Experiences of Parents of Children With Achondroplasia: Impacts on Quality of Life

Kathryn M. Pfeiffer[1], Meryl Brod[1], Dorthe Viuff[2], Sho Ota[3], Jill Gianettoni[3], Jonathan A. Leff[3]

[1]The Brod Group, [2]Ascendis Pharma A/S, [3]Ascendis Pharma, Inc.

BACKGROUND

- The clinical complications and medical impacts of achondroplasia (ACH) in children are well studied and frequently include recurrent ear infections, sleep apnea, hearing loss, teeth crowding, speech delay, and delayed developmental milestones, including gross motor and fine motor[1–5]
- Little is known about how having a child with ACH impacts parents' experiences and quality of life
 - Research has shown that at the age of 7 years, many children with ACH continue to require minimal to moderate parent/caregiver assistance with self-care, and some children still require supervision in social settings[5]
 - Evidence also suggests that parents may experience emotional and other impacts at the time of their child's diagnosis[6]

OBJECTIVE

The purpose of the study was to investigate the impacts of having a child aged 2 to <12 years with ACH on parents' general quality of life, including parents' caretaking and work responsibilities, emotional well-being, and social well-being.

METHODS

The qualitative research study design was based on an adapted grounded theory approach.

Based on a literature review and clinical expert interviews, a semi-structured interview guide was developed to elicit parents' experiences related to ACH.

It should be noted that this study was part of a larger study of parents of children with ACH under 18 years of age, and this study focused only on parents of children aged 2 to <12 years.

Inclusion criteria:

- adult aged 18 years or older
- able to read, write, and speak English (in the United States [US]) or Spanish (in Spain)
- parent of a child (<18 years of age) diagnosed with ACH; and
- actively involved in the child's care

Exclusion criteria:

- A cognitive impairment or other medical condition, including psychiatric conditions, that would affect a participant's ability to participate in a telephone interview or focus group

Individual telephone interviews and 1 parent focus group were conducted in the US and Spain with 36 parents of children aged 2 to <12 years with ACH.

Telephone interviews lasted approximately 1 hour, and the focus group lasted 2 hours.

The interviews/focus group were conducted in English (US) or Spanish (Spain), transcribed verbatim, and translated to English if necessary.

Interview and focus group transcripts were analyzed for content and coded by themes using a qualitative analysis software program.

RESULTS

Parent Participant Sample Description

Participant sample characteristics are shown in Table 1.

- Average age of parents was 41.5 years (SD, 6.6; range, 32-68)
- Thirty-one parents were mothers (86.1%), and 5 parents were fathers (13.9%)
- Most participants were married (80.6%, n=29), 8.3% were partnered (n=3), 5.6% were divorced (n=2), and 5.6% were single (n=2)
- Seven parents (19.4%), all residing in the US, also had a diagnosis of ACH

Table 1. Parent participant demographic characteristics

	Spain (n=11)	US (n=25)	Total (N=36)
Age, mean(SD)	40.4(3.1)	42.0(7.6)	41.5(6.6)
(range)	(35-43)	(32-68)	(32-68)
Relationship to child, n(%)			
mother	8(72.7)	23(92.0)	31(86.1)
father	3(27.3)	2(8.0)	5(13.9)
Marital status, n(%)			
single	2(18.2)	0	2(5.6)
married	6(54.5)	23(92.0)	29(80.6)
partnered	3(27.3)	0	3(8.3)
divorced	0	2(8.0)	2(5.6)
Education, n(%)			
less than high school	2(18.2)	1(4.0)	3(8.3)
high school or equivalent	4(36.4)	2(8.0)	6(16.7)
college degree	5(45.5)	12(48.0)	17(47.2)
post-graduate school	0	10(40.0)	10(27.8)
Work status, n(%)			
full-time	6(54.5)	10(40.0)	16(44.4)
part-time	3(27.3)	3(12.0)	6(16.7)
student	0	2(8.0)	2(5.6)
retired	0	1(4.0)	1(2.8)
not working (other)	2(18.2)	9(36.0)	11(30.6)
Parent has ACH			
n(%) yes	0	7(28.0)	7(19.4)

Percentages may not add to 100 due to rounding. ACH = achondroplasia; SD = standard deviation.

Demographic/Health Characteristics for Children of Parent Participants

Demographic/health characteristics for the children of parent participants are shown in Table 2.

- 30.6% of parent participants (n=11) had children aged 2 to <5 years with ACH, 36.1% of parents (n=13) had children aged 5 to <9 years, and 33.3% of parents (n=12) had children aged 9 to <12 years
- Nineteen children were female (52.8%) and 17 were male (47.2%)
- Twelve parents reported child's health status as "excellent" (33.3%), 14 reported as "very good" (38.9%), 7 (19.4%) reported as "good," and 3 (8.3%) reported as "fair"

RESULTS

Table 2. Demographic/health characteristics of children of parent participants

	Spain (n=11)	US (n=25)	Total (N=36)
Child age, n(%)			
2 to <5 years	5(45.5)	6(24.0)	11(30.6)
5 to <9 years	4(36.4)	9(36.0)	13(36.1)
9 to <12 years	2(18.2)	10(40.0)	12(33.3)
Child gender, n(%)			
female	7(63.6)	12(48.0)	19(52.8)
male	4(36.4)	13(52.0)	17(47.2)
Health status (parent-reported), n(%)			
excellent	3(27.3)	9(36.0)	12(33.3)
very good	3(27.3)	11(44.0)	14(38.9)
good	3(27.3)	4(16.0)	7(19.4)
fair	2(18.2)	1(4.0)	3(8.3)
Age/time diagnosed with ACH, n(%)			
in utero	9(81.8)	12(48.0)	21(58.3)
at birth	1(9.1)	4(16.0)	5(13.9)
<2 months of age	1(9.1)	2(8.0)	3(8.3)
2-6 months of age	0	5(20.0)	5(13.9)
unknown (adopted)	0	2(8.0)	2(5.6)

Percentages may not add to 100 due to rounding. ACH = achondroplasia; SD = standard deviation.

