P4507: TransCon™ IL-2 β/γ: a novel long-acting prodrug of receptor-biased IL-2 designed for improved pharmacokinetics and optimal activation of T cells for the treatment of cancer

David B Rosen[1], Burkhardt Laufer[2], Thomas Knappe[2], Meike Schnabel[2], Diana Begaliew[2], Joachim Zettler[2], Mathias Krusch[2], Yvonne Hehn[2], Ana Bernhard[2], Stefan Heinig[2], Valentino Konjik[2], Thomas Wegge[2], Yang Wu[1], Wenjie Song[1], Vibeke Breinholt[3], Mohammad Tabrizi[1], Kennett Sprogøe[3], and Juha Punnonen[1]

[1]Ascendis Pharma, Inc., Palo Alto, California, USA; [2]Ascendis Pharma GmbH, Heidelberg, Germany; [3]Ascendis Parma A/S, Copenhagen, Denmark

BACKGROUND AND METHODS

Recombinant Interleukin-2 (IL-2, aldesleukin) is an approved immunotherapy for metastatic melanoma and renal cell carcinoma. Yet, treatment with IL-2 can induce severe side effects, such as vascular leak syndrome (VLS) and eosinophilic infiltration of cardiac and pulmonary tissues[1]. IL-2 promotes the survival, proliferation and anti-tumor functions of CD8+ cytotoxic T cells, CD4+ effector T cells and natural killer (NK) cells, which mostly express the dimeric IL-2Rβ/γ receptor with low IL-2Rα levels[1]. However, IL-2 also potently activates IL-2Rα+ immunosuppressive regulatory T cells (Tregs) as well as IL-2Rα+ eosinophils, type 2 innate lymphoid cells and endothelial cells, which may promote VLS[2,3,4,5]. Aldesleukin is rapidly cleared with an elimination half-life ($t_{1/2}$) of 85 minutes, requires thrice daily dosing and results in large fluctuations in serum exposure, likely contributing to its toxicity[6]. Thus, IL-2 has two critical drawbacks for cancer immunotherapy: 1) potent activation of undesired cell types expressing IL-2Rα and 2) high C_{max} with rapid clearance.

Figure 1: TransCon IL-2 β/γ is a next generation IL-2 designed for 1) desired receptor selectivity *and* 2) optimized exposure

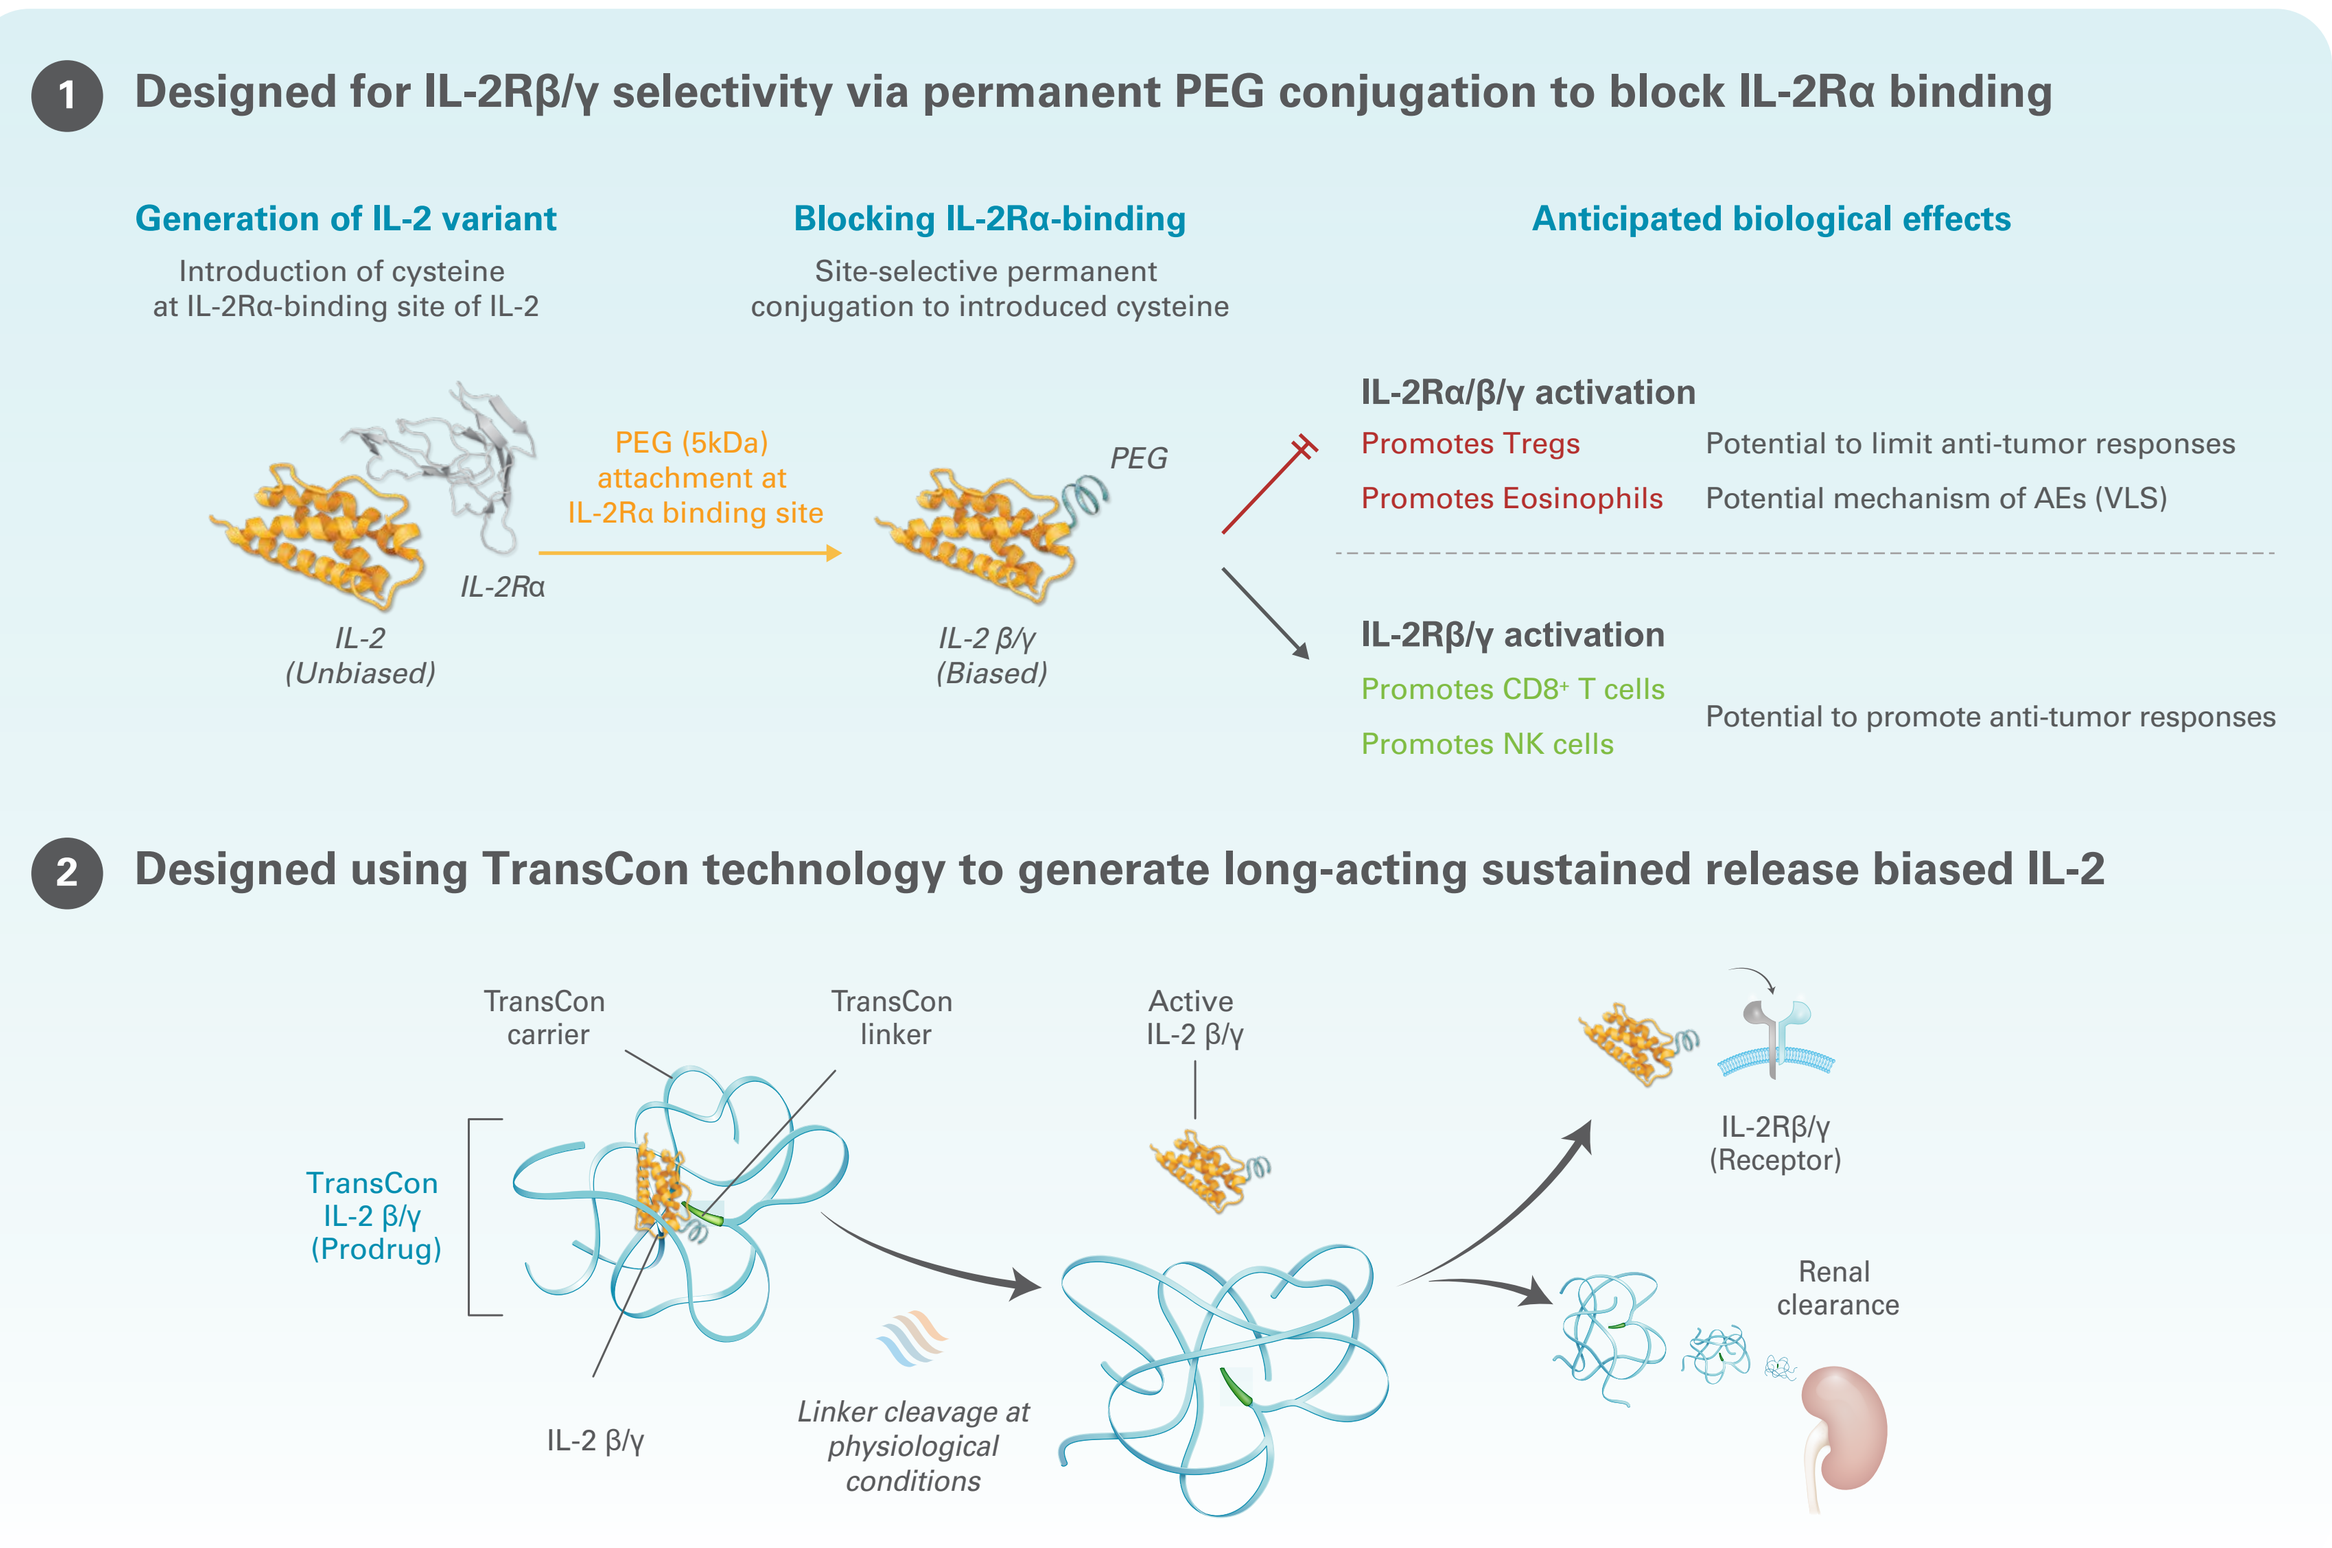
1 Designed for IL-2Rβ/γ selectivity via permanent PEG conjugation to block IL-2Rα binding
Generation of IL-2 variant
Introduction of cysteine at IL-2Rα-binding site of IL-2
Blocking IL-2Rα-binding
Site-selective permanent conjugation to introduced cysteine
Anticipated biological effects
PEG (5kDa) attachment at IL-2Rα binding site
IL-2 (Unbiased)
IL-2Rα
IL-2 β/γ (Biased)
PEG
IL-2Rα/β/γ activation
Promotes Tregs
Promotes Eosinophils
Potential to limit anti-tumor responses
Potential mechanism of AEs (VLS)
IL-2Rβ/γ activation
Promotes CD8+ T cells
Promotes NK cells
Potential to promote anti-tumor responses
2 Designed using TransCon technology to generate long-acting sustained release biased IL-2
TransCon carrier
TransCon linker
Active IL-2 β/γ
TransCon IL-2 β/γ (Prodrug)
IL-2 β/γ
Linker cleavage at physiological conditions
IL-2Rβ/γ (Receptor)
Renal clearance

