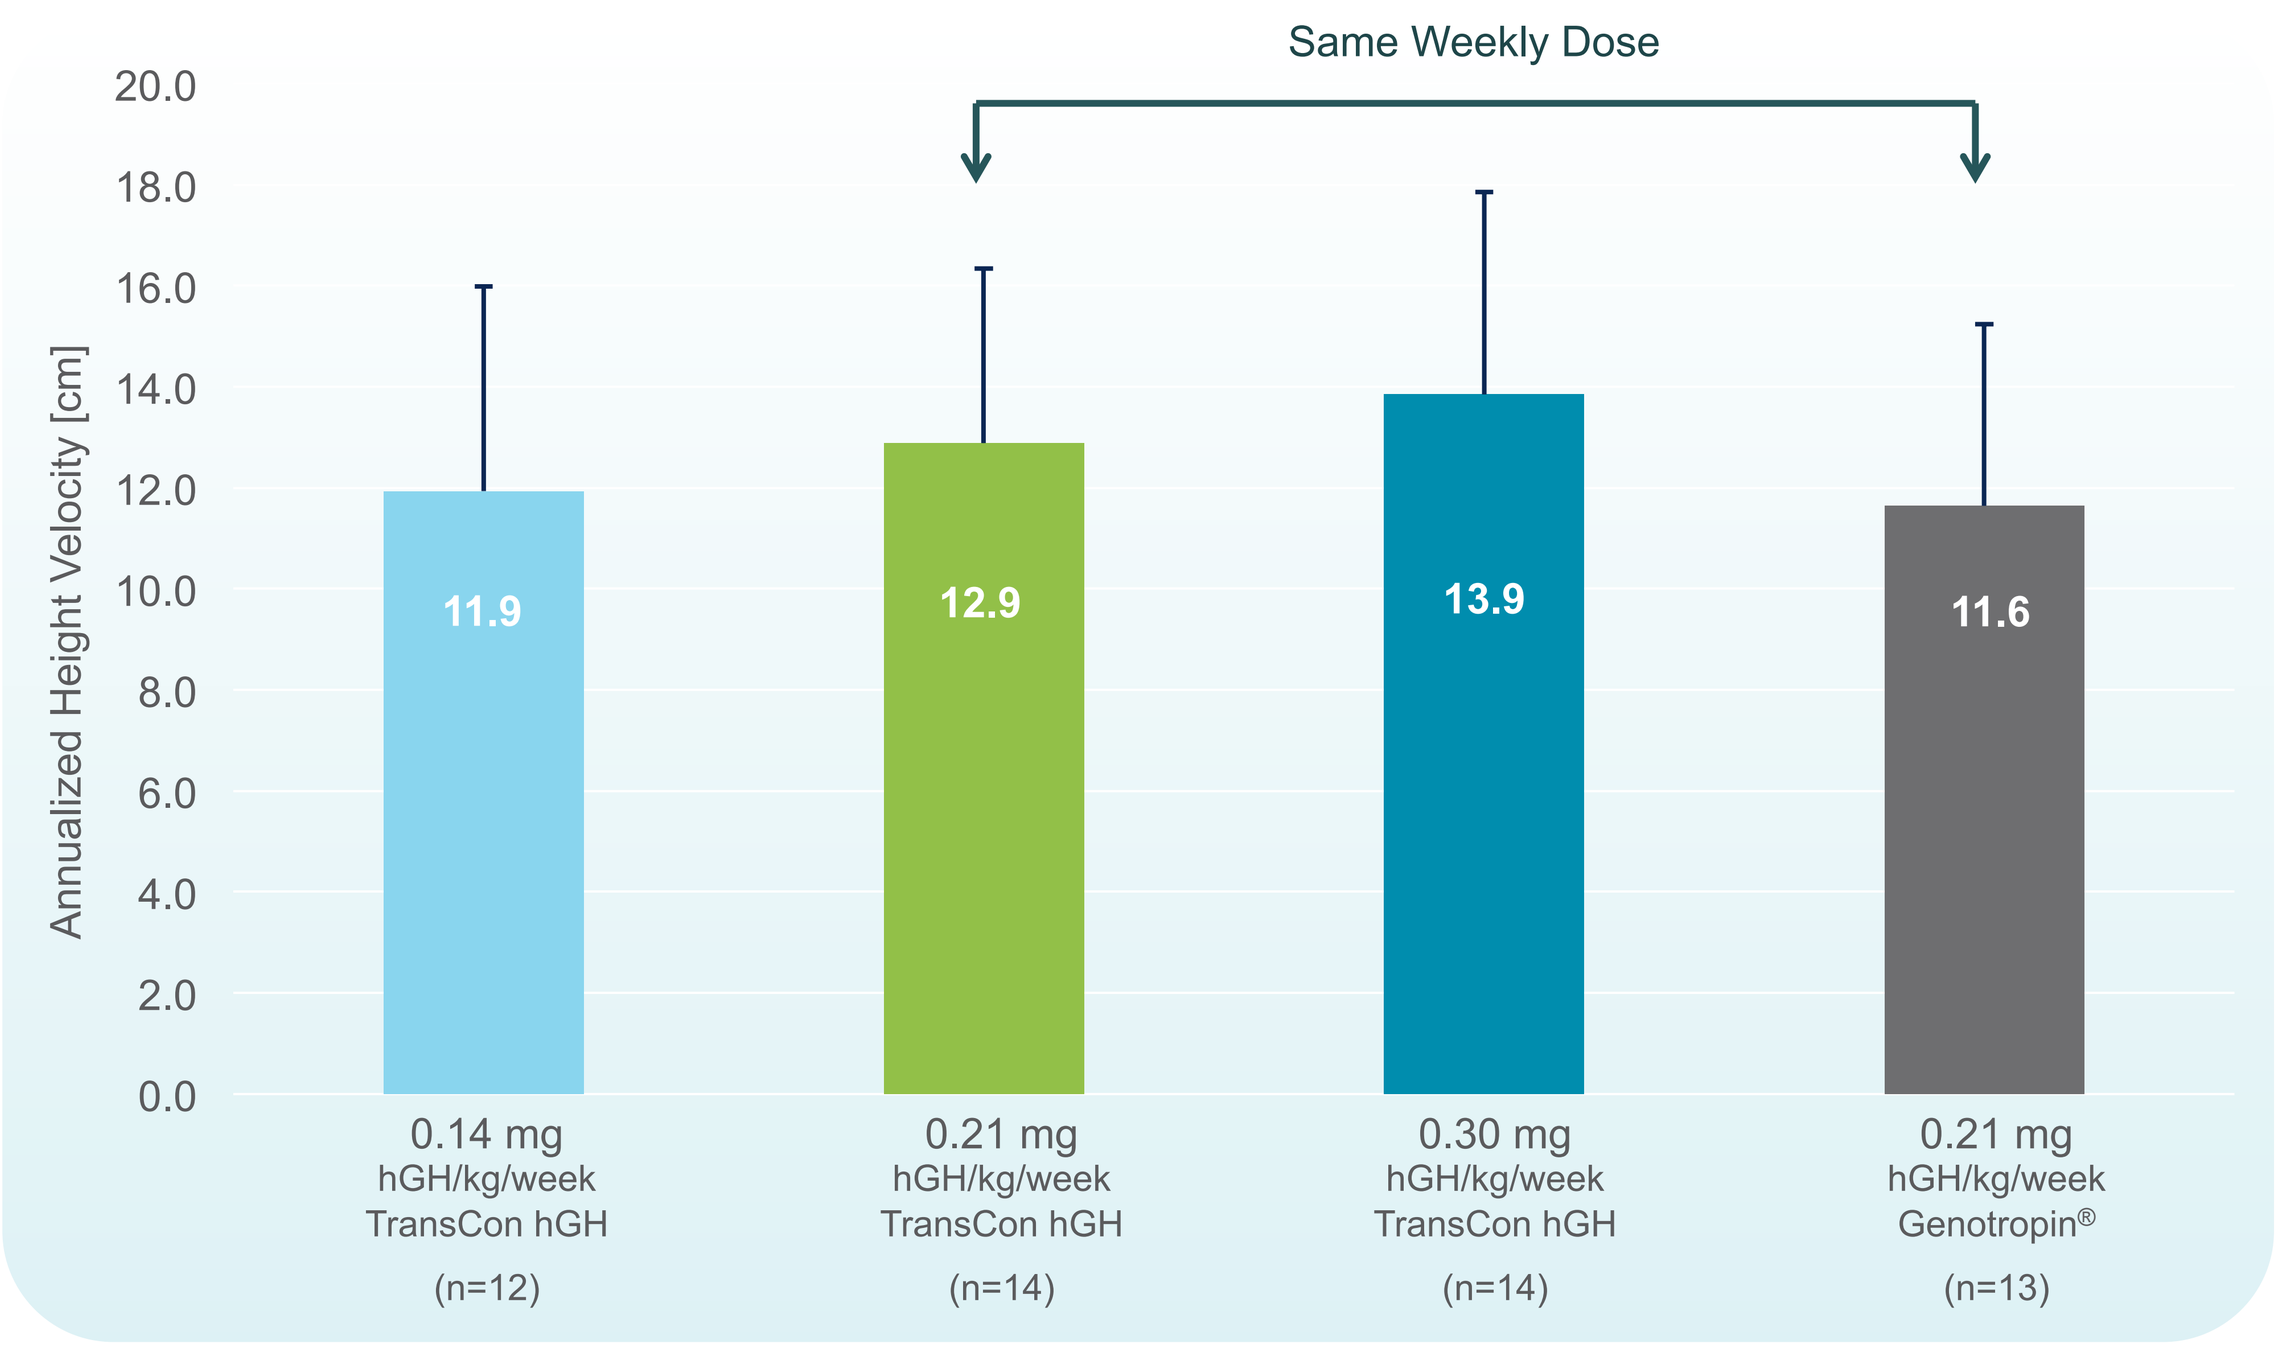
Same Weekly Dose
Annualized Height Velocity [cm]
0.0
2.0
4.0
6.0
8.0
10.0
12.0
14.0
16.0
18.0
20.0
11.9
12.9
13.9
11.6
0.14 mg hGH/kg/week TransCon hGH (n=12)
0.21 mg hGH/kg/week TransCon hGH (n=14)
0.30 mg hGH/kg/week TransCon hGH (n=14)
0.21 mg hGH/kg/week Genotropin® (n=13)