Impacts on Parents' Caretaking Responsibilities

The most frequently reported caretaking responsibilities included:

- Managing child's medical care (eg, doctor appointments, treatment decisions; 92%, n=33)
- Helping child with self-care (eg, toileting, bathing, dressing; 67%, n=24)
- Advocating for child (eg, for school accommodations, educating others; 64%, n=23)
- Providing assistance to child (eg, reaching objects; 56%, n=20)
- Providing support/guidance to child for living with/managing ACH (47%, n=17)
- Monitoring child (eg, for safety; 47%, n=17)

Figure 1. Impacts on Parents' Caretaking Responsibilities

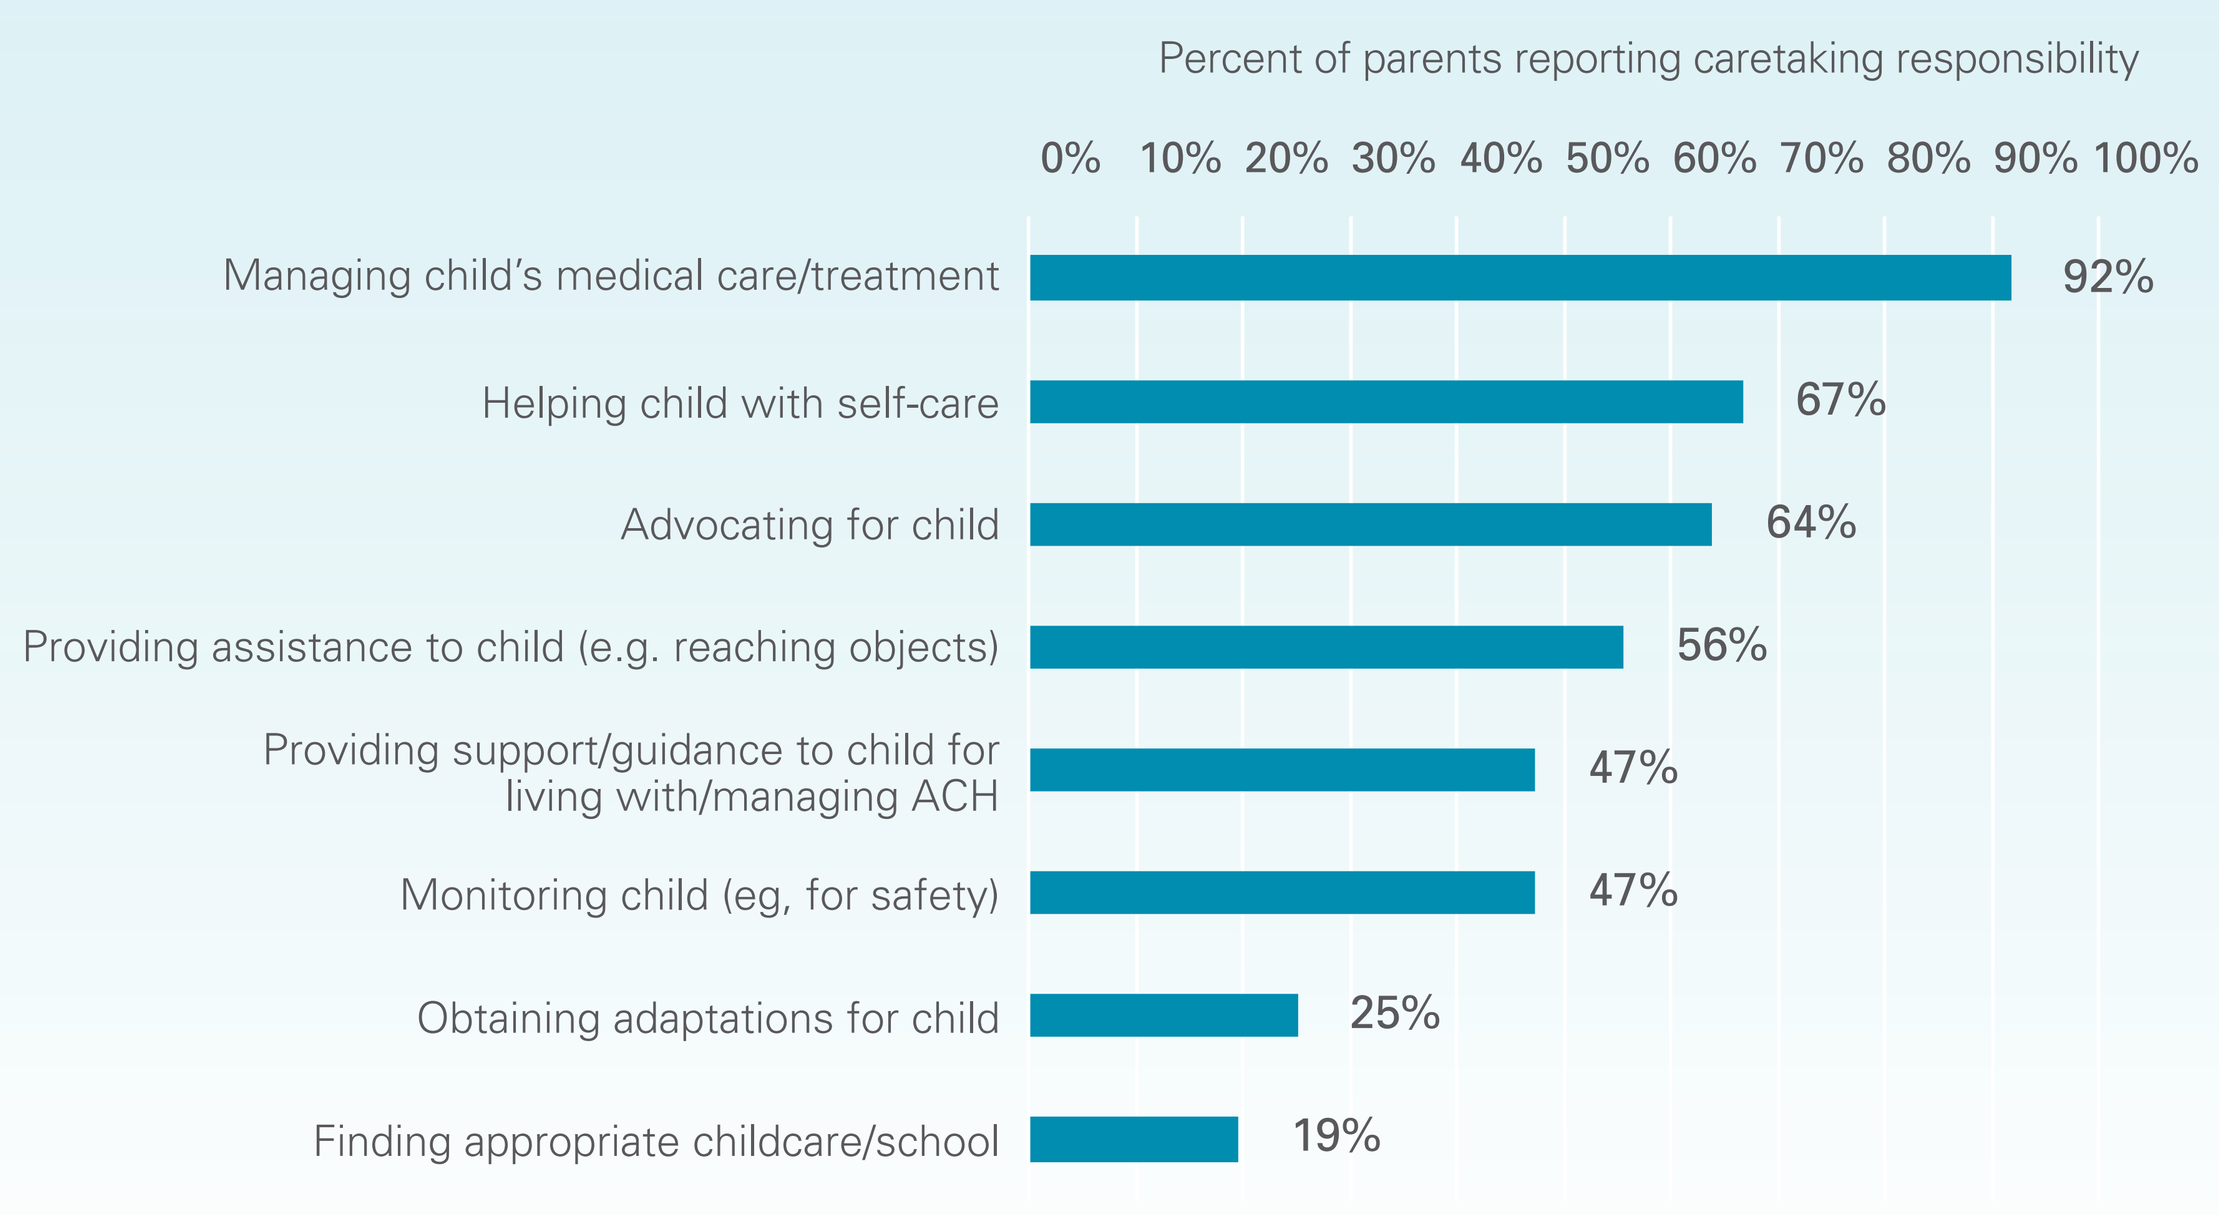
Percent of parents reporting caretaking responsibility
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Managing child's medical care/treatment 92%
Helping child with self-care 67%
Advocating for child 64%
Providing assistance to child (e.g. reaching objects) 56%
Providing support/guidance to child for living with/managing ACH 47%
Monitoring child (eg, for safety) 47%
Obtaining adaptations for child 25%
Finding appropriate childcare/school 19%