METHODS

We developed TransCon (transient conjugation) IL-2 β/γ, a novel long-acting prodrug of a receptor-biased IL-2 (IL-2 β/γ) to optimally address each of these drawbacks. First, to block IL-2Rα binding yet retain IL-2Rβ/γ activity, we created IL-2 β/γ by permanently attaching a small PEG moiety to an engineered cysteine placed at the IL-2Rα binding site.

Second, to improve PK properties, we attached the receptor-biased IL-2 β/γ to a TransCon carrier via a TransCon linker, shielding bioactivity and creating a prodrug. Under physiological conditions, TransCon IL-2 β/γ was designed for sustained release of the bioactive IL-2 β/γ from the PEG carrier, aiming for a much lower C_{max} and longer effective half-life of released IL-2 β/γ compared to aldesleukin.

RESULTS

In binding and cell-based functional assays, free IL-2 β/γ demonstrated desirable IL-2 receptor selectivity, maintaining IL-2R βγ potency while losing IL2Rα potency. *In vitro*, TransCon IL-2 β/γ showed expected slow-release kinetics. In mouse models, TransCon IL-2 β/γ promoted CD8+ T cell and NK cell proliferation and activation. In cynomolgous monkeys (non-human primates [NHP]), a single dose of TransCon IL-2 β/γ was well tolerated and induced a more robust expansion of CD8+ T cell subsets and NK cells relative to CD4+ T cell subsets or eosinophils as compared to daily aldesleukin treatment. TransCon IL-2 β/γ demonstrated a long prodrug half-life in mice (~22h) and monkeys (~32h), supporting every three week clinical dosing. Consistent with these observations, TransCon IL-2 β/γ induced lower levels of systemic inflammatory cytokines and endothelial activity biomarkers when compared to aldesleukin.

The data presented here provide evidence that TransCon IL-2 β/γ offers improved pharmacokinetic properties and optimal activation of cytolytic lymphocytes with improved tolerability.

For questions, email
oncology@ascendispharma.com

TransCon IL-2 β/γ was designed as a novel, long-acting prodrug of receptor-biased IL-2 that has the potential to overcome the challenges of existing IL-2 treatments.

Figure 2: Permanently PEGylated IL-2 β/γ demonstrated bias with reduced IL-2Rα binding and Treg activity but well-retained IL-2Rβ binding and activity

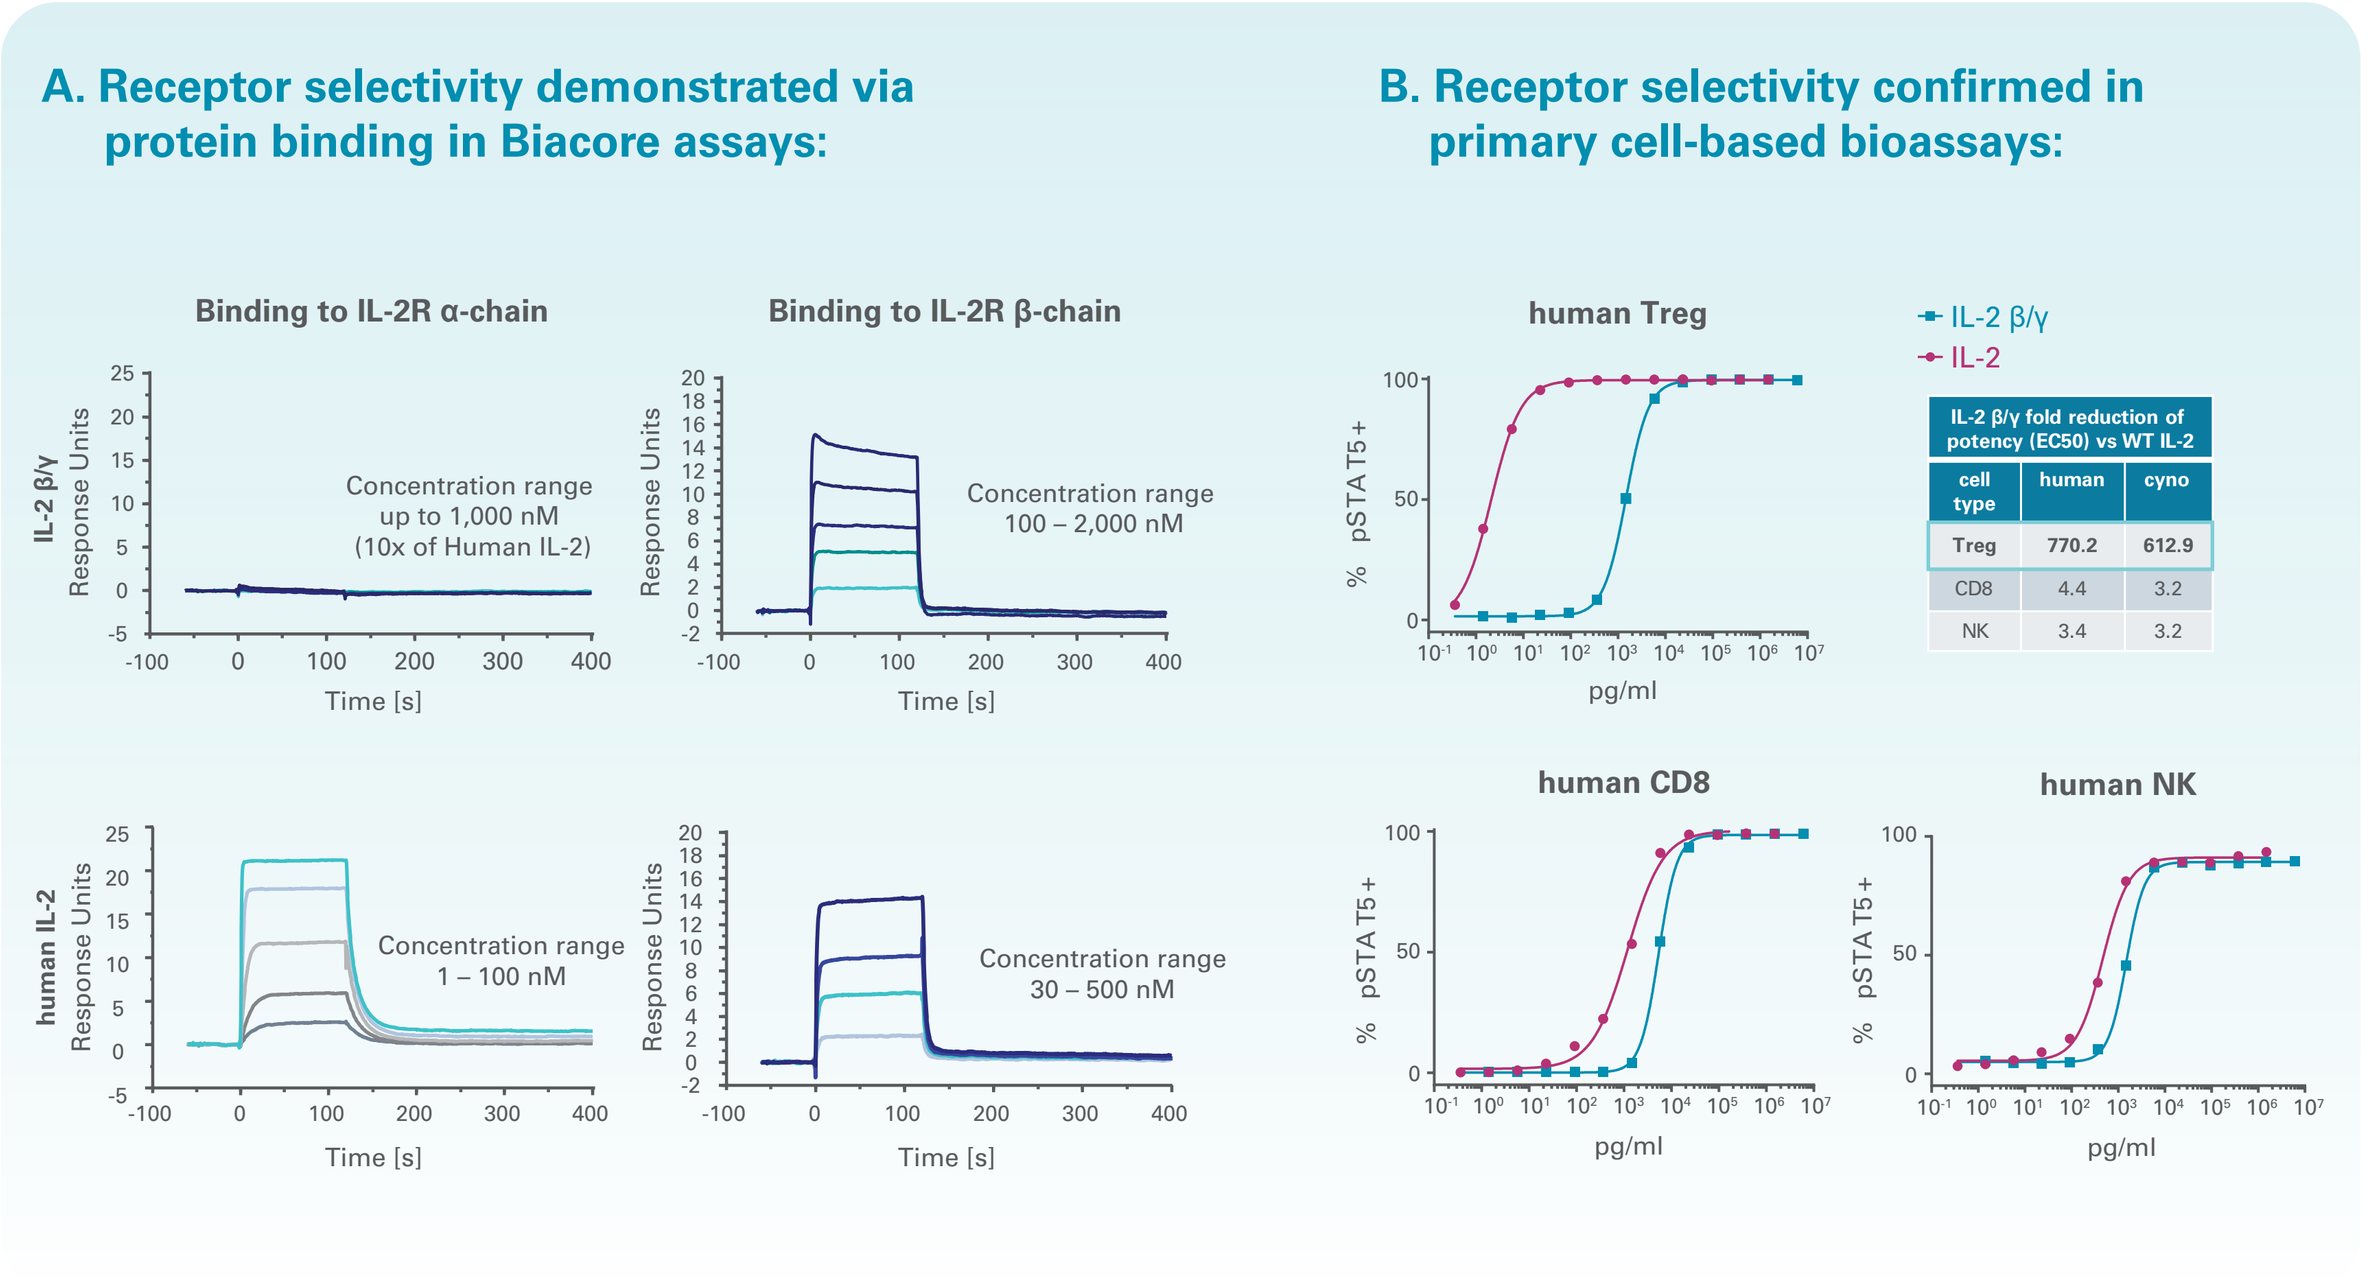
A. Receptor selectivity demonstrated via protein binding in Biacore assays:
Binding to IL-2R α-chain
Binding to IL-2R β-chain
Concentration range up to 1,000 nM (10x of Human IL-2)
Concentration range 100 – 2,000 nM
Concentration range 1 – 100 nM
Concentration range 30 – 500 nM
IL-2 β/γ
human IL-2
Response Units
Time [s]
B. Receptor selectivity confirmed in primary cell-based bioassays:
human Treg
human CD8
human NK
IL-2 β/γ
IL-2
% pSTAT5+
pg/ml
IL-2 β/γ fold reduction of potency (EC50) vs WT IL-2
cell type | human | cyno
Treg | 770.2 | 612.9
CD8 | 4.4 | 3.2
NK | 3.4 | 3.2