Figure 4: Annualized Height Velocity (Mean + SD) of full dataset of 53 patients after 6 months of treatment in the Phase 2 pediatric GHD study.

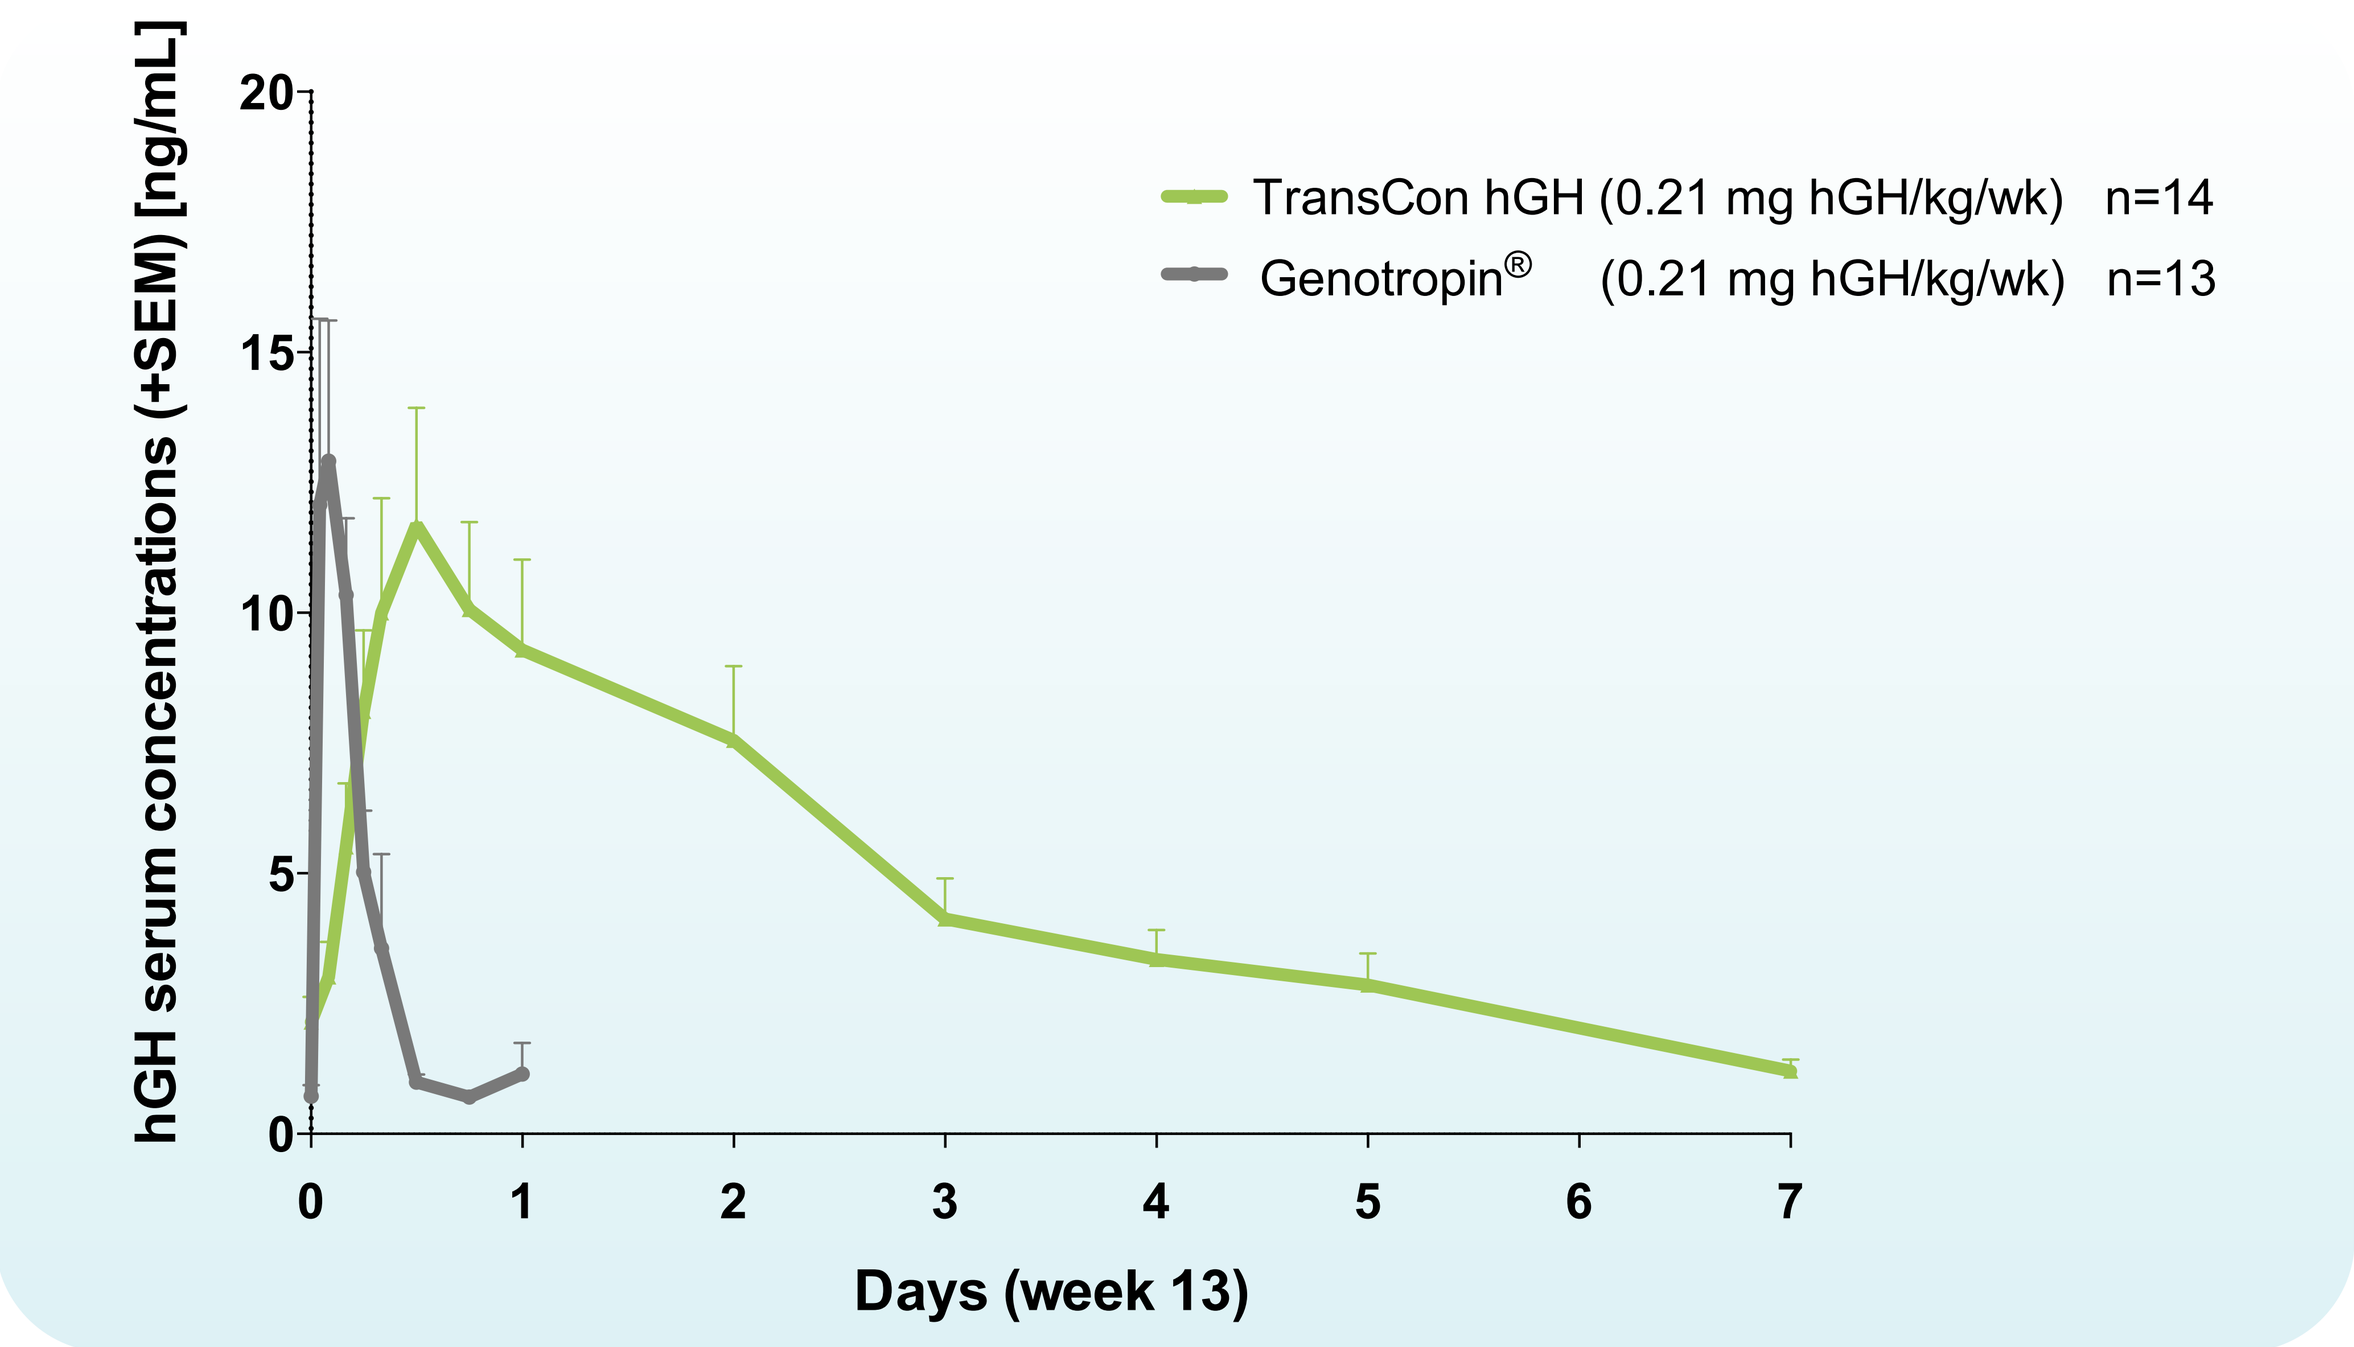
TransCon hGH (0.21 mg hGH/kg/wk) n=14
Genotropin® (0.21 mg hGH/kg/wk) n=13
hGH serum concentrations (+SEM) [ng/mL]
0
5
10
15
20
0
1
2
3
4
5
6
7
Days (week 13)