N=36. ACH = achondroplasia.

Impacts on Work

The most often reported impacts on parents' work or employment included:

- Missed work time to care for child (50%, n=18)
- Changed work schedule (19%, n=7)
- Discontinued work (14%, n=5)

Figure 2. Impacts on Parents' Work or Employment

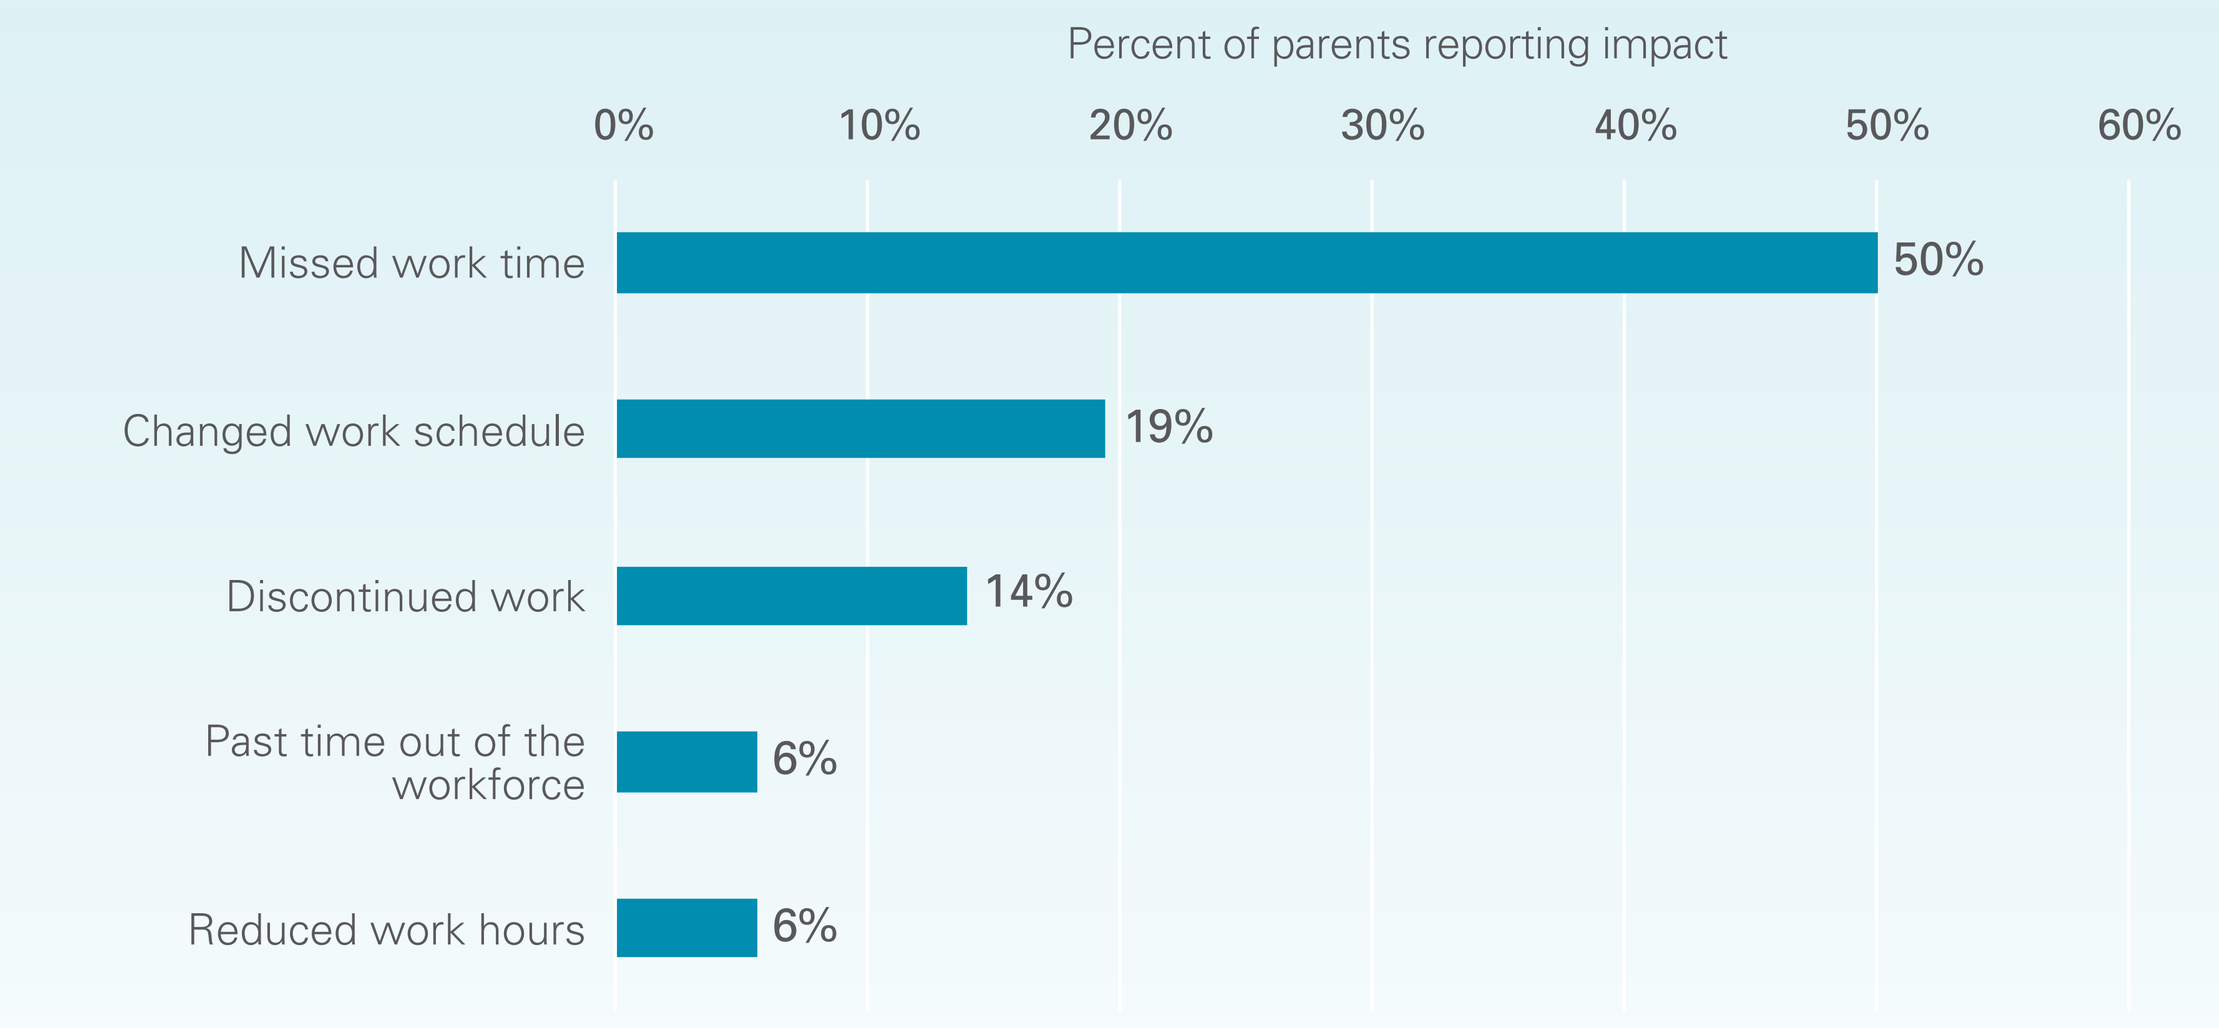
Percent of parents reporting impact
0% 10% 20% 30% 40% 50% 60%
Missed work time 50%
Changed work schedule 19%
Discontinued work 14%
Past time out of the workforce 6%
Reduced work hours 6%

N=36.

Impacts on Parents' Emotional Well-being

Frequently reported impacts of having a child with ACH on parents' emotional well-being included:

- Worry about child's future (75%, n=27)
- Worry about child's physical health (67%, n=24)
- Concerns about child's safety (50%, n=18)
- Feeling stressed/overwhelmed (44%, n=16)
- Worry about child's social well-being (42%, n=15)
- Initial shock/grief following diagnosis (42%, n=15)
- Concerns about child's ability to function independently (33%, n=12)