A) Biacore was performed by injecting IL-2 β/γ and unbiased recombinant human IL-2 for 2 min onto human IL-2Rα-Fc or human IL-2Rβ-Fc coated sensor chips and then dissociating for 10 min at pH 7.4 and 25.0 °C. IL-2 receptor subunits were captured via immobilized mouse anti-human IgG1(Fc) antibody. Displayed are double reference subtracted sensorgrams at the indicated concentrations of IL-2 β/γ (top) or recombinant human IL-2 (bottom). B) Whole blood from humans or cynomolgus monkeys was stimulated with IL-2 or IL-2 β/γ for 30 minutes at indicated doses then analyzed by flow cytometry for p-STAT5 in the indicated cellular populations.

RESULTS

Figure 3: TransCon IL-2 β/γ provided sustained release of IL-2 β/γ in a controlled and predictable fashion

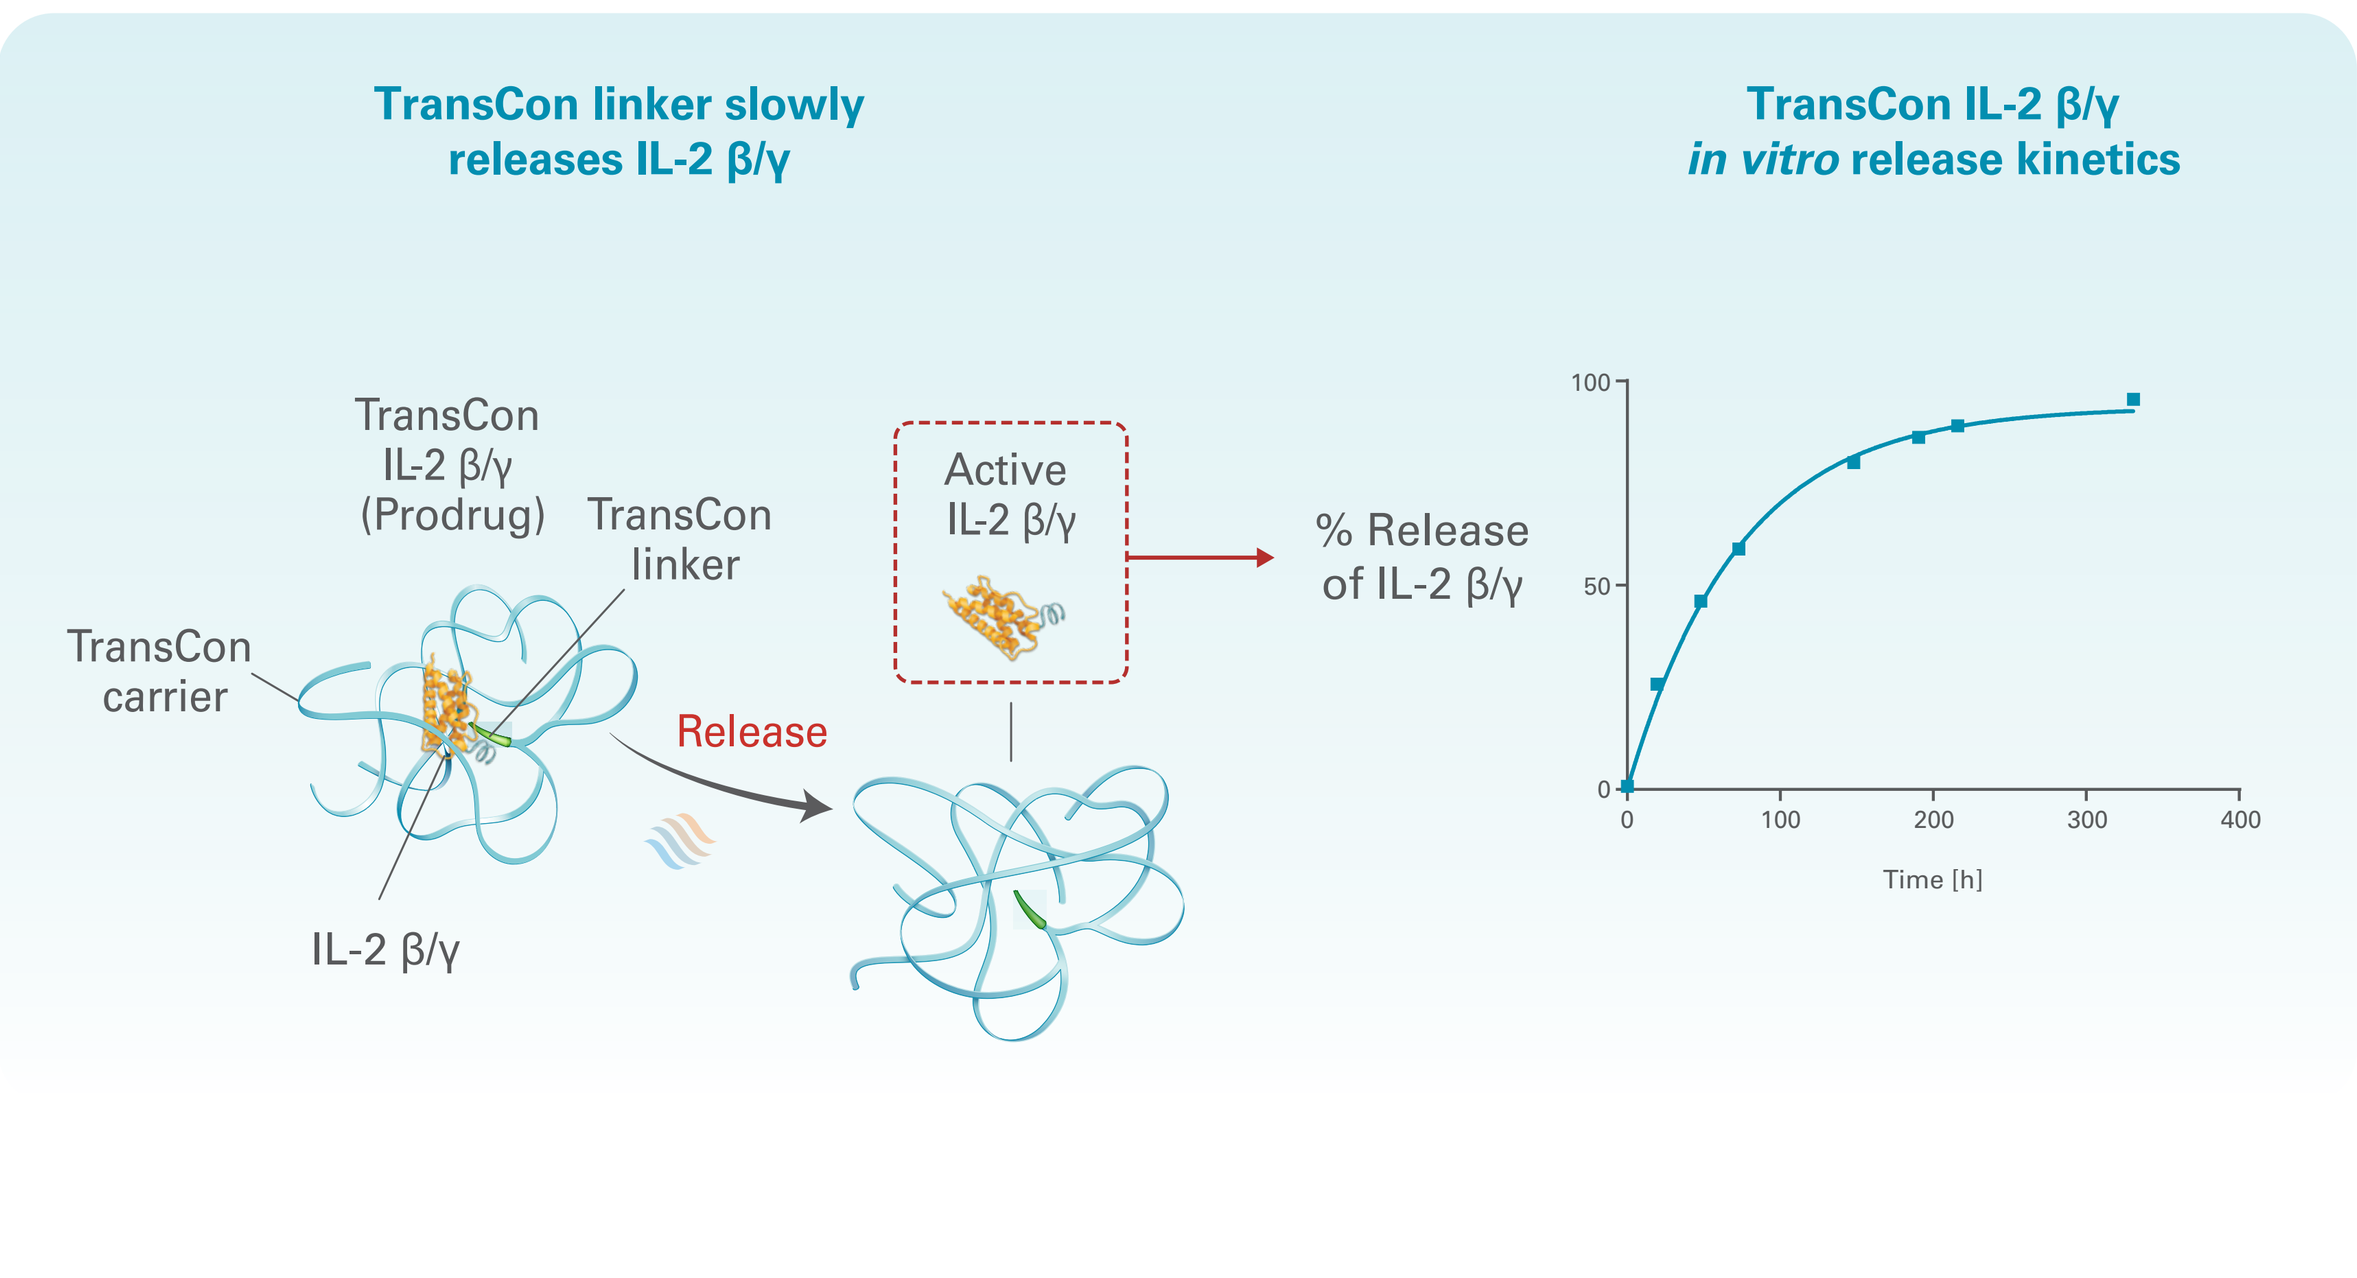
TransCon linker slowly releases IL-2 β/γ
TransCon IL-2 β/γ *in vitro* release kinetics
TransCon IL-2 β/γ (Prodrug)
TransCon linker
TransCon carrier
IL-2 β/γ
Release
Active IL-2 β/γ
% Release of IL-2 β/γ
Time [h]

In vitro release kinetics were performed on TransCon IL-2 β/γ over time at 37°C in PBS. Concentrations of released IL-2 β/γ were measured by RP-HPLC at the indicated timepoints.