Figure 5: hGH levels for TransCon hGH (0.21 mg hGH/kg/week) and daily hGH (0.21 mg hGH/kg/week) at Week 13 in the Phase 2 pediatric GHD study.

Recently Initiated – Phase 3 Pediatric GHD Study

This is a Phase 3, randomized, open-label, active-controlled trial investigating the safety, tolerability and efficacy of TransCon hGH as compared to standard daily growth hormone, over 52 weeks in prepubertal children with GHD.

It was initiated in August 2016 and will be conducted in North America, Europe, the Middle East, North Africa and Oceania. A brief description of this clinical study is available on www.heighttrial.com (NCT02781727).

Conclusion

To date, the TransCon Growth Hormone has demonstrated efficacy and safety comparable to that observed with daily hGH. Injection site reactions were generally mild and similar to daily hGH injections, with no nodule formation or lipoatrophy noted. In addition, no neutralizing anti-hGH antibodies occured.

TransCon hGH was shown to be dose-linear across different dose-levels (hGH and IGF-1) and demonstrated the same therapeutic effect (annualized height velocity) to daily hGH on the same weekly dose-level (pediatric GHD study; 0.21 mg hGH/kg/week).

TransCon hGH can be administered in small volumes with a fine (30G or smaller) needle at comparable weekly dose-levels to daily hGH. The product can be stored at room temperature.

The completed clinical studies support Phase 3 development with TransCon hGH. A Phase 3 study in pediatric GHD, the heiGHt Trial, was recently initiated.

Disclosure Statement: All authors listed above are employees and shareholders of Ascendis Pharma.