Figure 3. Impacts on Parents' Emotional Well-being

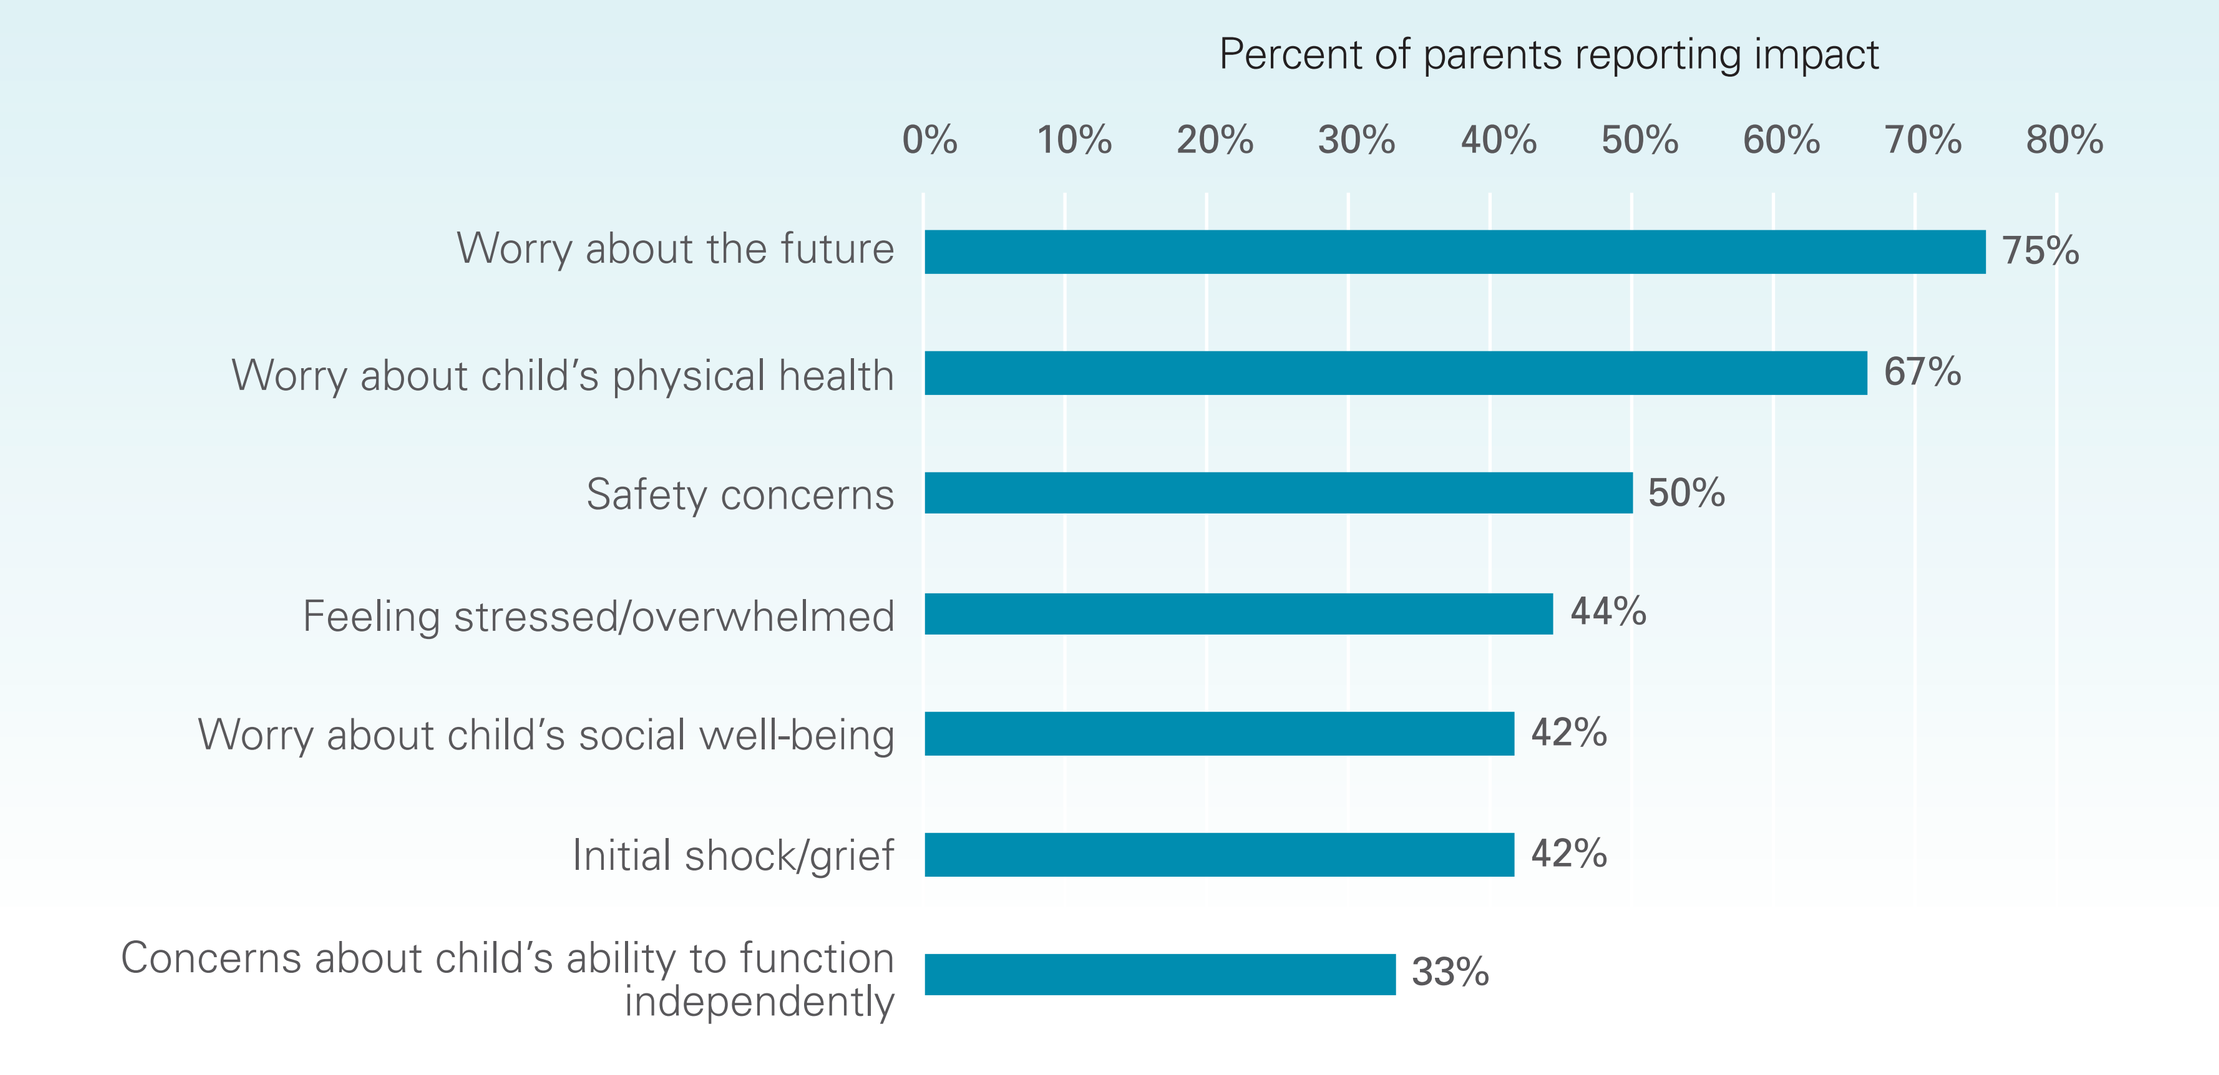
Percent of parents reporting impact
0% 10% 20% 30% 40% 50% 60% 70% 80%
Worry about the future 75%
Worry about child's physical health 67%
Safety concerns 50%
Feeling stressed/overwhelmed 44%
Worry about child's social well-being 42%
Initial shock/grief 42%
Concerns about child's ability to function independently 33%

N=36.