Figure 4: TransCon IL-2 β/γ robustly expanded CD8+ T cells and NK cells in mice

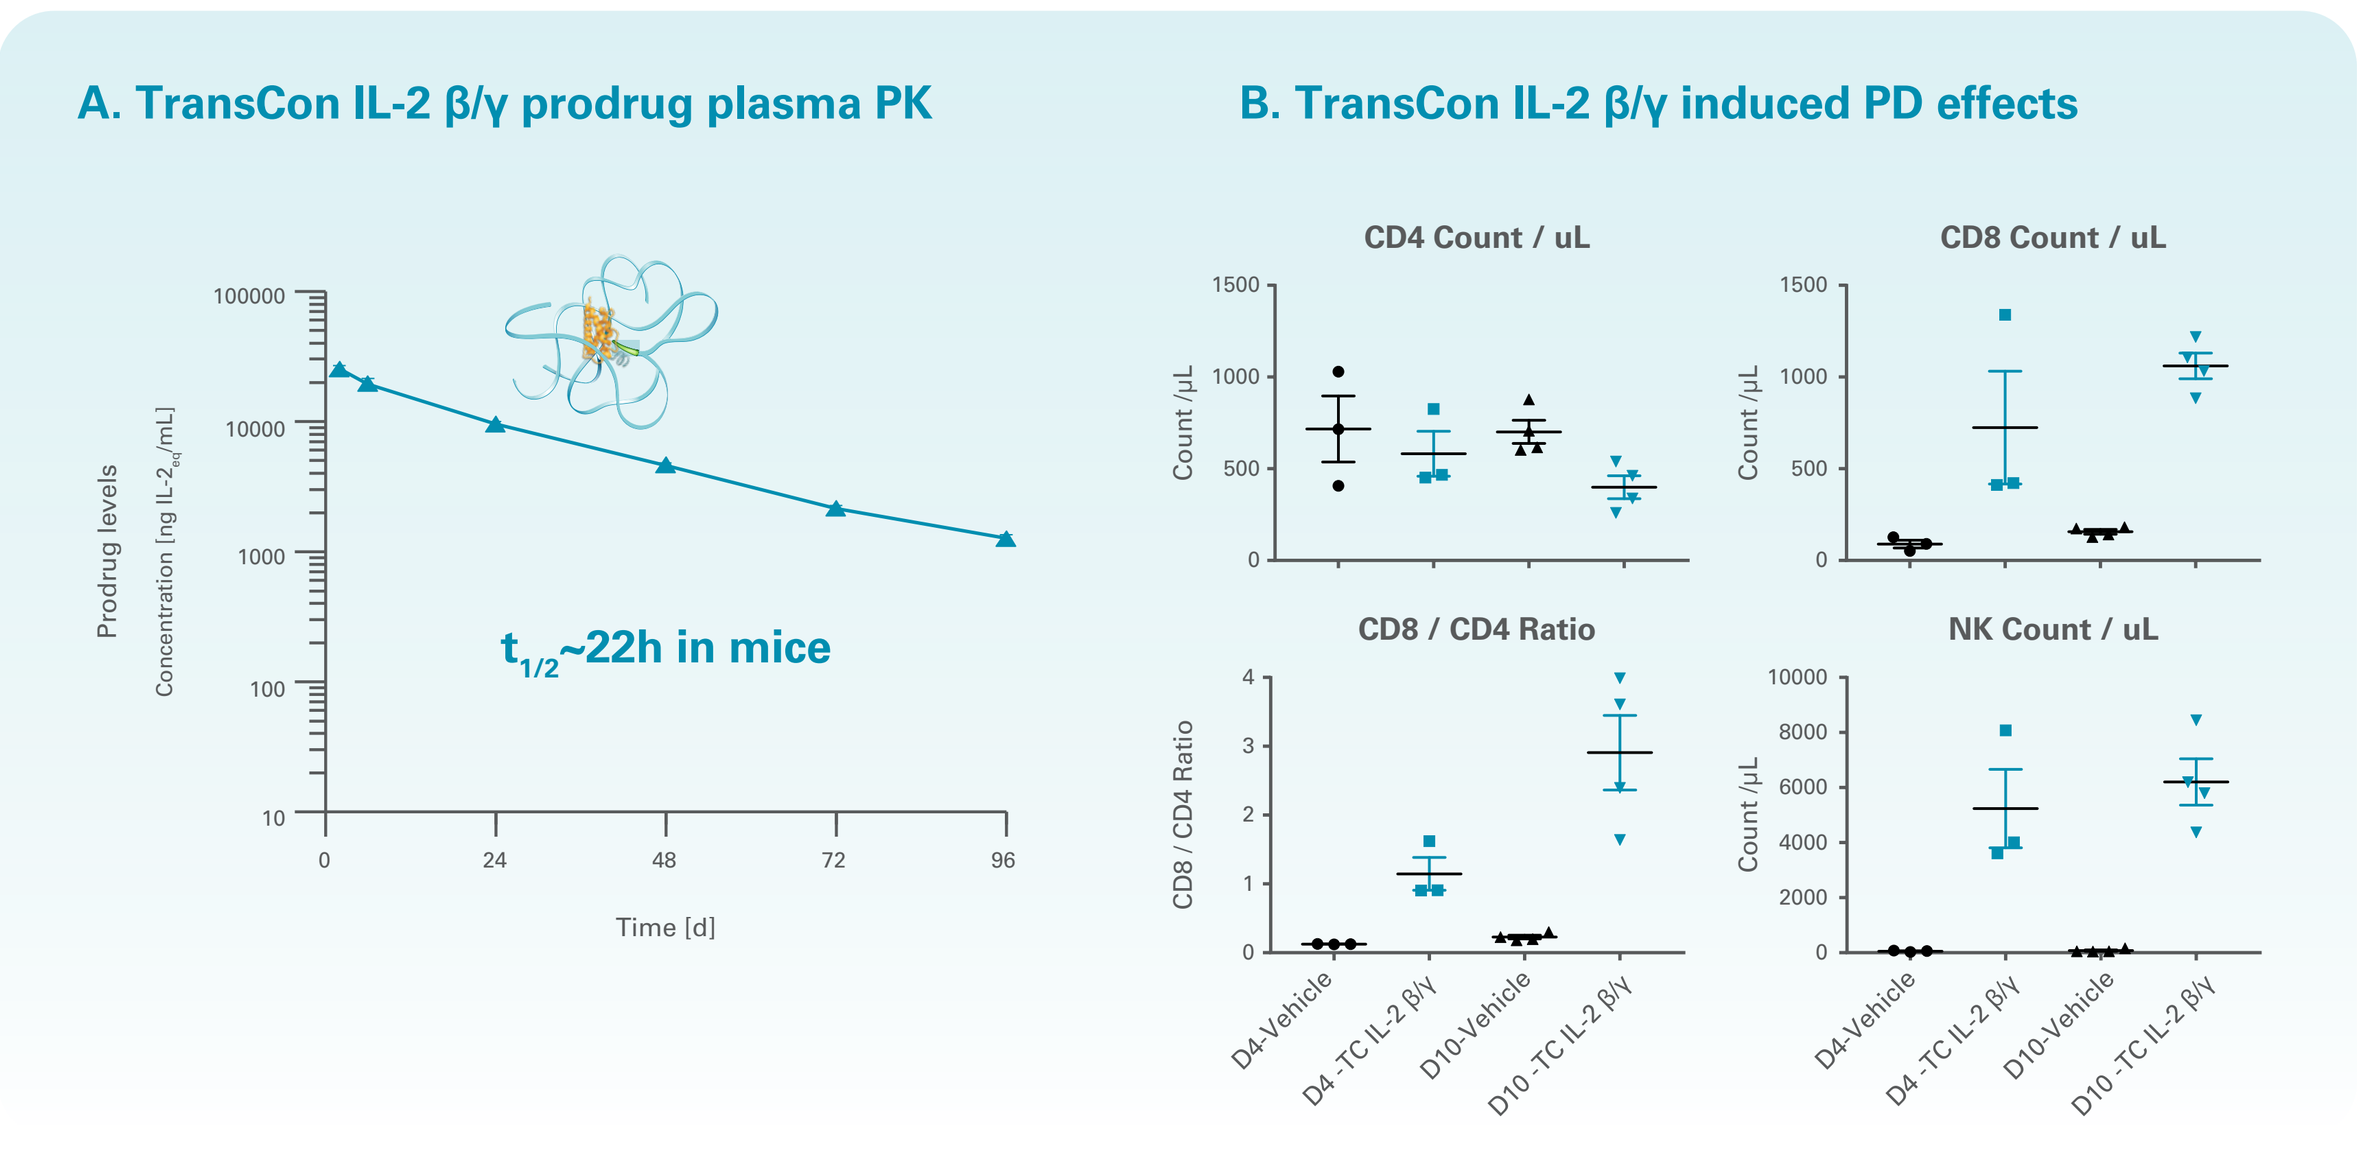
A. TransCon IL-2 β/γ prodrug plasma PK
Prodrug levels
$t_{1/2}$~22h in mice
Time [d]
B. TransCon IL-2 β/γ induced PD effects
CD4 Count / uL
CD8 Count / uL
CD8 / CD4 Ratio
NK Count / uL
D4-Vehicle
D4 -TC IL-2 β/γ
D10-Vehicle
D10 -TC IL-2 β/γ

A) Female BALB/C mice were administered 20 µg of TransCon IL-2 β/γ once via i.v. route and peripheral blood was analyzed from 0h - 96h post dose for plasma TransCon IL-2 β/γ prodrug levels by ELISA. These analyses suggested an *in vivo* TransCon IL-2 β/γ prodrug half life of approximately 22h in mice. B) Female BALB/C mice were administered 60 µg of TransCon IL-2 β/γ on Day 0 and Day 6 via i.v. route. Peripheral blood was drawn on Day 4 (D4) and Day 10 (D10) and stimulated with BD Leukocyte Activation Cocktail (PMA and Ionomycin) for 5h before analysis by flow cytometry. Data were acquired along with counting beads for quantitation of CD4+ T cell, CD8+ T cell and NK cell numbers (above).

Figure 5: TransCon IL-2 β/γ robustly activated CD8+ T cells and NK cells in mice

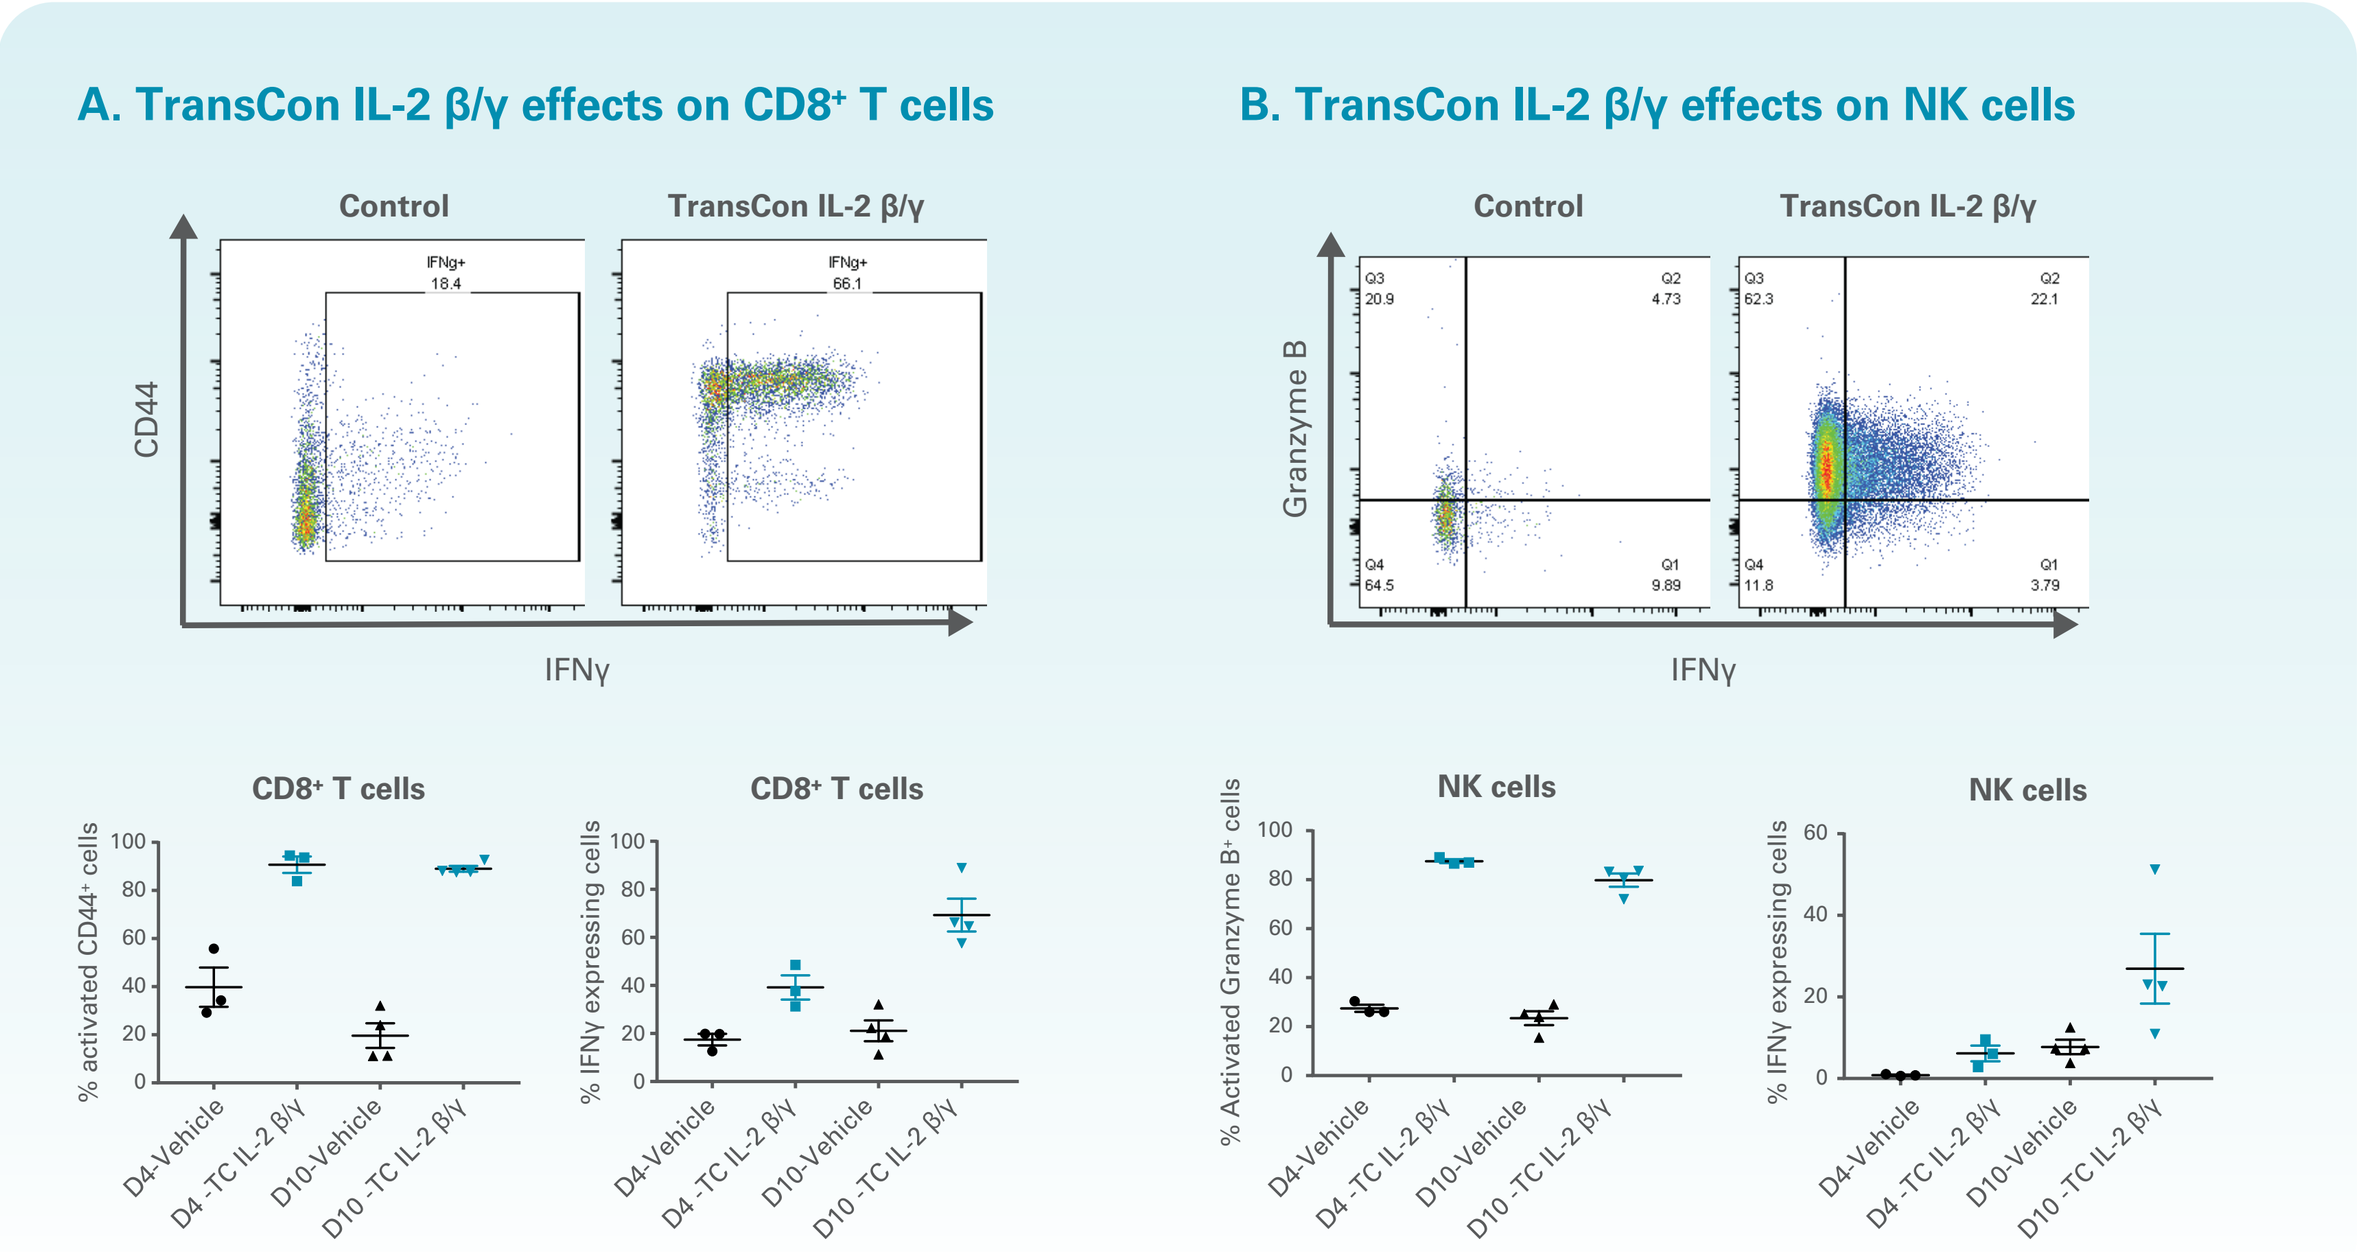
A. TransCon IL-2 β/γ effects on CD8+ T cells
B. TransCon IL-2 β/γ effects on NK cells
Control
TransCon IL-2 β/γ
CD44
Granzyme B
IFNγ
CD8+ T cells
NK cells
D4-Vehicle
D4 -TC IL-2 β/γ
D10-Vehicle
D10 -TC IL-2 β/γ

Female BALB/C mice were administered 60 µg of TransCon IL-2 β/γ on Day 0 and Day 6 via i.v. route. Peripheral blood was drawn on Day 4 (D4) and Day 10 (D10) and stimulated with BD Leukocyte Activation Cocktail for 5h to induce cytokine production before analysis by flow cytometry for activation markers and cytokines within lymphocyte populations. Example data from Day 10 are shown as flow cytometry plots (top). Percentages of activated CD44+ or IFNγ+ cells within CD8+ T cells (A) or percentages of activated Granzyme B+ or IFNγ+ cells within NK cells (B) are shown.

Figure 6: Single dose TransCon IL-2 β/γ induced robust lymphocyte but minimal eosinophil expansion in NHP when compared to aldesleukin

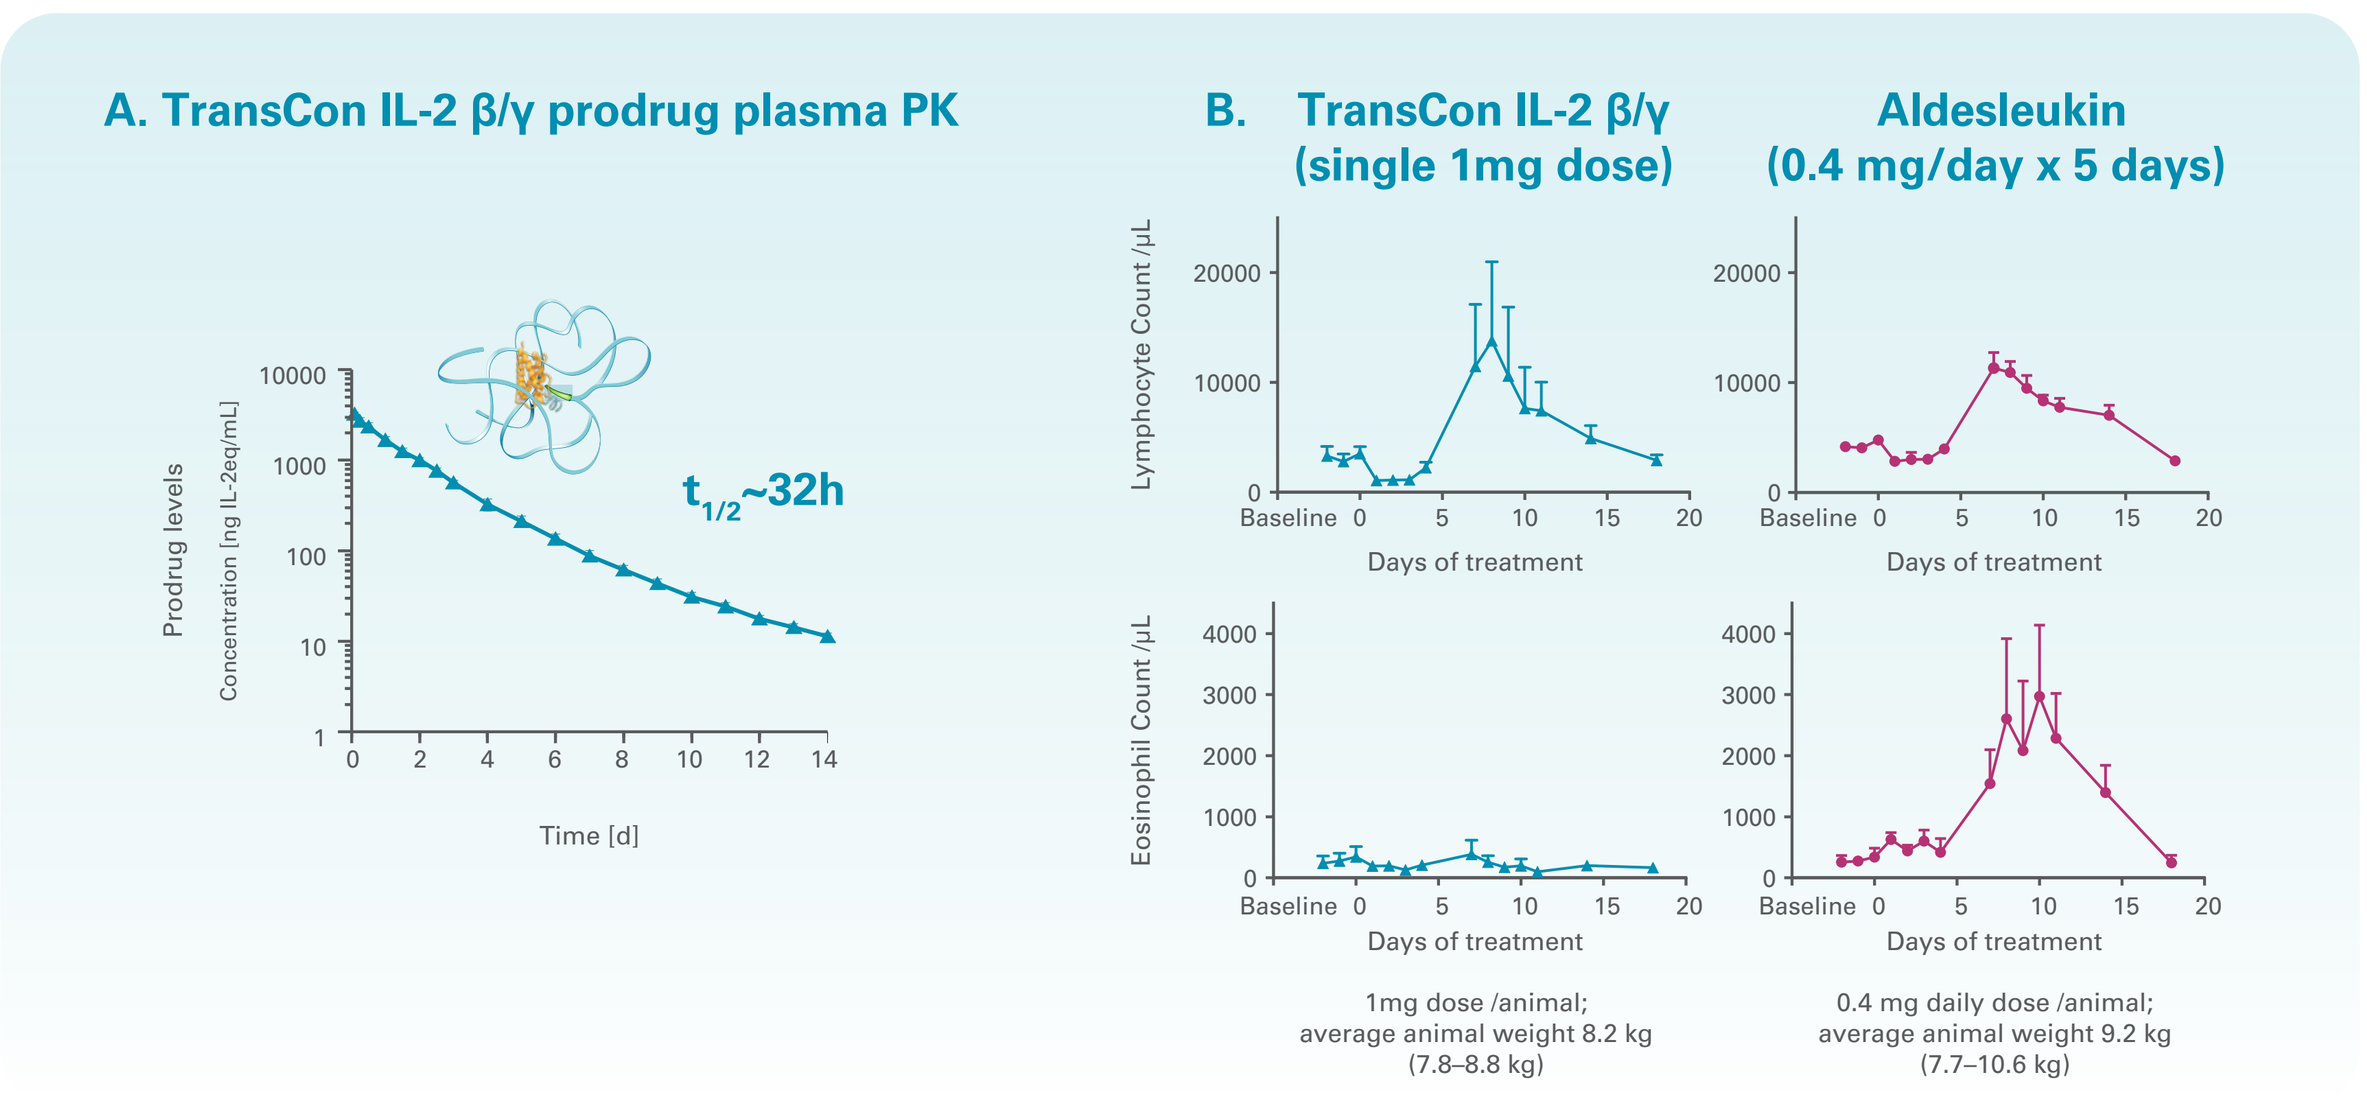
A. TransCon IL-2 β/γ prodrug plasma PK
$t_{1/2}$~32h
Prodrug levels
Time [d]
B. TransCon IL-2 β/γ (single 1mg dose)
Aldesleukin (0.4 mg/day x 5 days)
Lymphocyte Count /µL
Eosinophil Count /µL
Days of treatment
1mg dose /animal; average animal weight 8.2 kg (7.8–8.8 kg)
0.4 mg daily dose /animal; average animal weight 9.2 kg (7.7–10.6 kg)