RESULTS

Impacts on Parents' Social Well-being

The most frequently reported impact on parents' social well-being were:

- Friendships, support, and social activities through the dwarfism community/advocacy organizations (69%, n=25)
- Limit social or other activities, such as hobbies (28%, n=10)
- Support from family and/or friends (28%, n=10)

Figure 4. Impacts on Parents' Social Well-being

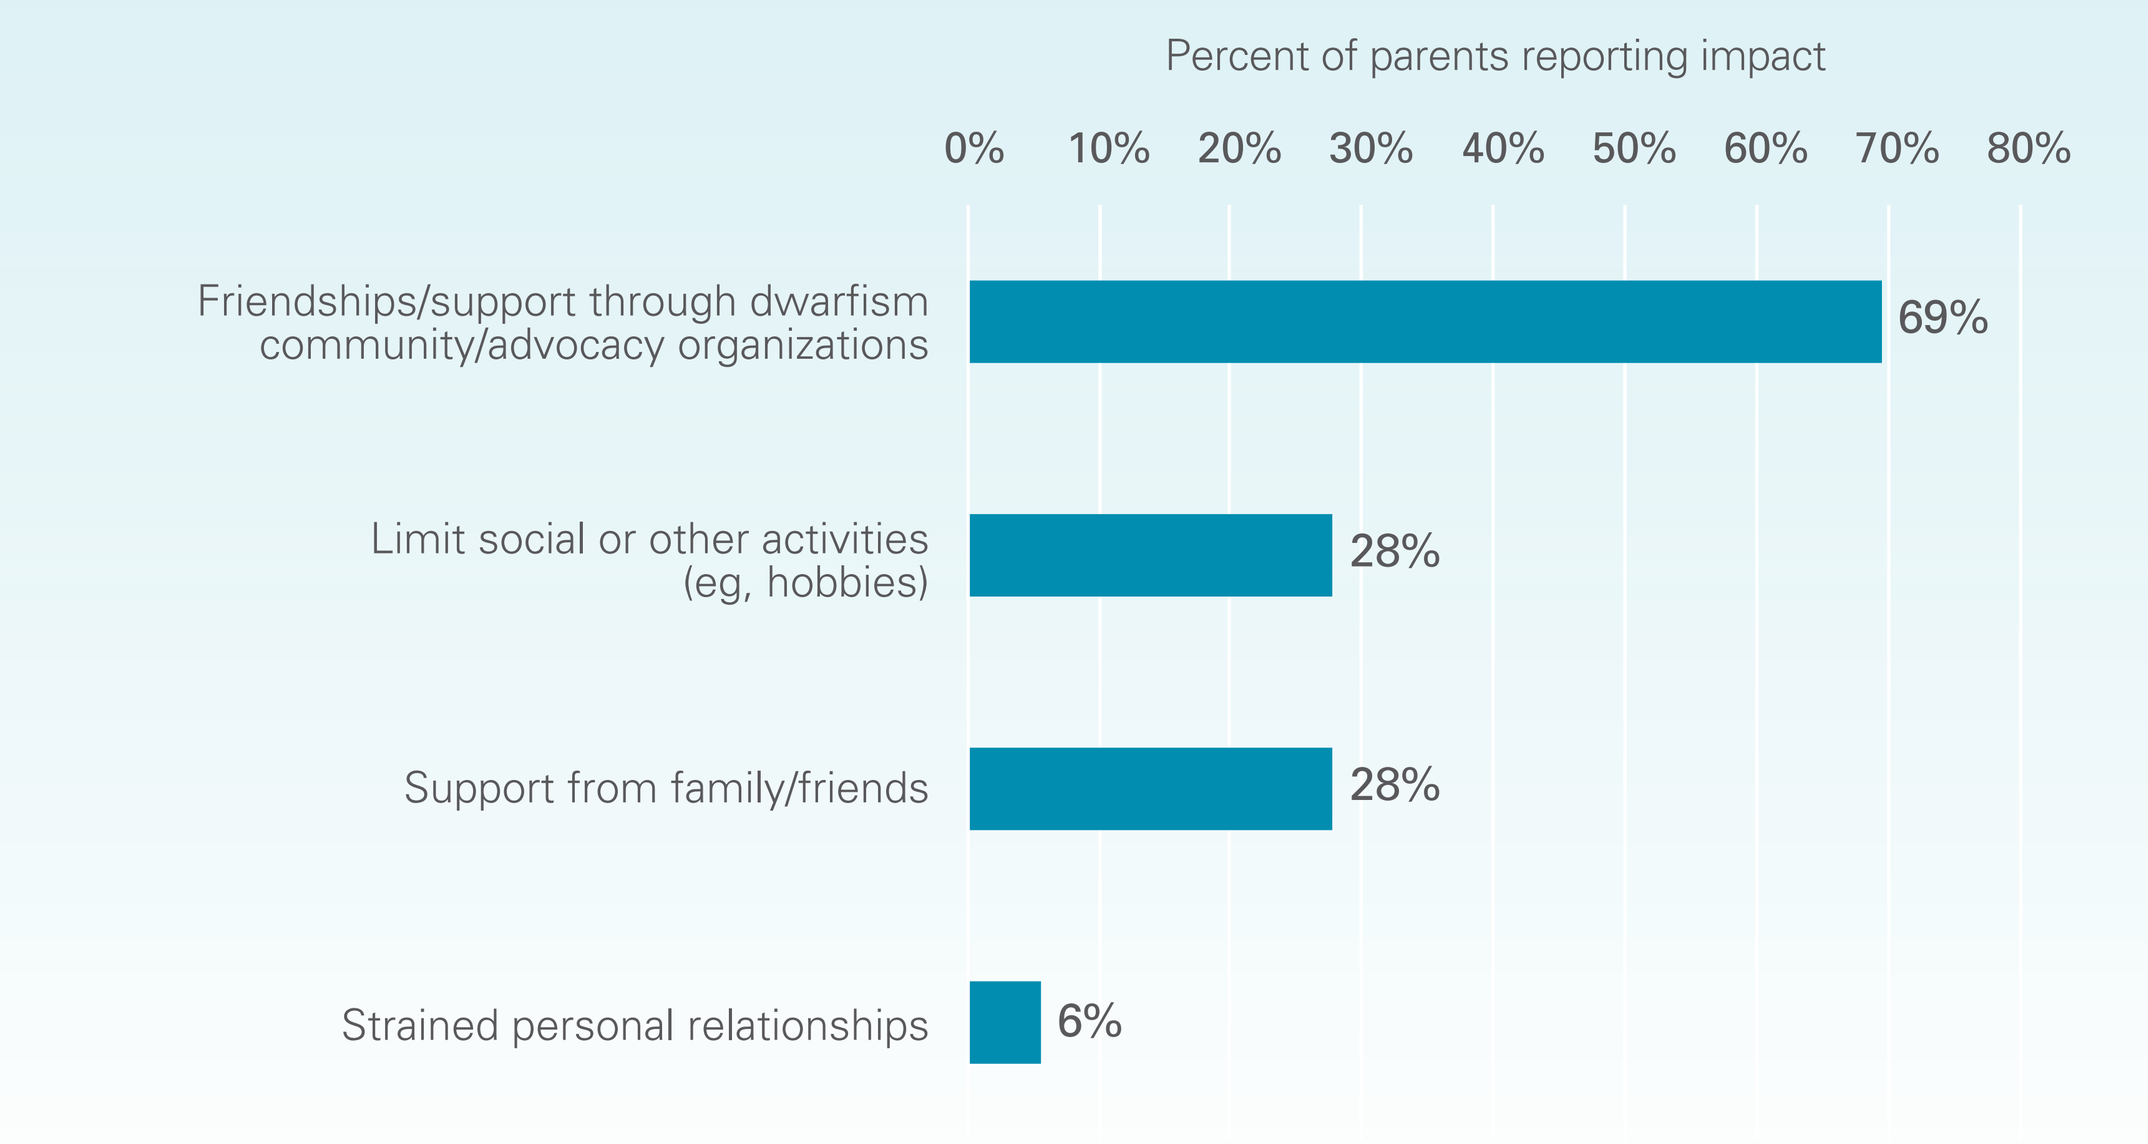
Percent of parents reporting impact
0% 10% 20% 30% 40% 50% 60% 70% 80%
Friendships/support through dwarfism community/advocacy organizations 69%
Limit social or other activities (eg, hobbies) 28%
Support from family/friends 28%
Strained personal relationships 6%