Cynomolgus monkeys were administered with either 5 daily 0.4mg doses of aldesleukin (n=4) or a single 1 mg dose of TransCon IL-2 β/γ (n=4, measured in IL-2 equivalents) via i.v. route. Peripheral blood was acquired pre- and post-dosing and was analyzed for plasma TransCon IL-2 β/γ prodrug levels by ELISA (A) or for Complete Blood Counts (B) using a hemocytometer. A) Analysis of TransCon IL-2 β/γ plasma prodrug levels during the first week of exposure suggested an *in vivo* TransCon IL-2 β/γ prodrug half life of approximately 32h in NHP. B) Absolute Lymphocyte Count (top panels) and Absolute Eosinophil Count (bottom panels) values are shown as mean + SEM for each group. While either treatment with TransCon IL-2 β/γ or aldesleukin robustly induced increases in blood lymphocyte counts, treatment with aldesleukin but not TransCon IL-2 β/γ induced increases in eosinophil counts.

Figure 7: Single dose TransCon IL-2 β/γ induced minimal systemic endothelial cell injury and inflammation markers in NHP when compared to aldesleukin

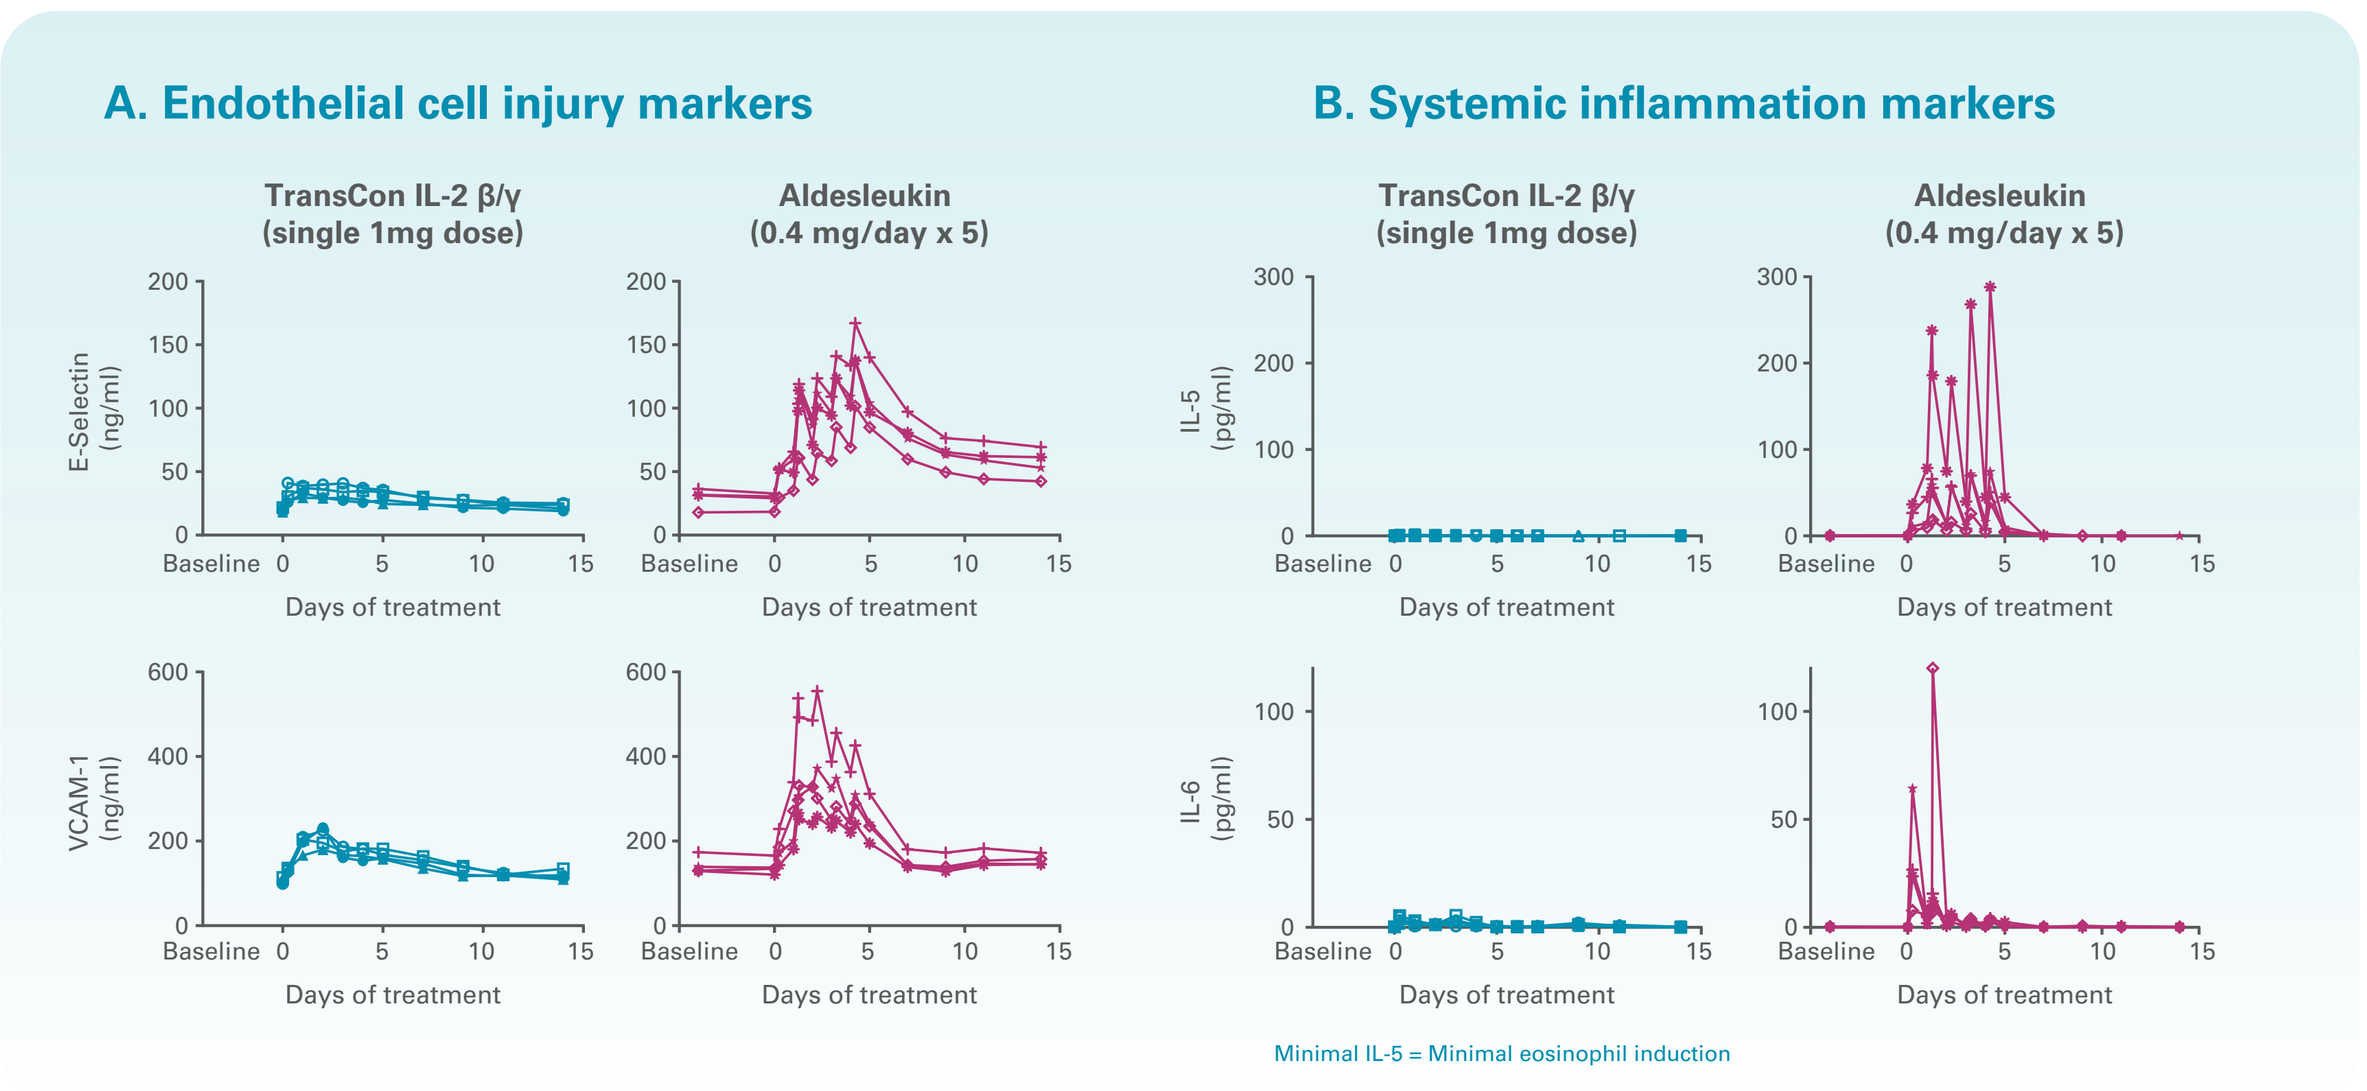
A. Endothelial cell injury markers
B. Systemic inflammation markers
TransCon IL-2 β/γ (single 1mg dose)
Aldesleukin (0.4 mg/day x 5)
E-Selectin (ng/ml)
VCAM-1 (ng/ml)
IL-5 (pg/ml)
IL-6 (pg/ml)
Days of treatment
Minimal IL-5 = Minimal eosinophil induction

Cynomolgus monkeys were administered with either 5 daily 0.4 mg doses of aldesleukin (n=4) or a single 1 mg dose of TransCon IL-2 β/γ (n=4). Serum was acquired pre- and post-dosing and was analyzed for soluble analytes using multiplexed immunodetection with electrochemiluminescence on the MSD platform Individual animal values are shown. A) In contrast to aldesleukin treatment, minimal induction of endothelial cell injury markers (E-Selectin, VCAM-1) was seen in serum after treatment with TransCon IL-2 β/γ. B) Similarly, minimal induction of systemic inflammatory markers such as IL-5 (eosinophil inflammation marker) and IL-6 (acute phase response marker) was seen in serum after treatment with TransCon IL-2 β/γ as opposed to treatment with aldesleukin.

Figure 8: Single dose TransCon IL-2 β/γ induced robust memory CD8+ T cell and NK expansion in NHP

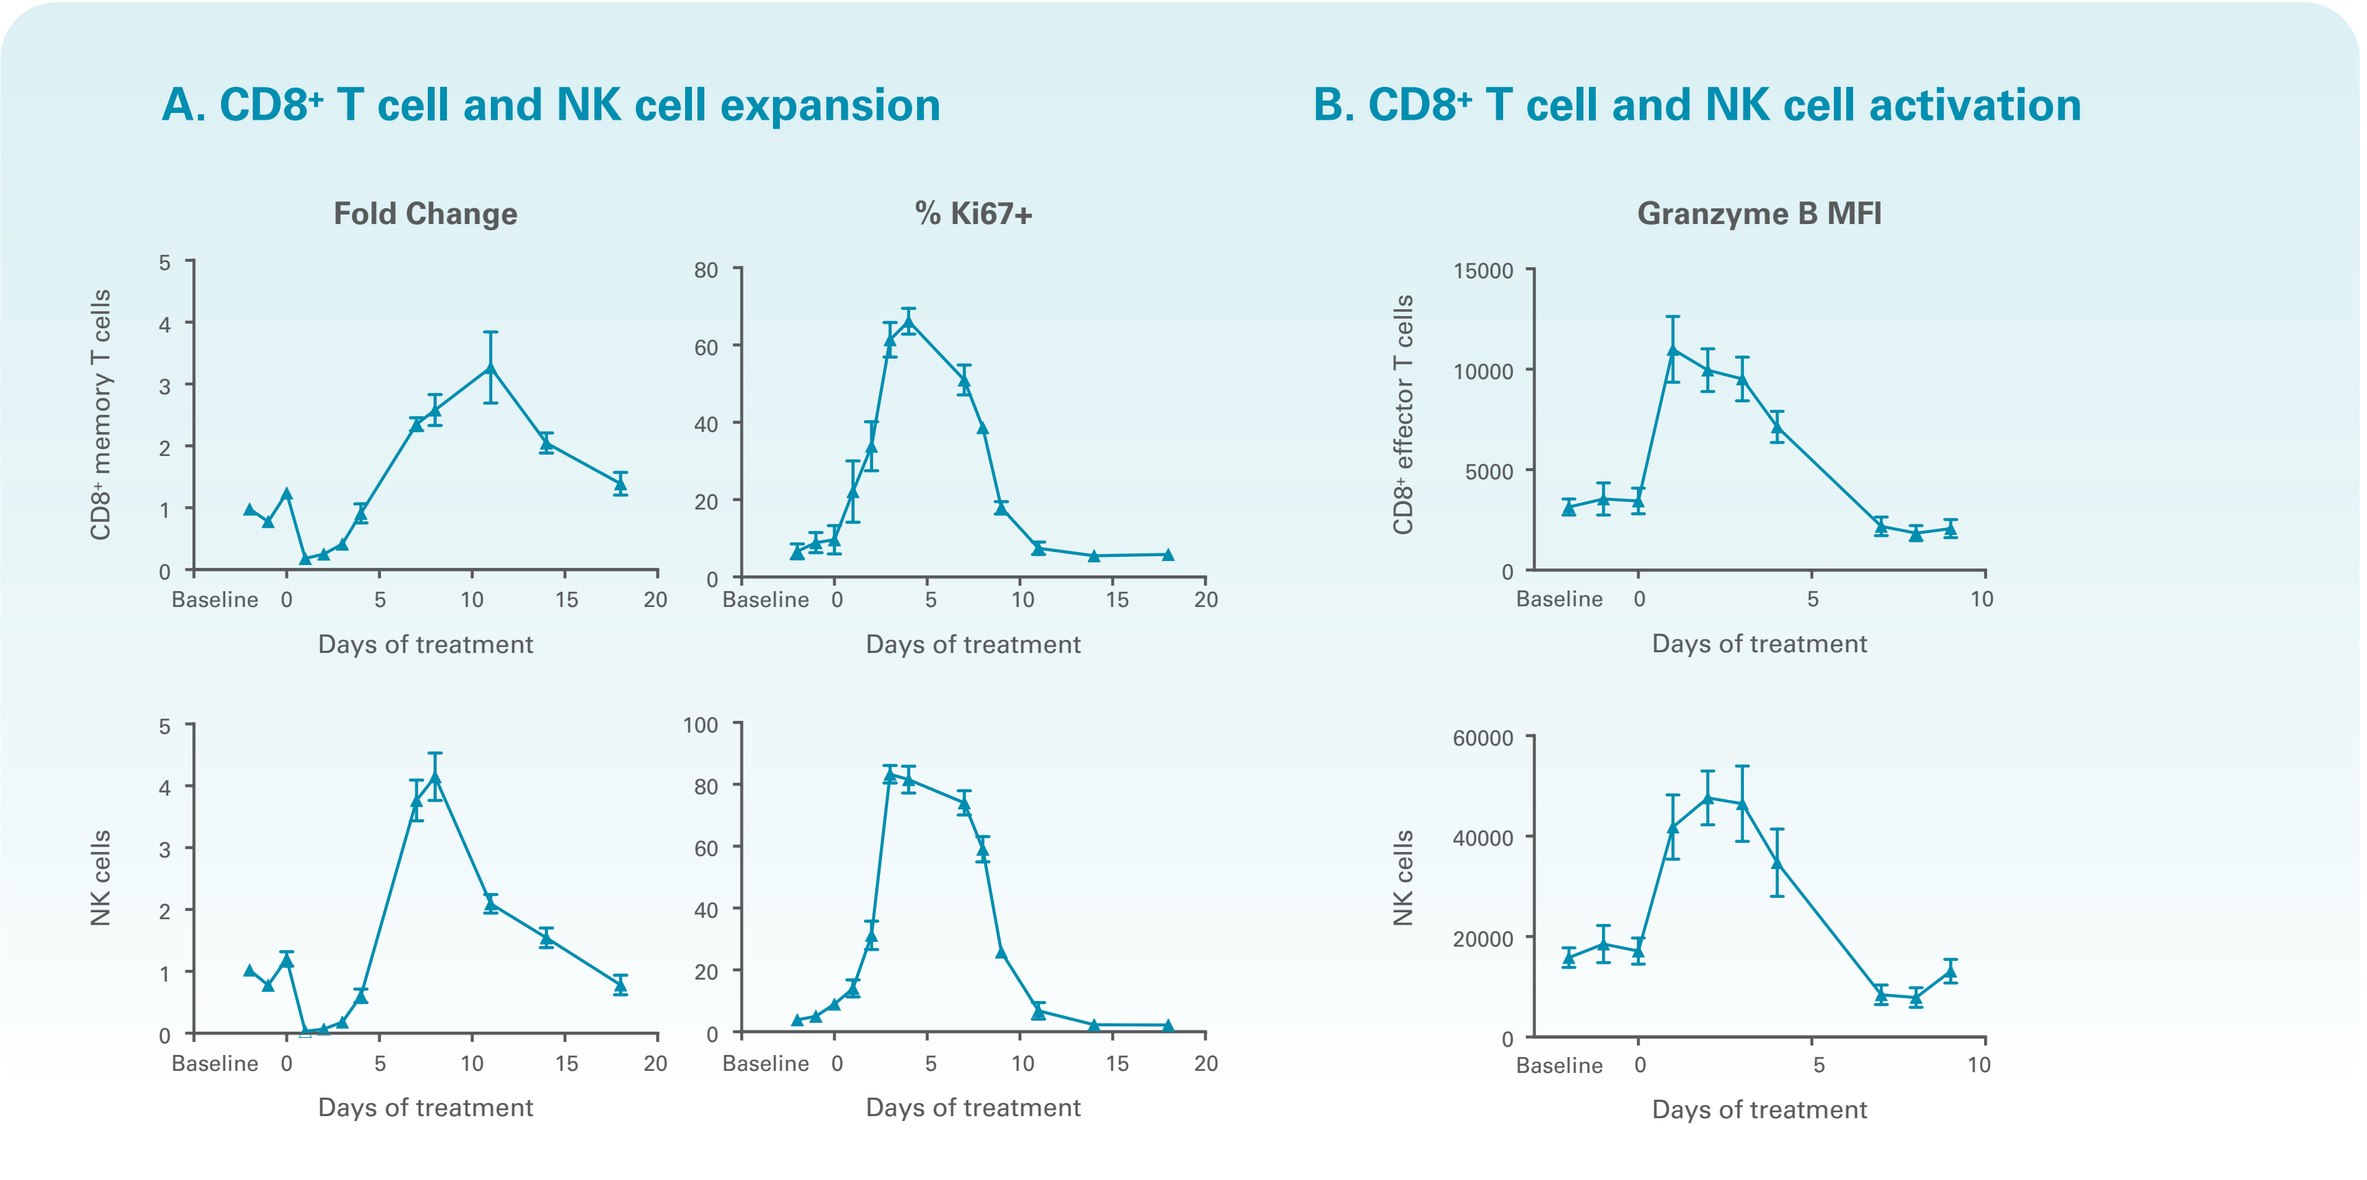
A. CD8+ T cell and NK cell expansion
B. CD8+ T cell and NK cell activation
Fold Change
% Ki67+
Granzyme B MFI
CD8+ memory T cells
NK cells
CD8+ effector T cells
Days of treatment

Cynomolgus monkeys (n=4) were treated with a single 1 mg dose of TransCon IL-2 β/γ and peripheral blood was acquired pre- and post-dosing and analyzed for lymphocyte populations and activation state by flow cytometry. Fold changes were calculated using lymphocyte subpopulation counts derived from flow cytometry data combined with Absolute Lymphocyte Counts. Analysis included gating for CD28- effector as well as CD28+ CD95+ CD8+ memory T cells. A) Shown are fold changes in the number of CD8+ memory T cells and NK cells (left), and increases in the percentage of proliferating Ki67+ CD8+ memory T cells and NK cells (middle). B) Shown are increases in the cytolytic effector molecule Granzyme B in CD8+ effector T cells and NK cells. Values are represented as mean +/- SEM for each group.

Figure 9: Single dose TransCon IL-2 β/γ induced selective lymphocyte expansion in NHP