N=36.

Impacts on Families

The most frequently reported impacts on families included:

- Strain on family (eg, having less time; 56%, n=20)
- Family travel/vacation plans (eg, prioritizing advocacy organization meetings/events; 53%; n=19)
- Limiting or adapting family activities (42%, n=15)
- Increased family closeness (17%, n=6)

Figure 5. Impacts on Families

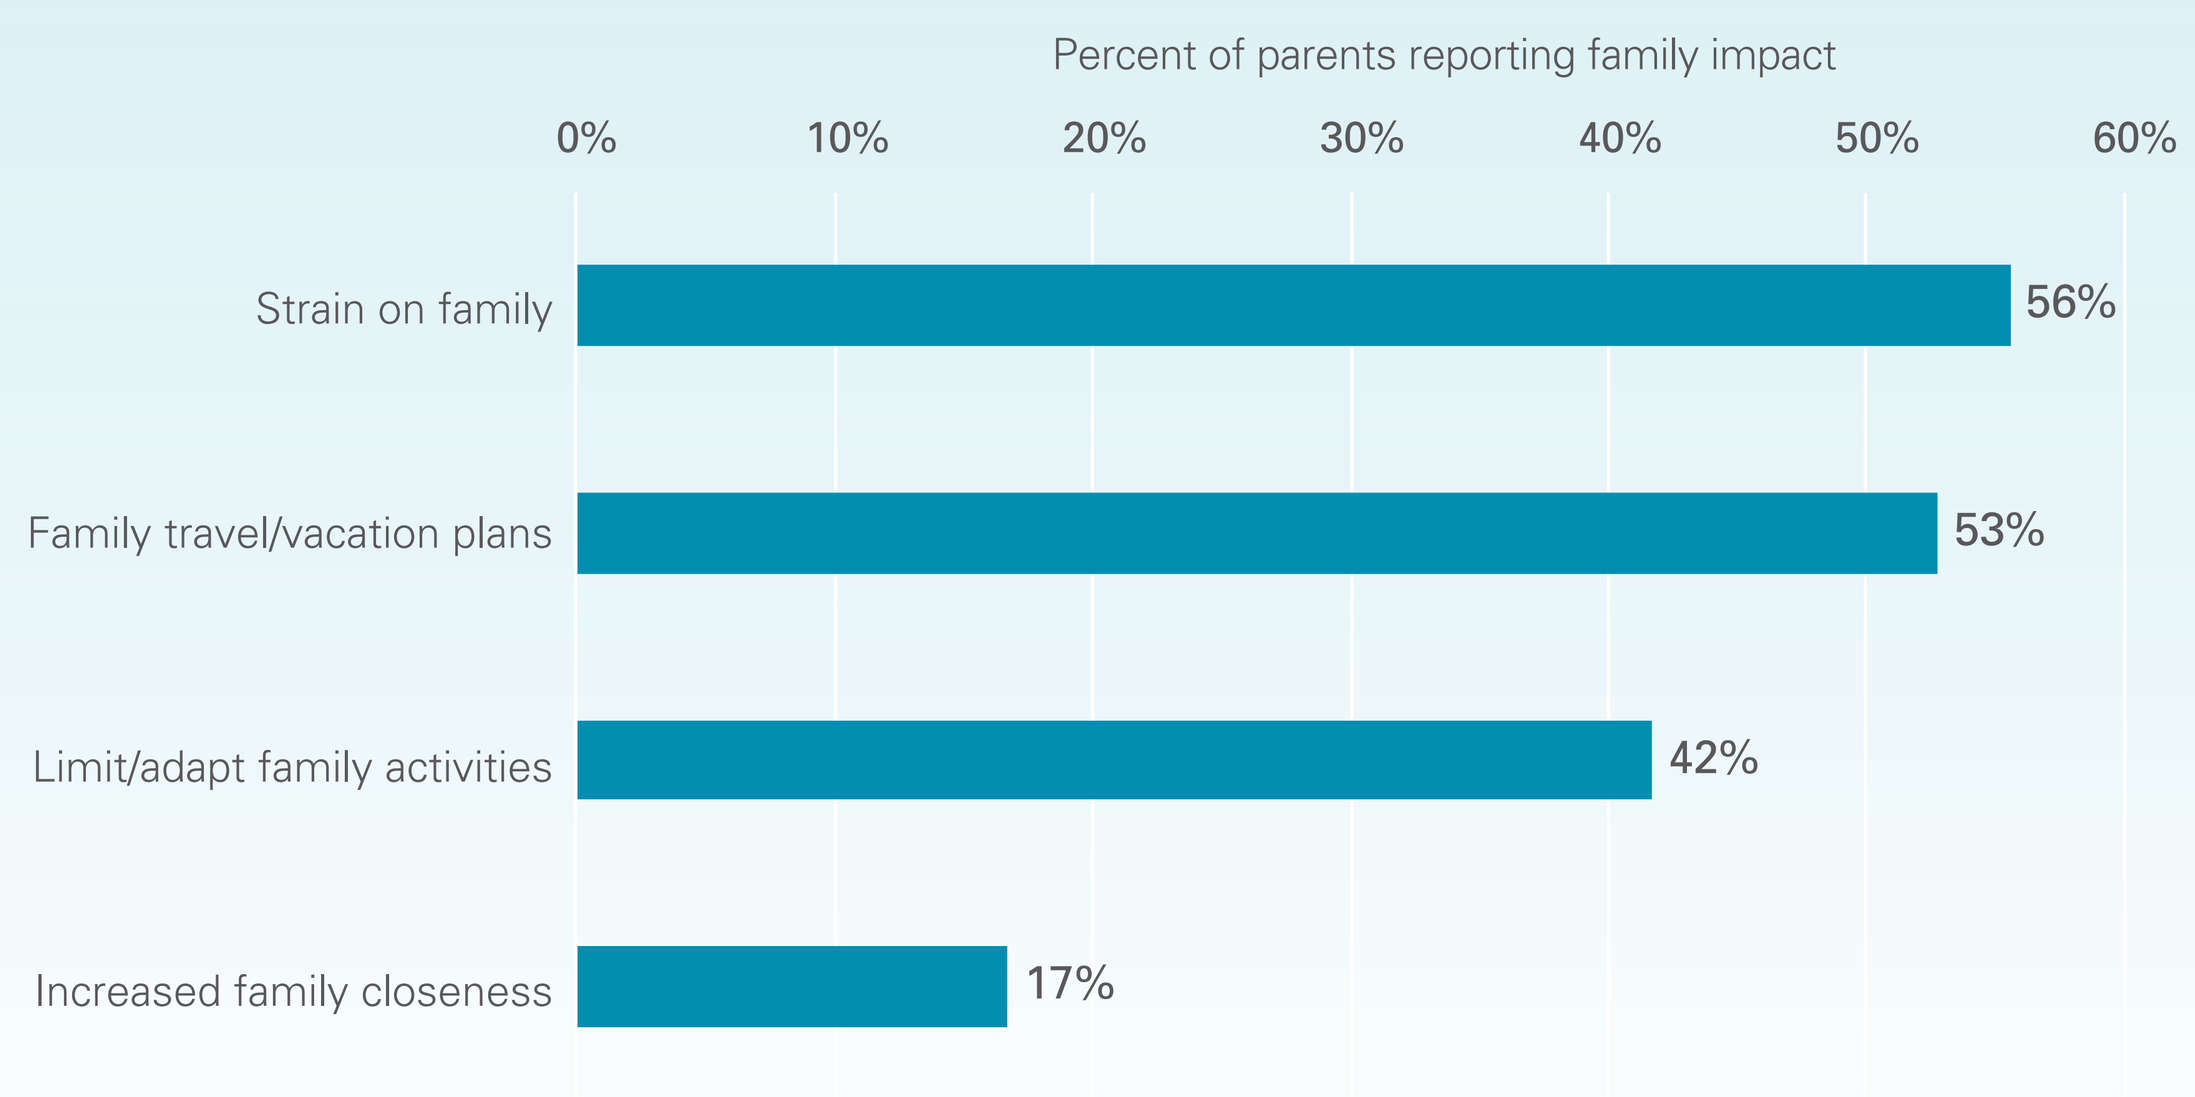
Percent of parents reporting family impact
0% 10% 20% 30% 40% 50% 60%
Strain on family 56%
Family travel/vacation plans 53%
Limit/adapt family activities 42%
Increased family closeness 17%