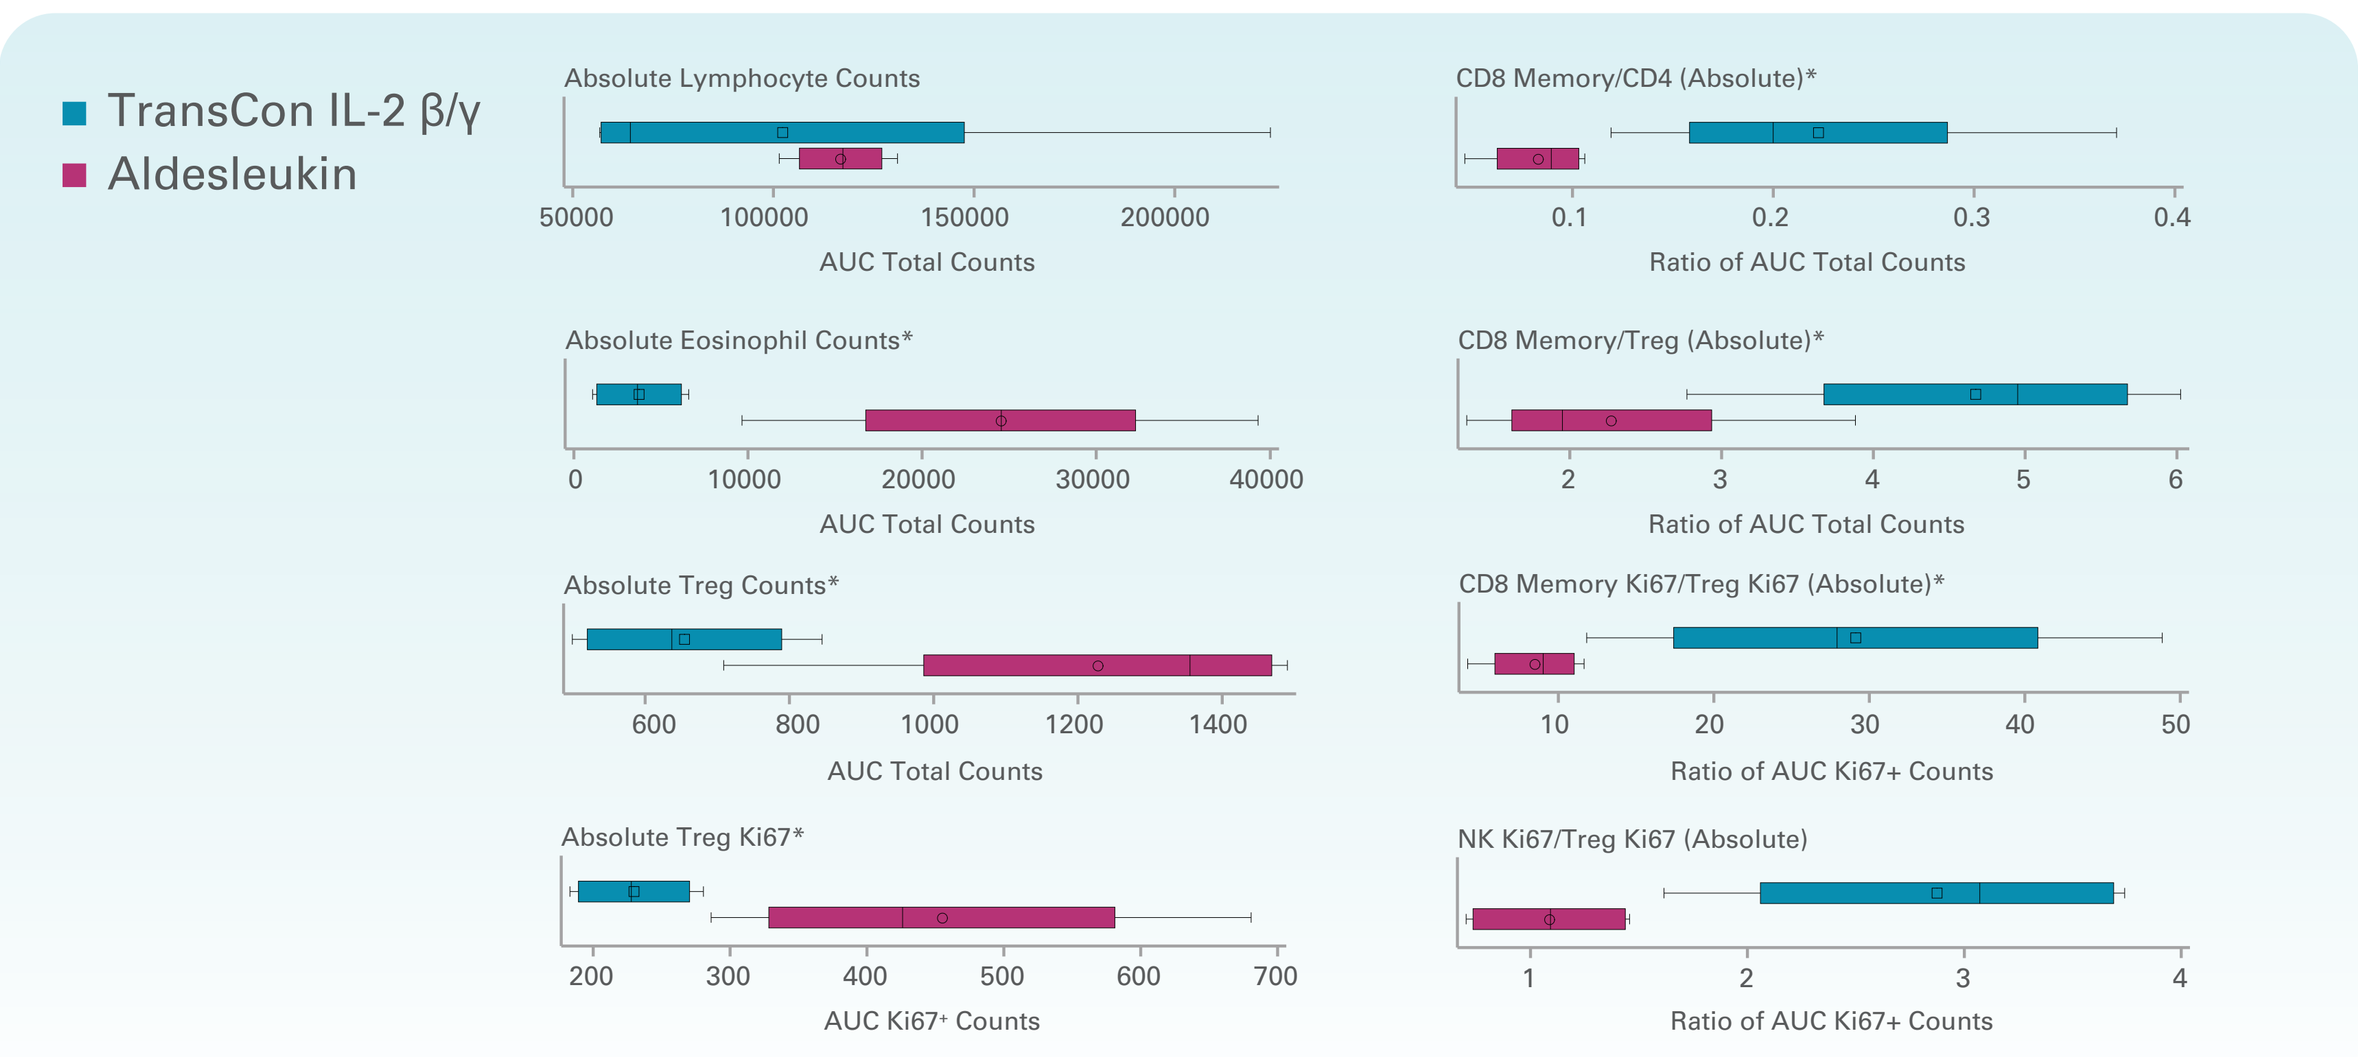
TransCon IL-2 β/γ
Aldesleukin
Absolute Lymphocyte Counts
Absolute Eosinophil Counts*
Absolute Treg Counts*
Absolute Treg Ki67*
AUC Total Counts
AUC Ki67+ Counts
CD8 Memory/CD4 (Absolute)*
CD8 Memory/Treg (Absolute)*
CD8 Memory Ki67/Treg Ki67 (Absolute)*
NK Ki67/Treg Ki67 (Absolute)
Ratio of AUC Total Counts
Ratio of AUC Ki67+ Counts

Flow cytometry based cell counts from experiments described in Figures 6-8 were further analyzed using an Area Under the Curve (AUC) analysis to quantify the total amount (area) of response over time. AUCs of various cell subsets were used to compare cellular responses following TransCon IL-2 β/γ (single dose of 1 mg/animal) or aldesleukin (0.4 mg/animal/day x 5). Shown are the AUC calculated values for selected populations (left) as well as the comparison of AUC values as ratios between multiple populations (right) as an indicator of relative expansion of one cell type over another under different treatment modalities. Metrics which were statistically significant ($p < 0.05$) between treatment groups using a Student's T-test (*) are marked.

SUMMARY

TransCon IL-2 β/γ demonstrated:

- Sustained release of a novel IL-2 variant (IL-2 β/γ) with selective binding and activation of IL-2Rβ/γ
- Long prodrug half-life in mice (~22h) and NHP (~32h)
- Robust expansion and activation of CD8+ T cells and NK cells in mice
- Robust lymphocyte expansion without eosinophil expansion in NHP
- Minimal induction of systemic acute phase response inflammatory markers and endothelial damage markers as compared to aldesleukin
- Selective expansion of CD8+ T cells and NK subsets in mice and NHP *in vivo*, consistent with receptor bias observed *in vitro*
- No dose limiting toxicities observed in NHPs; the maximum tolerated dose was not reached

Next steps:

- Future studies will test increased doses and combinations with other immunotherapies

CONCLUSIONS

TransCon IL-2 β/γ has the clinical potential to:

- Deliver sustained release of receptor-biased IL-2 avoiding high C_{max} exposure, expected to support every 3 week dosing
- Selectively expand and activate cytolytic lymphocytes with anti-tumor functions after a single dose with reduced risk of endothelial cell injury
- Offer optimized efficacy/safety ratio as a monotherapy and when combined with other systemic and intratumoral oncology therapeutics

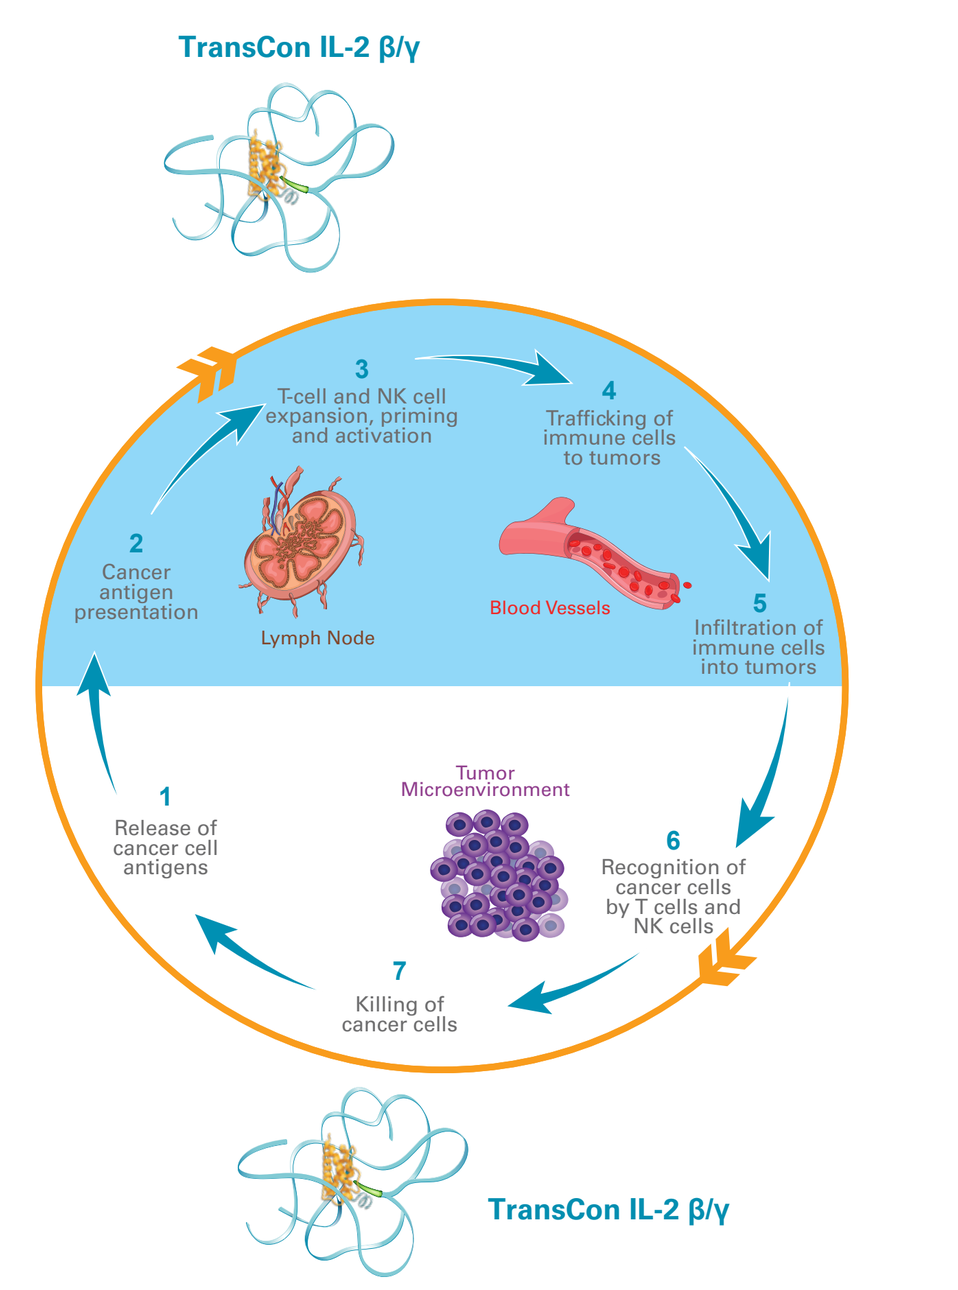
TransCon IL-2 β/γ
1 Release of cancer cell antigens
2 Cancer antigen presentation
3 T-cell and NK cell expansion, priming and activation
4 Trafficking of immune cells to tumors
5 Infiltration of immune cells into tumors
6 Recognition of cancer cells by T cells and NK cells
7 Killing of cancer cells
Lymph Node
Blood Vessels
Tumor Microenvironment
TransCon IL-2 β/γ

REFERENCES:
[1]Harding et al. Cytokine: X , 2019 [2]Rand et al., Journal of Clinical Investigation, 1991 [3]Van Haelst Pisani C et al., Blood, 1991, [4]Krieg et al. PNAS 2010, [5]Van Gool et al. Blood, 2014 [6]Konrad et al. Cancer Research, 1990.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group
© June 2020 Ascendis Pharma A/S