N=36.

A NOTE ON SUB-GROUP DIFFERENCES

The findings suggest that there may be some sub-group differences among parents, but significance tests could not be conducted due to small subsample sizes.

Among parents with and without ACH, results were generally similar, though parents with ACH were less likely to report some impacts compared to parents without ACH.

Additionally, the results suggested that there may be some differences by child age group:

- For example, helping child with self-care was more frequently reported by parents of younger children (2 to <9 years; 75%; n=18) than parents of older children (9 to <12 years; 50%, n=6)
- Results were generally similar for parents in the US and Spain

STUDY LIMITATIONS

Given the relatively small sample size, results should be interpreted with caution. Percentage differences in parents' reported impacts may not reflect actual differences in the population.

Due to small subsample sizes, significance tests for differences between parent subgroups could not be conducted.

Results may not be generalizable to other groups/populations. For example, parents' experiences related to having a child with ACH may vary in different countries, cultures, and healthcare systems.

CONCLUSIONS

- To our knowledge, this is the first study of how having a child with ACH affects parents' general quality of life and well-being
- The findings indicate that parents have a range of caretaking responsibilities for their children with ACH, including managing medical care/treatment, help with self-care, and advocating for child
- Additionally, many parents reported missed work time to care for child or take him/her to doctor appointments
- Parents also frequently expressed worries about their child's future, worries about child's physical health, child safety concerns, worries about child's social well-being, and feelings of being stressed/overwhelmed
- Despite these challenges, a majority of parents reported making social connections and friendships through dwarfism advocacy organizations, which were an important source of education and support

1. Pauli RM. Achondroplasia: a comprehensive clinical review. *Orphanet J Rare Dis.* 2019;14(1):1.
2. Hunter AG, Bankier A, Rogers JG, Sillence D, Scott CI, Jr. Medical complications of achondroplasia: a multicentre patient review. *J Med Genet.* 1998;35(9):705-712.
3. Wright MJ, Irving MD. Clinical management of achondroplasia. *Arch Dis Child.* 2012;97(2):129-134.
4. Ireland PJ, Donaghey S, McGill J, et al. Development in children with achondroplasia: a prospective clinical cohort study. *Dev Med Child Neurol.* 2012;54(6):532-537.
5. Ireland PJ, Johnson S, Donaghey S, et al. Developmental milestones in infants and young Australian children with achondroplasia. *J Dev Behav Pediatr.* 2010;31(1):41-47.
6. Hill V, Sahhar M, Aitken M, Savarirayan R, Metcalfe S. Experiences at the time of diagnosis of parents who have a child with a bone dysplasia resulting in short stature. *Am J Med Genet A.* 2003;122A(2):100-107.

Ascendis, Ascendis Pharma, the Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group. © September 2019 Ascendis Pharma A/S.